Risk Prediction of Hepatocellular Carcinoma in Patients with Cirrhosis: The ADRESS-HCC Risk Model

Flemming JA[1], Vittinghoff, E[2], Kim WR[3] and Terrault, NA[1].

Departments of Medicine[1], Epidemiology and Biostatistics[2], University of California San Francisco, San Francisco CA. [3]Department of Medicine, Mayo Clinic. Rochester MN.

Background and Aims

•AASLD guidelines recommend surveillance for HCC with ultrasonography every six months in cirrhotic patients who have a risk of HCC of greater than 1.5% per year.

•We aimed to develop an HCC risk prediction model that estimates the one-year probability of developing HCC in an individual patient with cirrhosis.

Methods

•Study design: Retrospective cohort using the Scientific Registry of Transplant Recipients database from 03/2002 – 12/2011.

•Adult liver transplant (LT) waitlisted candidates with cirrhosis without prevalent HCC and followed at least 6 months were included.

•Incident HCC was defined by the date of application for HCC MELD exception points if it occurred at least 6 months after listing for LT.

•Cox proportional hazards regression was used to develop and validate a risk prediction model from two non-random subsets of the cohort.

•Model discrimination was determined using c-indices.

•Model calibration was evaluated by comparing the predicted and observed cumulative incidences of HCC at one year in the validation set.

Results

•A total of 34,931 patients with cirrhosis were included in the cohort, 1,960 (5.6%) were diagnosed with HCC during follow-up.

•Demographics of the total cohort, derivation and validation sets are shown in table1.

Table 1: Baseline Characteristics of the Cohort

Variable	Entire Cohort (n=34,932)	Derivation Set (n=17,124)	Validation Set (n=17,808)	P Value*
Age (median years, IQR)	54 (48–59)	54 (48-59)	54 (48-59)	0.90
Sex, %				<.001
• Male	62.1	60.8	63.3	
• Female	37.9	39.2	36.7	
Race, %				<.001
• Caucasian non-Hispanic	74.0	70.5	77.3	
• Hispanic / Latino	16.1	19.6	12.7	
• African American	6.3	5.4	7.1	
• Asian	2.9	3.7	2.2	
• Other	0.7	0.8	0.7	
Body Mass Index (median, IQR)	28 (25–32)	28 (25–32)	28 (25–32)	0.16
Diabetes, %				<.001
• Yes	17.1	16.3	17.9	
• No	73.1	73.8	72.5	
• Missing	9.8	9.9	9.7	
Etiology of Cirrhosis, %				
• HCV	45.7	46.0	45.5	0.018
• HBV	2.8	3.0	2.6	
• NASH / Cryptogenic	17.5	17.6	17.4	
• Alcohol	18.3	18.3	18.3	
• PBC	4.6	4.5	4.6	
• PSC	6.0	5.7	6.4	
• AIH	3.8	3.8	3.7	
• A1AT	0.7	0.6	0.8	
• HH	0.5	0.5	0.5	
• Wilson	0.2	0.2	0.2	
MELD Score (median, IQR)	13 (10–16)	12 (10–15)	13 (11–16)	<.001
Childs-Turcotte-Pugh Class, %				<.001
• A	12.9	14.8	11.2	
• B	54.1	54.6	53.6	
• C	33	30.6	35.3	
Bilirubin (median mg/dl, IQR)	2 (1.3–3.3)	1.9 (1.2–3.2)	2.1 (1.3–3.4)	<.001
Albumin (median g/dl, IQR)	3.1 (2.6–3.5)	3.1 (2.7–3.6)	3.0 (2.6–3.5)	<.001
INR (median, IQR)	1.3 (1.2–1.5)	1.3 (1.2–1.5)	1.3 (1.2–1.5)	<.001
Creatinine (median mg/dl, IQR)	0.91 (0.8–1.2)	0.9 (0.8–1.2)	1.0 (0.8–1.2)	<.001
Sodium (median Meq/L, IQR)	137 (135–140)	137 (135–140)	137 (134–139)	<.001
HCC during follow-up, % (n)	5.6 (1,960)	6.5 (1,116)	4.7 (844)	
Follow-up time (median yrs, IQR)	1.26 (0.47–2.83)	1.51 (0.58–3.2)	1.07 (0.39–2.5)	
Total person-time, years	67,014	36,719	30,295	

