1897

Blockage of HBV Virus Replication and Inhibition of cccDNA Establishment by Core Protein Allosteric Modifiers (CpAMs)

Qi Huang, Yuhua Zong, Alex Mercier, Emily Connelly, G. Renuka Kumar, Dawei Cai, Cathal Mahon, Pao-Chen Li, Lida Guo, Yi Zhou, Shawn Sun, Geoffrey Chen, Katherine Nabel, Lee D. Arnold, William Turner, Leping Li, Simon Haydar, Uri Lopatin and Richard Colonno

Assembly Biosciences, Inc., San Francisco, CA, and Bloomington, IN, United States

Background

Approximately 240 million people worldwide are chronically infected with Hepatitis B virus (HBV) and are at risk of developing chronic liver diseases, such as hepatitis, fibrosis, cirrhosis and hepatocellular carcinoma (HCC). Currently approved treatments for chronic HBV include nucleos(t)ide analogs (Nucs) such as entecavir (ETV) and tenofovir, and interferons (IFN and pegylated-interferon (PEG-IFN)). While Nucs are very effective at inhibiting HBV replication for prolonged periods, cure rates occur in <10% of treated patients. The deficiency of these therapies appears to be their limited effect on cccDNA formation. Hence, new classes of anti-HBV molecules are needed to significantly improve cure rates in chronic HBV patients. HBV core protein (Cp) is involved in multiple steps of the HBV life cycle, including the formation, amplification and maintenance of cccDNA. A novel class of direct acting HBV antivirals, Core Protein Allosteric Modifiers (CpAMs), have been discovered to target HBV core protein and reduce HBV replication and cccDNA levels.[1,2] Here we report on the mechanism of action of a series of proprietary CpAMs using a variety of preclinical assays.

Materials and Methods

Inhibitors: Assembly Biosciences' CpAMs were synthesized at Sai Life Sciences. GLS4 (CpAM) was synthesized by published procedures.[3,4] ETV[5] (Nuc) was purchased from ACME Bioscience.

Cell culture and compound treatment: Huh7 cells were transfected with plasmids expressing HBV constructs in 96-well plates and treated with compounds. Supernatants and cells were harvested at 5 days post-infection. Primary Human Hepatocytes (PHH; Lonza) were infected with HBV from HepAD38 at an MOI of 50-250 and treated with inhibitors at the indicated time points and concentrations. Supernatants and cells were harvested at indicated time points.

Molecular profiling of CpAMs: HepAD38 cells[6] were treated with inhibitors and induced simultaneously. Cells were retreated 3 days later and harvested 6 days post induction. HBV total RNA/encapsidated pgRNA, Cp, capsid, and capsid associated core DNA were detected by Northern Blot, Western Blot, Enzyme Immunoassay (EIA), and Southern Blot, respectively, as previously described.[7]

HBcAg quenching assay: Huh7 cells were transfected with a plasmid expressing the complete Cp open reading frame under the control of a CMV promoter. Supernatants were harvested 4 days post transfection and subjected to a custom in-house electrochemiluminescent (ECL) ELISA. This assay detects an HBc epitope that gets masked upon addition of CpAMs.

HBV DNA quantification in transient transfected cells: HBV DNA levels in DNase I-treated supernatants were quantified by Taqman quantitative PCR (qPCR) using primers and a probe specific to the HBV core gene.

HBV DNA/pgRNA quantification in PHH: HBV DNA/pgRNA levels were quantified using bDNA probes (Affimetrix) specific to the relevant HBV sequences.

HBeAg/HBsAg levels quantification: HBe and HBs antigens were quantified using commercial enzyme-linked immunosorbent assay (ELISA) kits (Autobio Diagnostics and Beijing Wantai Biological Pharmacy Enterprise, respectively) as well as custom in-house ECL ELISAs.

HBV RNA staining: Total HBV RNA levels were visualized using ViewRNA technology (Affymetrix) and a set of bDNA probes designed to detect all species of HBV mRNAs.

cccDNA Southern Blots: Cells infected with HBV from HepAD38 were treated with inhibitors. Four days post infection, cells were harvested and extrachromosomal DNA extracted by a Hirt-modified method.[8] Non-supercoiled DNA was removed via T5 exonuclease (NEB) digestion. High-fidelity EcoRI endonuclease (NEB) was then used to linearize supercoiled HBV cccDNA.

EC_{50} calculations: EC_{50} values were calculated using GraphPad Prism software.

Cp Involved in Multiple Steps of HBV Lifecycle

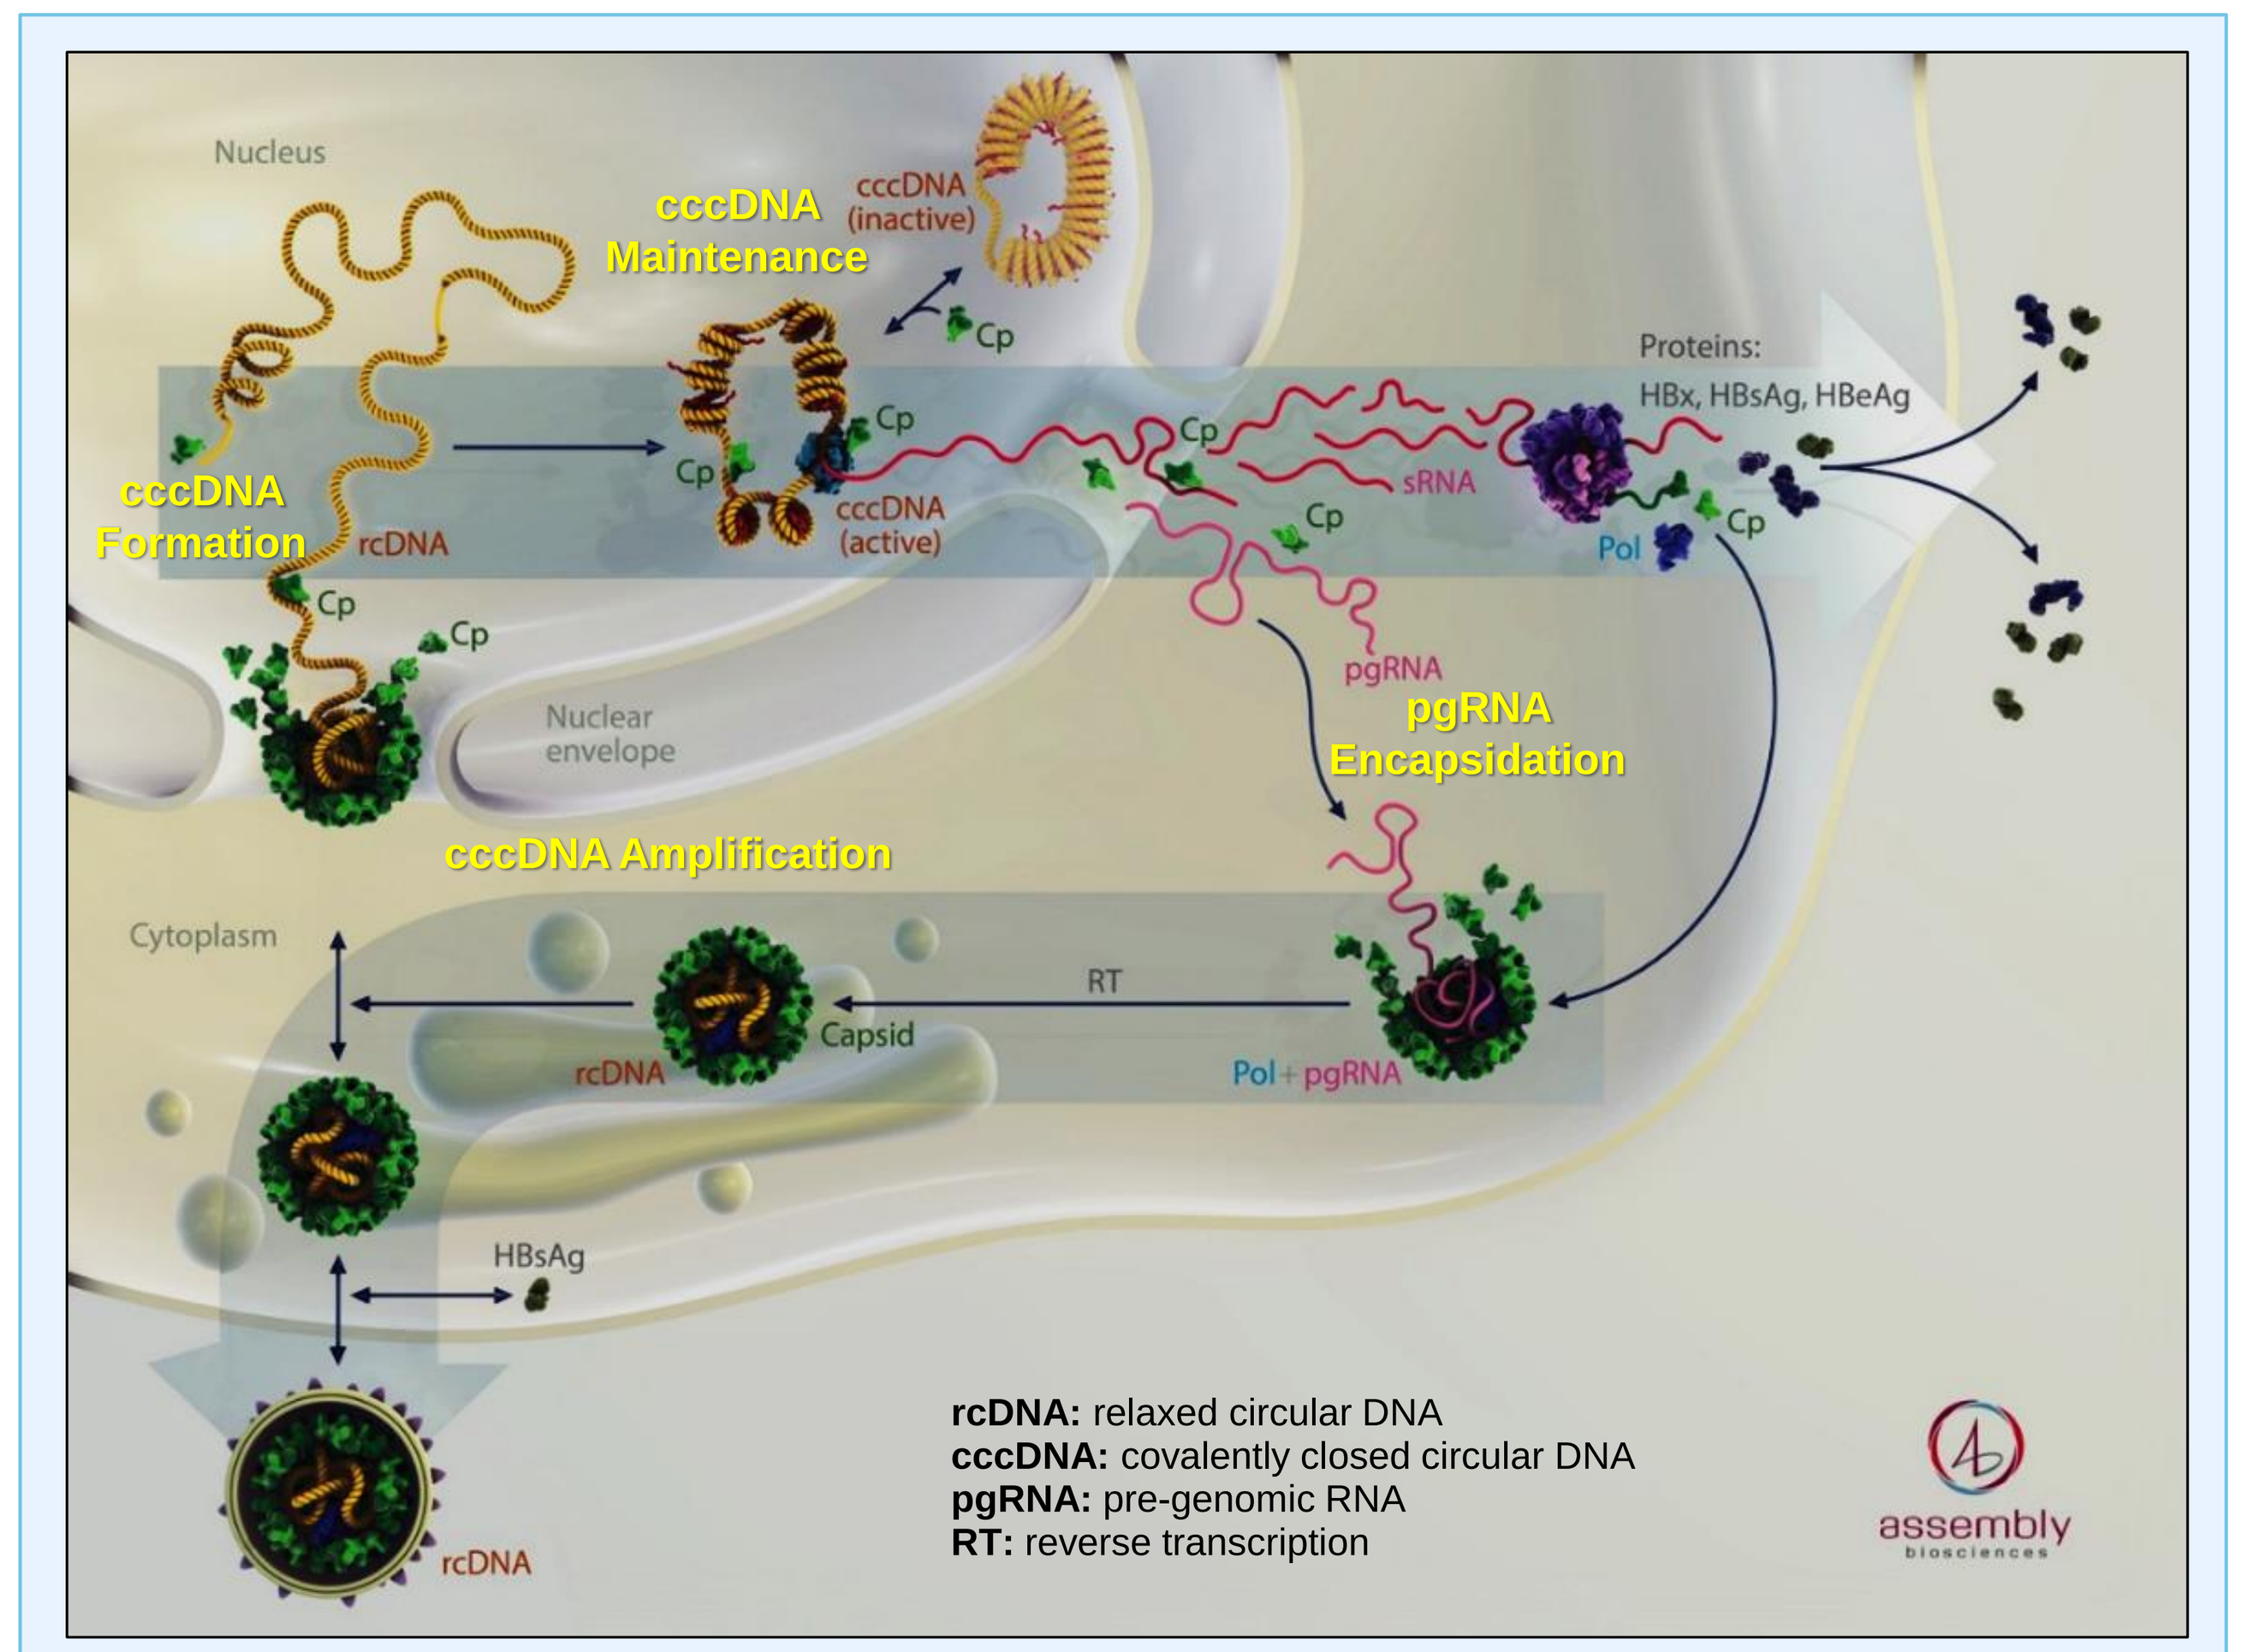
Nucleus
cccDNA Maintenance
cccDNA (inactive)
cccDNA (active)
cccDNA Formation
rcDNA
Cp
Proteins: HBx, HBsAg, HBeAg
sRNA
Pol
pgRNA
pgRNA Encapsidation
Nuclear envelope
cccDNA Amplification
Cytoplasm
RT
Capsid
Pol + pgRNA
HBsAg
rcDNA: relaxed circular DNA
cccDNA: covalently closed circular DNA
pgRNA: pre-genomic RNA
RT: reverse transcription