* P value for comparison between the derivation and validation sets.
HCV: Hepatitis C; HBV: Hepatitis B; NASH: non-alcoholic steatohepatitis; EtOH: alcohol; PBC: primary biliary cirrhosis; PSC: primary sclerosing cholangitis; AIH: autoimmune hepatitis; A1AT: alpha-1 anti-trypsin; HH: hereditary hemchromatosis.

Results

•**<u>A</u>**ge, **<u>D</u>**iabetes, **<u>R</u>**ace, **<u>E</u>**tiology of cirrhosis, **<u>S</u>**ex and **<u>S</u>**everity of liver disease were associated with HCC (table 2) and used to develop the ADRESS-HCC risk model (table 3).

Table 2: Cox models evaluating risk factors for ADRESS-HCC model (derivation cohort n=17,124)

Variable	Univariate Model HR (95% CI)	Univariate Model P Value	Multivariate Model HR (95% CI)	Multivariate Model P value
Age (per year increase)	1.04 (1.03-1.04)	<0.001	1.05 (1.04-1.06)	<0.001
Etiology of Cirrhosis‡				
- Autoimmune	ref	-	ref	-
- EtOH/Metabolic	2.28 (1.69-3.07)	<0.001	1.43 (1.04-1.97)	0.030
- Viral	4.72 (3.55-6.26)	<0.001	3.38 (2.50-4.55)	<0.001
Diabetes present	1.31 (1.11-1.54)	0.001	1.25 (1.06-1.48)	0.008
Male Sex	2.03 (1.76-2.33)	<0.001	1.99 (1.71-2.30)	<0.001
CTP Score (per unit increase)	1.17 (1.13-1.21)	<0.001	1.17 (1.13-1.21)	<0.001
Non-Caucasian Race	1.26 (1.12-1.43)	<0.001	1.20 (1.06-1.37)	0.005
MELD Score (per unit increase)	1.02 (1.01-1.04)	0.007	-	-
BMI (per 5 unit increase)	1.01 (0.96-1.07)	0.681	-	-
Presence of TIPS	0.88 (0.69-1.12)	0.315	-	-
Previous Variceal Hemorrhage	0.93 (0.67-1.29)	0.643	-	-
Bilirubin (per unit increase)	1.03 (1.01-1.05)	<0.001	-	-
INR (per unit increase)	1.36 (1.22-1.52)	<0.001	-	-
Creatinine (log)	1.16 (1.00-1.33)	0.040	-	-

‡Autoimmune: AIH, PBC, PSC; EtOH/Metabolic: HH, A1AT, NASH/cryptogenic, alcohol; Viral: HBV, HCV

Table 3: Calculation of one-year HCC risk using ADRESS-HCC model

<u>Step 1</u>: Calculate ADRESS-HCC Score
= ([<u>A</u>ge] + [<u>D</u>iabetes] + [<u>R</u>ace] + [<u>E</u>tiology] + [<u>S</u>ex] + [<u>S</u>everity]) where:
Age = [age in years x 0.0532]
Diabetes = 0.2135 if present; = 0 if absent
Race = 0.2058 if non-Caucasian or Hispanic; = 0 if Caucasian, non-Hispanic
Etiology = 0 if Autoimmune; = 0.3509 if EtOH/Metabolic; = 1.246 if Viral
Sex = 0.5114 if male; = 0 if female
Severity = [CTP score (5-15) x 0.1170]

<u>Step 2</u>: Baseline hazard at one year: S0(t) = 0.99986

<u>Step 3</u>: $[100 * (1 - S0(t)^{*}\exp^{(\text{ADRESS-HCC Score})})]$ = one-year HCC risk (%)

Results

•The C-indices in the derivation and validation sets were 0.704 and 0.691 respectively.