CpAMs Perturb Cp Assembly and HBV Replication

Molecular Profiling of CpAMs

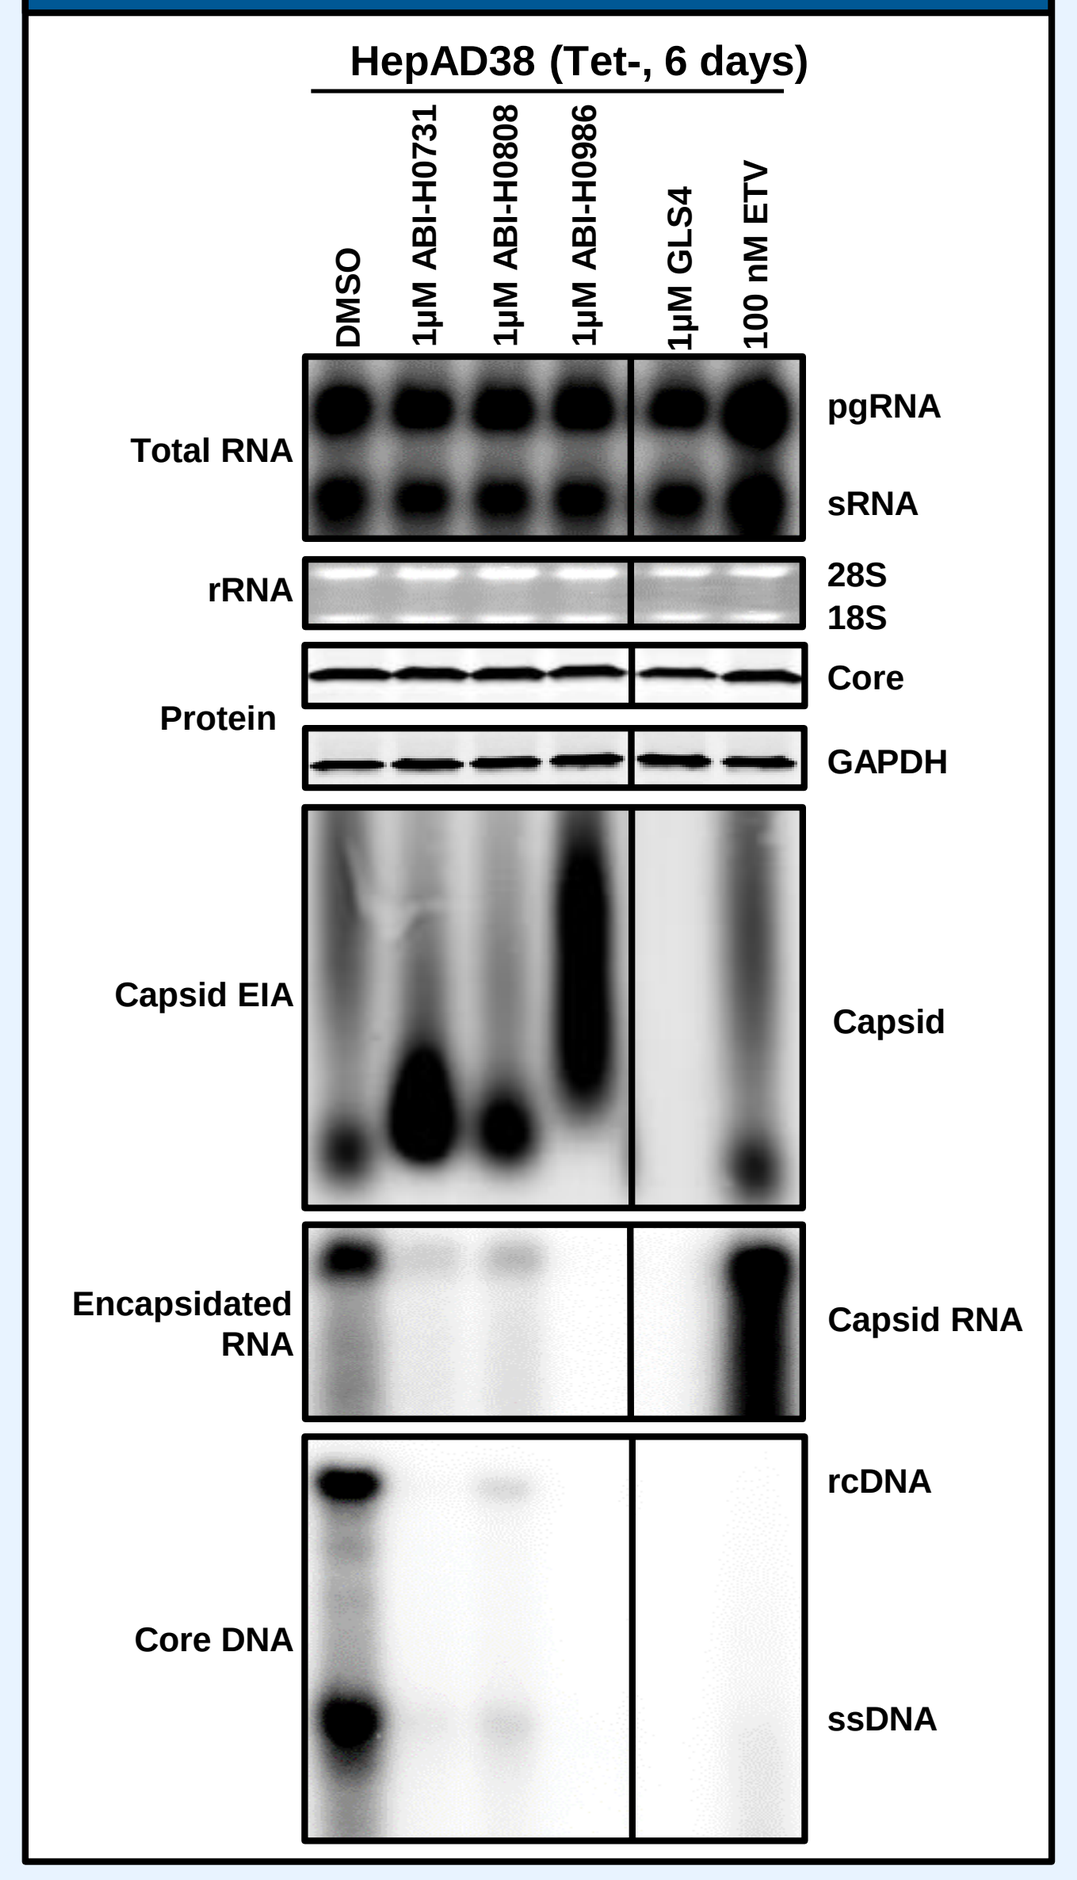
HepAD38 (Tet-, 6 days)
DMSO
1µM ABI-H0731
1µM ABI-H0808
1µM ABI-H0986
1µM GLS4
100 nM ETV
Total RNA
pgRNA
sRNA
rRNA
28S
18S
Protein
Core
GAPDH
Capsid EIA
Capsid
Encapsidated RNA
Capsid RNA
Core DNA
rcDNA
ssDNA

HBcAg Quenching Assay

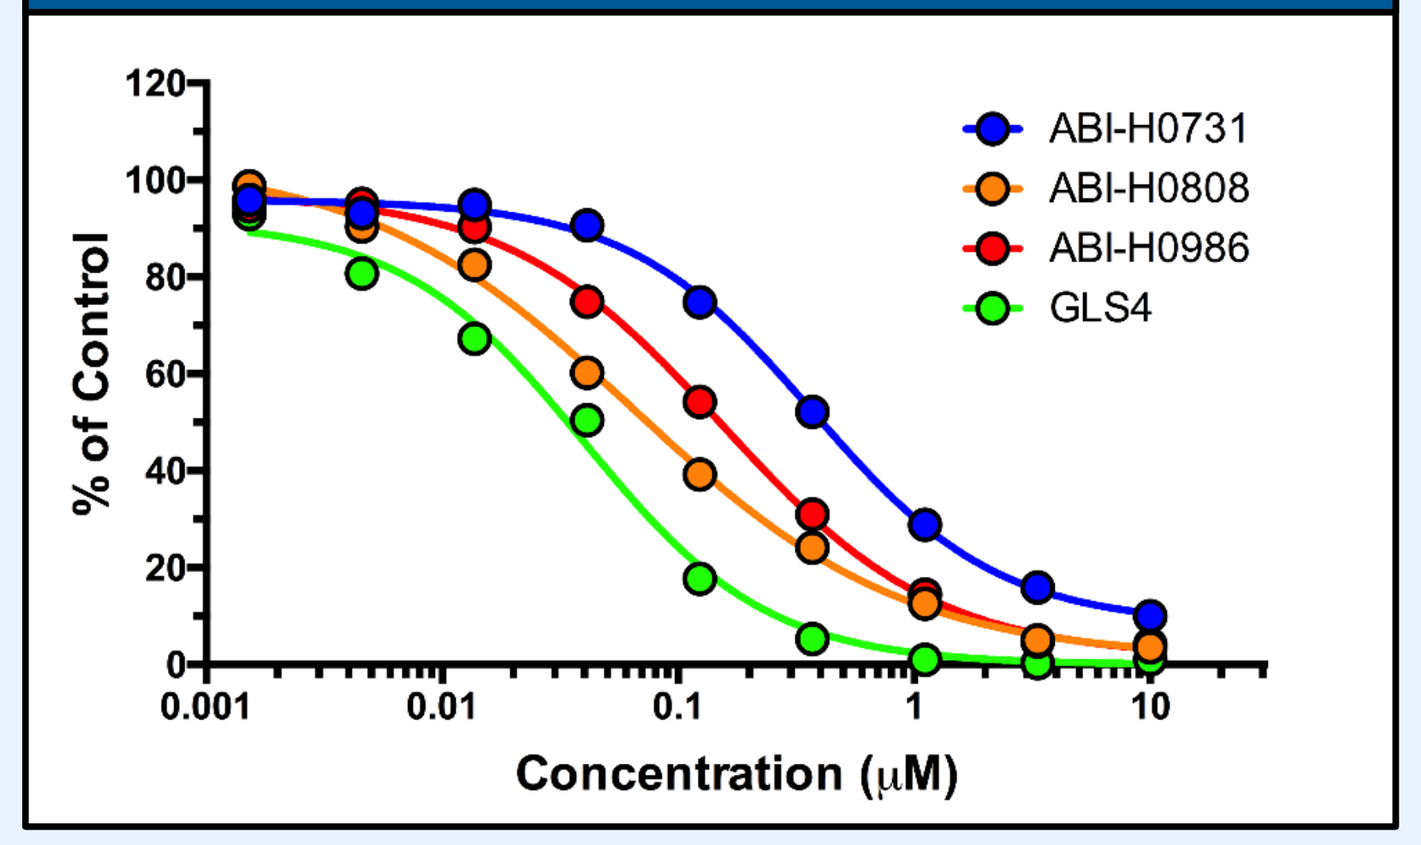
ABI-H0731
ABI-H0808
ABI-H0986
GLS4
% of Control
Concentration (µM)

Compound	EC_{50} ± SD (nM)
ABI-H0731	374 ± 120
ABI-H0808	64 ± 22
ABI-H0986	160 ± 64
GLS4	40 ± 9

- CpAMs perturbed capsid assembly based on EIA and HBcAg quenching results
- CpAMs inhibited, whereas ETV greatly increased, the amount of packaged HBV RNA
- CpAMs resulted in a blockage of viral DNA synthesis

CpAMs Exhibit Broad Genotypic Activity

HBV Genotype	EC_{50} ± SD (nM)	
	ABI-H0731	ETV
A	113 ± 22	1.9 ± 0.5
B	86 ± 11	2.3 ± 0.5
C	88 ± 27	1.2 ± 0.1
D	142 ± 48	5.0 ± 1.9