•The calibration of the ADRESS-HCC risk model was excellent as the observed HCC risk closely and matched the predicted values (figure 1).

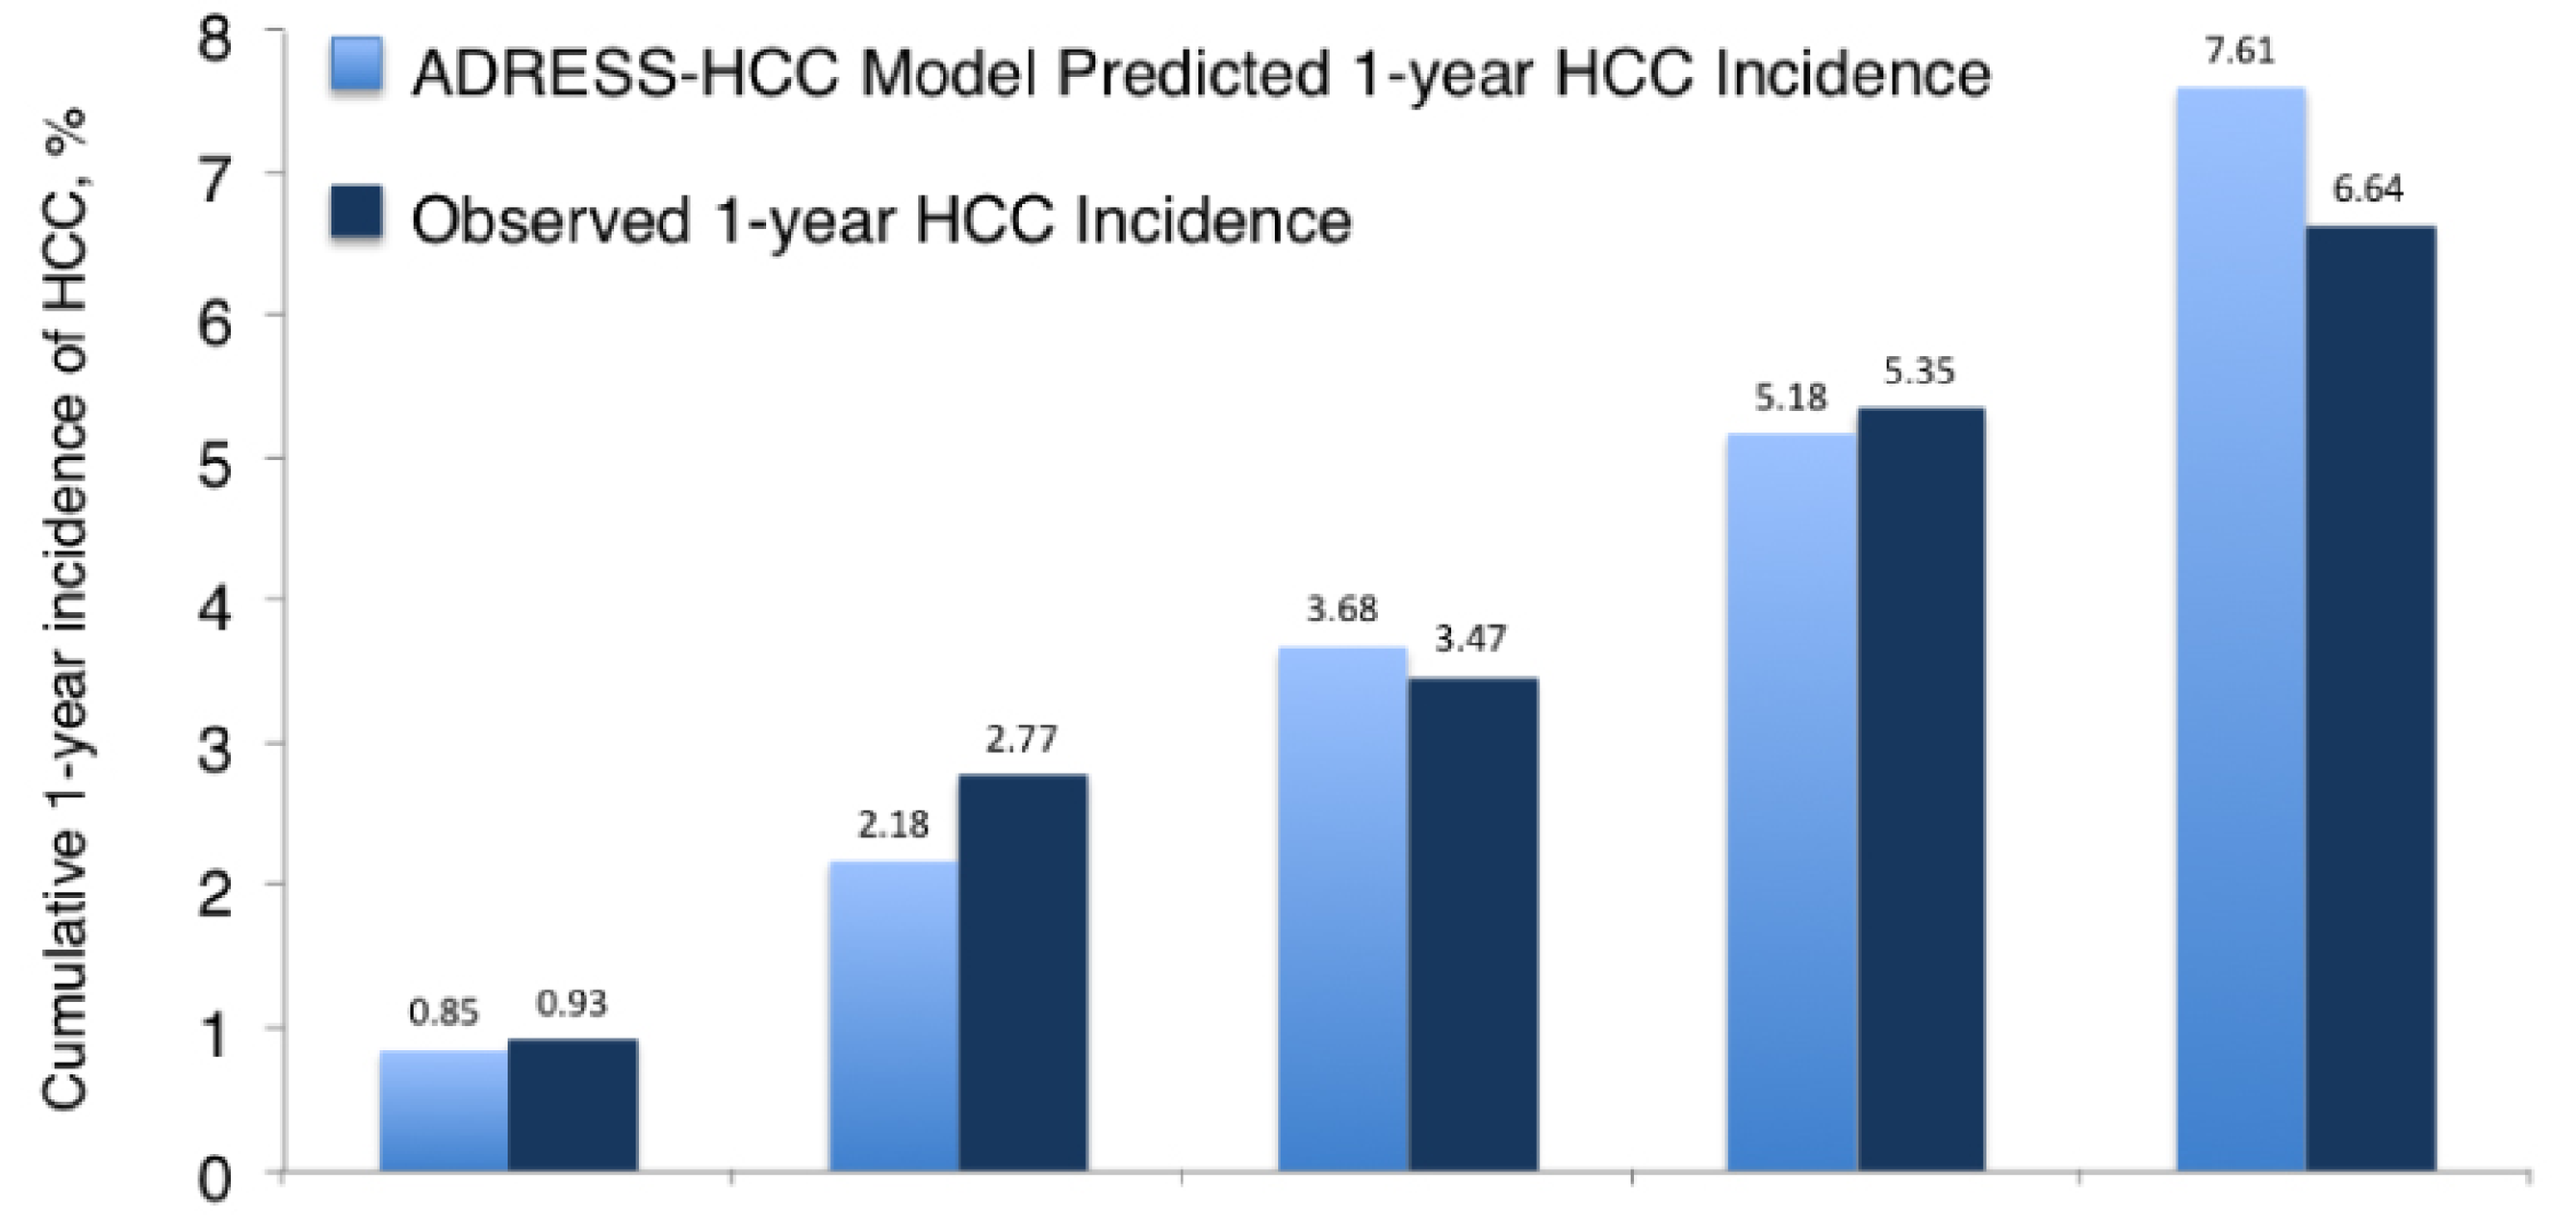
ADRESS-HCC Model Predicted 1-year HCC Incidence
Observed 1-year HCC Incidence
Cumulative 1-year incidence of HCC, %
0
1
2
3
4
5
6
7
8
0.85
0.93
2.18
2.77
3.68
3.47
5.18
5.35
7.61
6.64

Model Predicted 1-year Incidence	< 1.5%	1.5% - 3%	3% - 4.5%	4.5% - 6%	> 6%
Model Point Score Range	< 4.67	4.67 – 5.37	5.37 – 5.77	5.77 – 6.06	> 6.06

Figure 1: Calibration of the ADRESS-HCC risk model

Conclusions

•The ADRESS-HCC risk model is a validated tool that can be used to counsel a wide variety of cirrhotic patients regarding their risk of HCC using individual clinical characteristics.

•It can also be used to identify patients who meet the AASLD criteria for entrance into HCC surveillance programs.

•Validation in other cohorts of cirrhotics is needed.

Supported by the Canadian Association for the Study of the Liver (CASL) / Merck Fellowship in Clinical Hepatology (Flemming)