- CpAM ABI-H0731 exhibited mean EC_{50}s ranging from 86 to 142 nM in reducing viral replication of the four major HBV genotypes (A-D)
- Assembly CpAMs possess broad genotypic antiviral activity

CpAMs Active Against Nuc Resistant Mutant

HBV Construct	EC_{50} ± SD (nM)	
	ABI-H0731	ETV
Wild type	183 ± 14	13 ± 7
L180M+M204V	194 ± 1	> 100

- CpAM ABI-H0731 retained antiviral potency against the Nuc resistant variant L180M+M204V (Genotype D)
- CpAM activity should be retained in patients harboring Nuc-resistant variants

Time Dependent Inhibition of cccDNA Formation

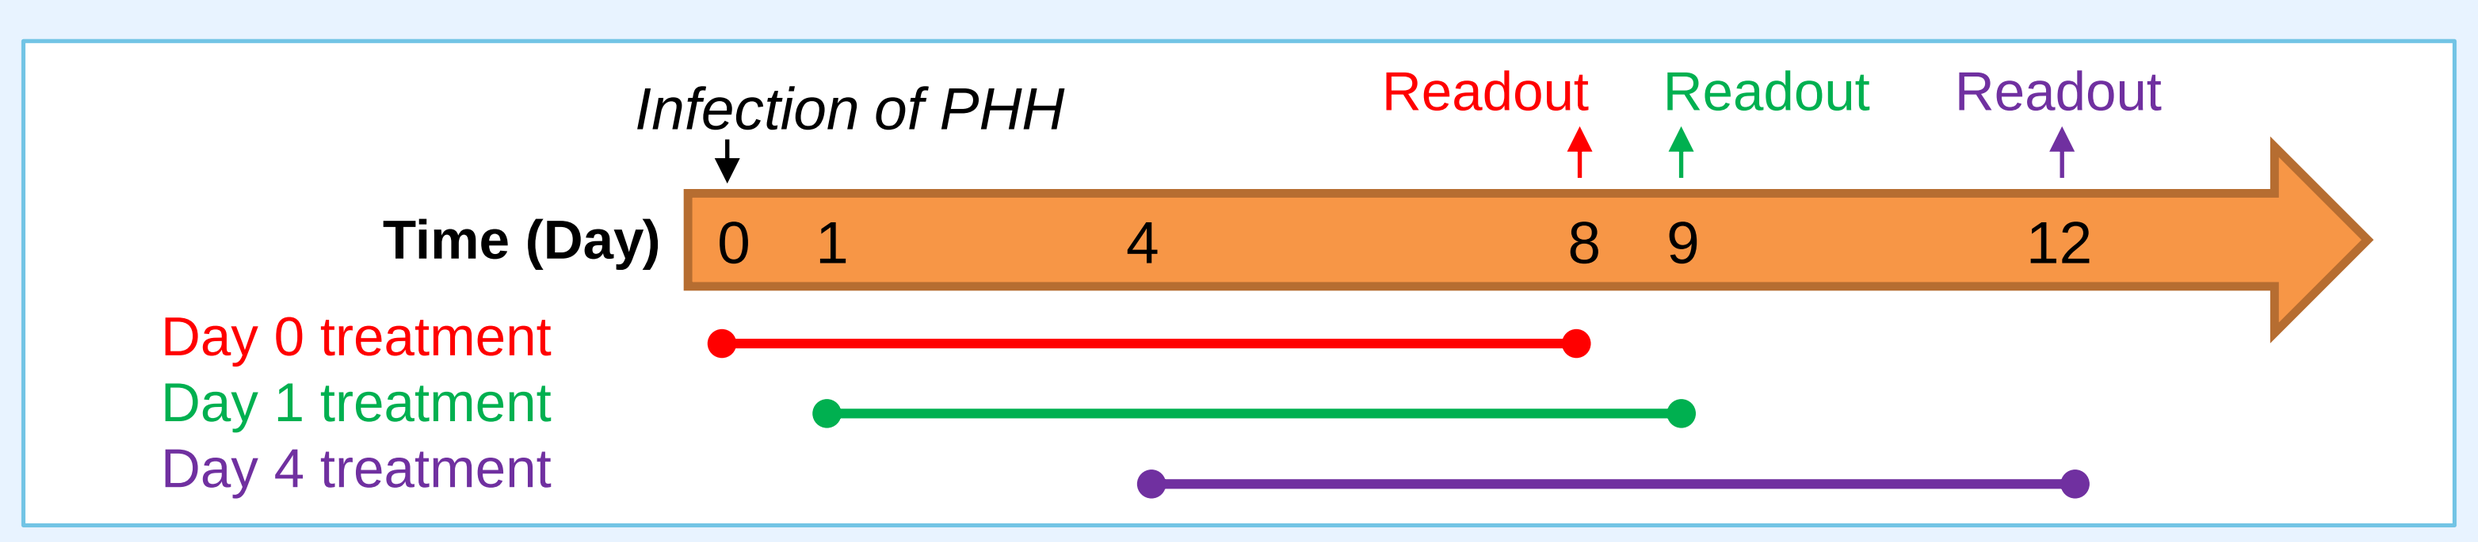
Infection of PHH
Readout
Readout
Readout
Time (Day)
0 1 4 8 9 12
Day 0 treatment
Day 1 treatment
Day 4 treatment

Inhibitor	Day 0 EC_{50} (nM)			
	HBV DNA	HBeAg	HBsAg	pgRNA
ABI-H0731	124	2,530	2,610	1,090
ABI-H0808	54.6	181	286	226
ABI-H0986	78.9	778	834	265
GLS4	1,980	1,660	1,610	1,900
ETV	<0.1	>>100*	>>100*	>>100*

Inhibitor	Day 1 EC_{50} (nM)			
	HBV DNA	HBeAg	HBsAg	pgRNA
ABI-H0731	153	3,710	2,170	1,580
ABI-H0808	112	1,440	911	834
ABI-H0986	74.5	943	1,020	1,550
GLS4	3,270	2,610	999	4,250
ETV	<0.1	>>100*	>>100*	>>100*

Inhibitor	Day 4 EC_{50} (nM)			
	HBV DNA	HBeAg	HBsAg	pgRNA
ABI-H0731	184	>10,000	>10,000	>10,000
ABI-H0808	85.3	>10,000	>10,000	>10,000
ABI-H0986	112	3,150	4,640	8,840
GLS4	9,460	>10,000	>10,000	>10,000
ETV	<0.1	>>100*	>>100*	>>100*

*Incomplete inhibition

Time addition of ABI-H0986: HBV DNA and HBsAg

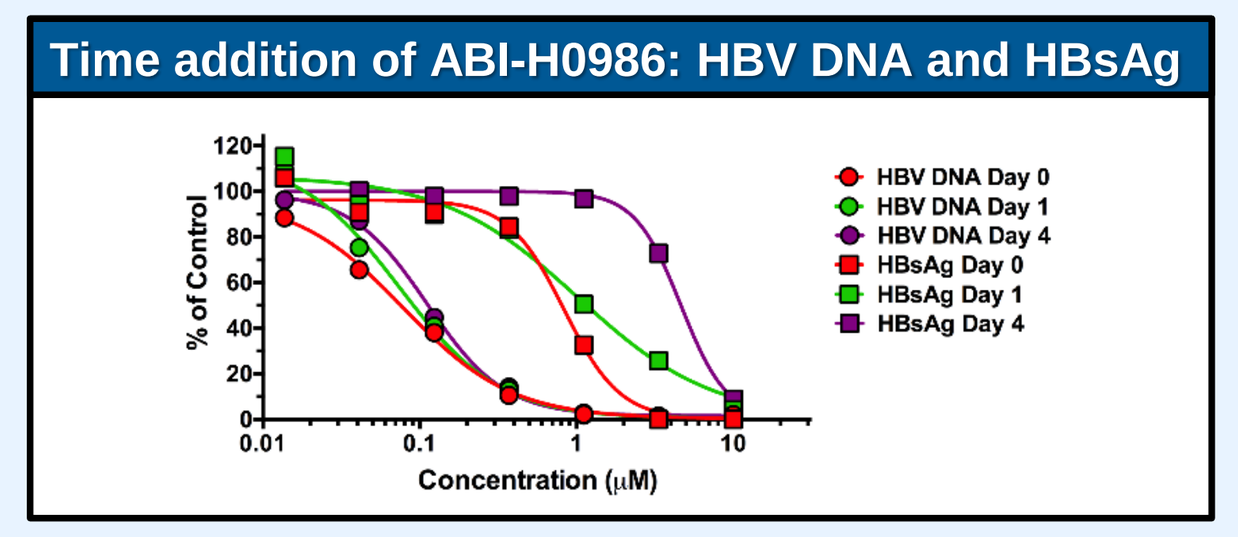
HBV DNA Day 0
HBV DNA Day 1
HBV DNA Day 4
HBsAg Day 0
HBsAg Day 1
HBsAg Day 4
% of Control
Concentration (µM)

- CpAMs and ETV inhibited HBV replication independent of the time of treatment
- CpAMs inhibited cccDNA formation when cells were treated early following HBV infection. They had limited effects against HBeAg, HBsAg and pgRNA (all surrogates of cccDNA activity) when added to cells four days following infection
- ETV had no effect on cccDNA formation, independent of the time of compound addition

CpAMs Impair cccDNA Formation in HepG2-NTCP and PHH Infection Models

Profiling of CpAMs Antiviral Activities

Parameter	HepG2-NTCP EC_{50} ± SD (nM)				
	ABI-H0731	ABI-H0808	ABI-H0986	GLS4	ETV
Viral DNA	307 ± 41	95 ± 36	57 ± 9	58 ± 6	0.8 ± 0.2
HBeAg	4,950 ± 520	372 ± 27	920 ± 126	549 ± 48	>>100
HBsAg	7,300 ± 770	945 ± 82	1,990 ± 170	673 ± 41	>>100
pgRNA	2,680 ± 580	419 ± 31	1,050 ± 100	279 ± 43	>>100

Parameter	PHH EC_{50} ± SD (nM)				
	ABI-H0731	ABI-H0808	ABI-H0986	GLS4	ETV
Viral DNA	154 ± 18	80 ± 17	70 ± 7	1,940 ± 40	<0.1
HBeAg	2,210 ± 310	196 ± 63	677 ± 102	1,480 ± 170	>>100*
HBsAg	3,000 ± 270	310 ± 64	892 ± 133	1,860 ± 430	>>100*
pgRNA	1,840 ± 380	305 ± 67	611 ± 233	1,930 ± 30	>>100*

*Incomplete inhibition

- CpAMs reduced HBeAg, HBsAg and pgRNA expression (markers of cccDNA formation)
- Assembly CpAMs exhibited improved antiviral activities, unlike other CpAMs such as GLS4, which lost potency in PHH
- ETV showed minimal inhibition of antigen production in PHH when tested up to 100 nM (>1,000x EC_{50})

Detection of HBV RNAs by ViewRNA in PHH

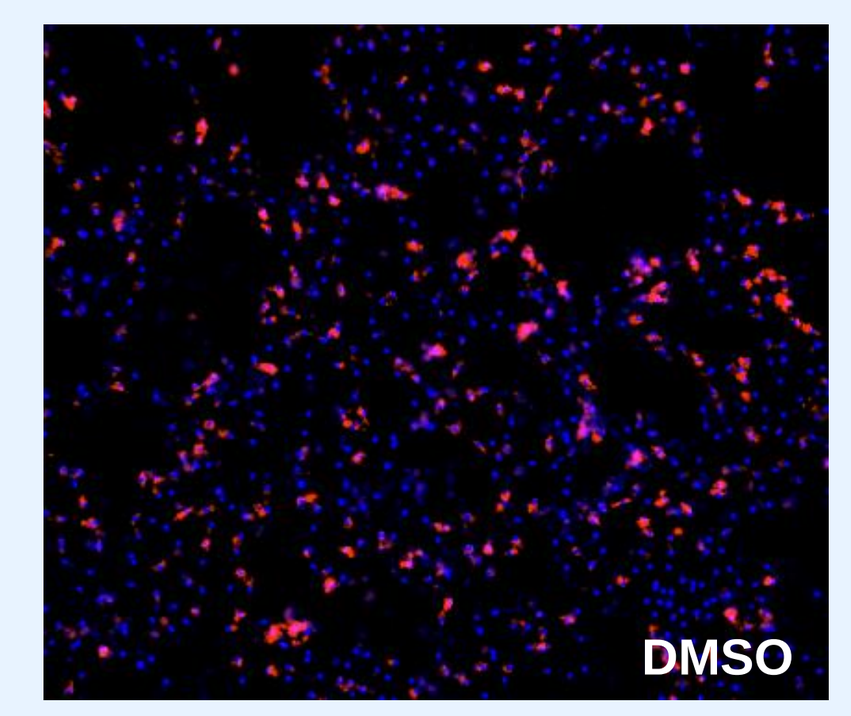
DMSO

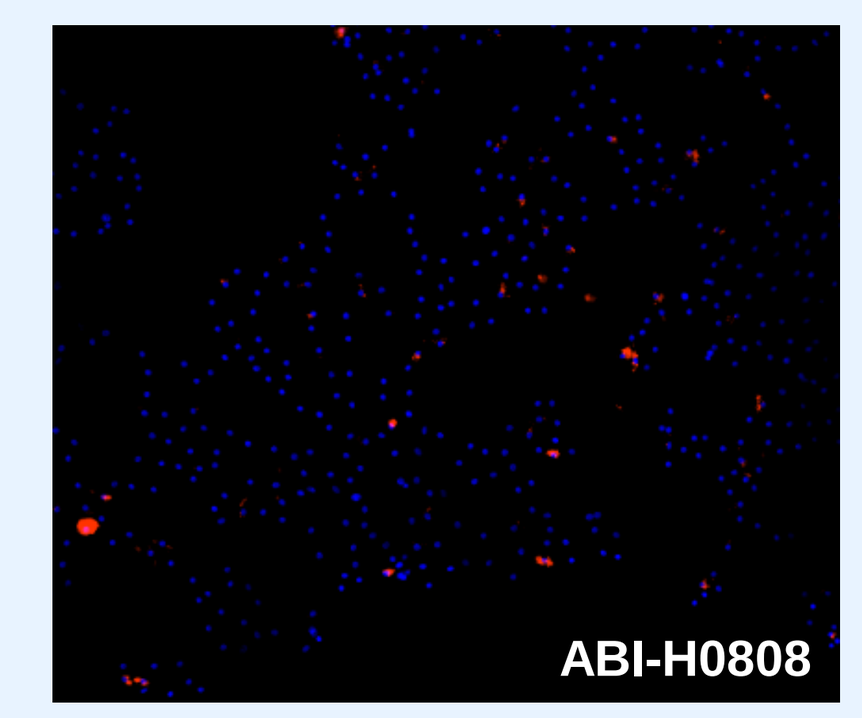
ABI-H0808

- HBV RNA levels in HBV infected PHH were detected using ViewRNA technology following early treatment with 3 µM CpAM ABI-H0808
- ABI-H0808 reduced HBV RNA levels by >80% in infected PHH

HBV cccDNA Detection by Southern Blot

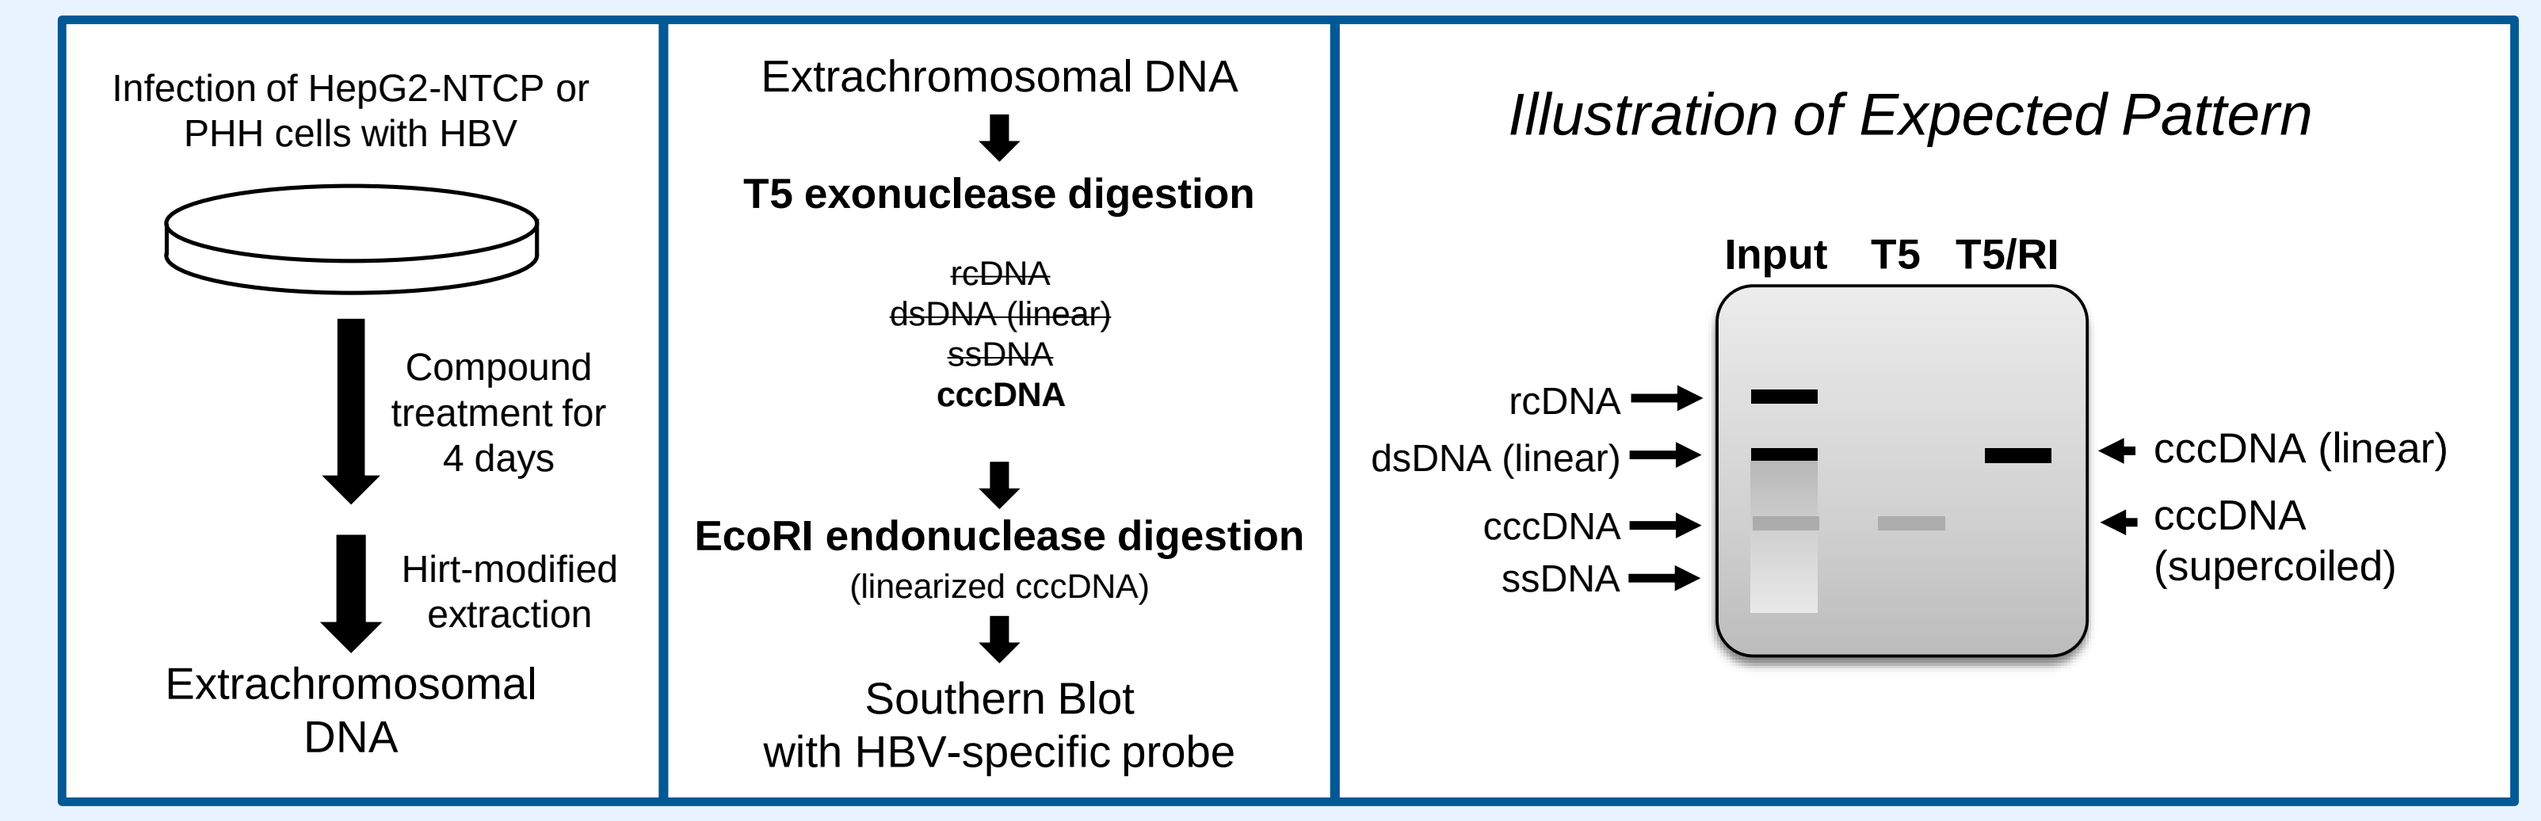
Infection of HepG2-NTCP or PHH cells with HBV
Compound treatment for 4 days
Hirt-modified extraction
Extrachromosomal DNA
Extrachromosomal DNA
T5 exonuclease digestion
rcDNA
dsDNA (linear)
ssDNA
cccDNA
EcoRI endonuclease digestion
(linearized cccDNA)
Southern Blot with HBV-specific probe
Illustration of Expected Pattern
Input T5 T5/RI
rcDNA
dsDNA (linear)
cccDNA
ssDNA
cccDNA (linear)
cccDNA (supercoiled)

HepG2-NTCP

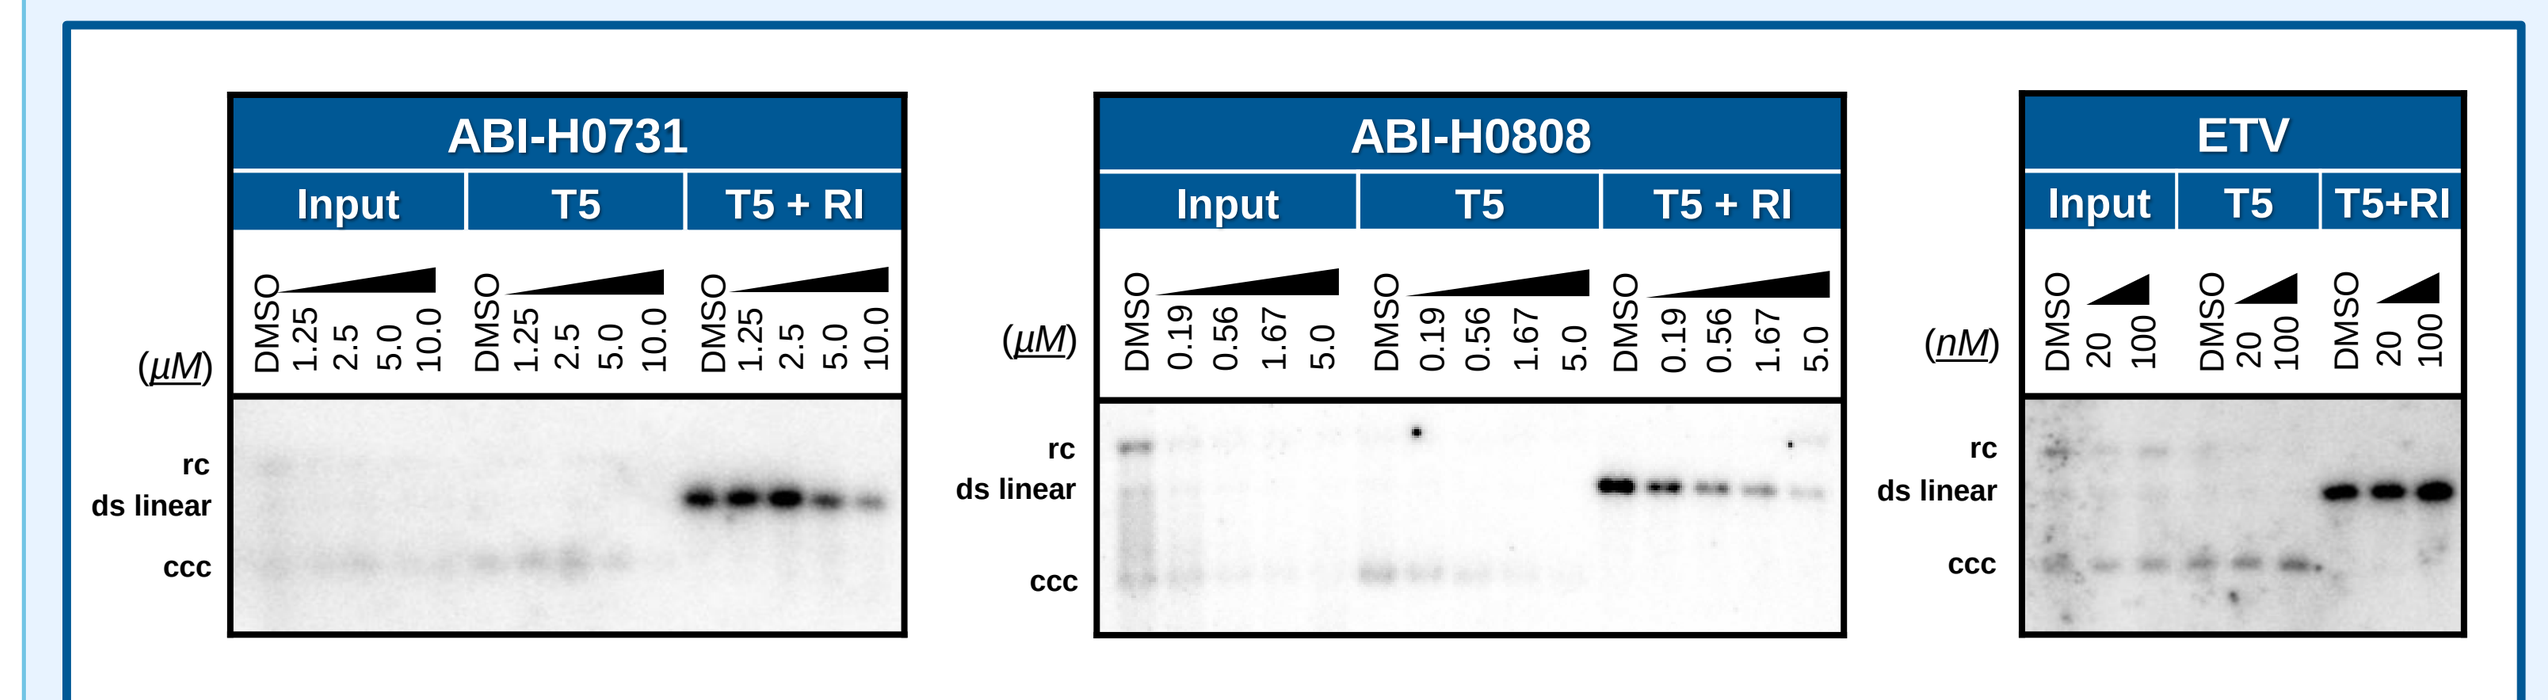
ABI-H0731
Input T5 T5 + RI
(µM)
DMSO 1.25 2.5 5.0 10.0
ABI-H0808
DMSO 0.19 0.56 1.67 5.0
ETV
Input T5 T5+RI
(nM)
DMSO 20 100
rc
ds linear
ccc

PHH

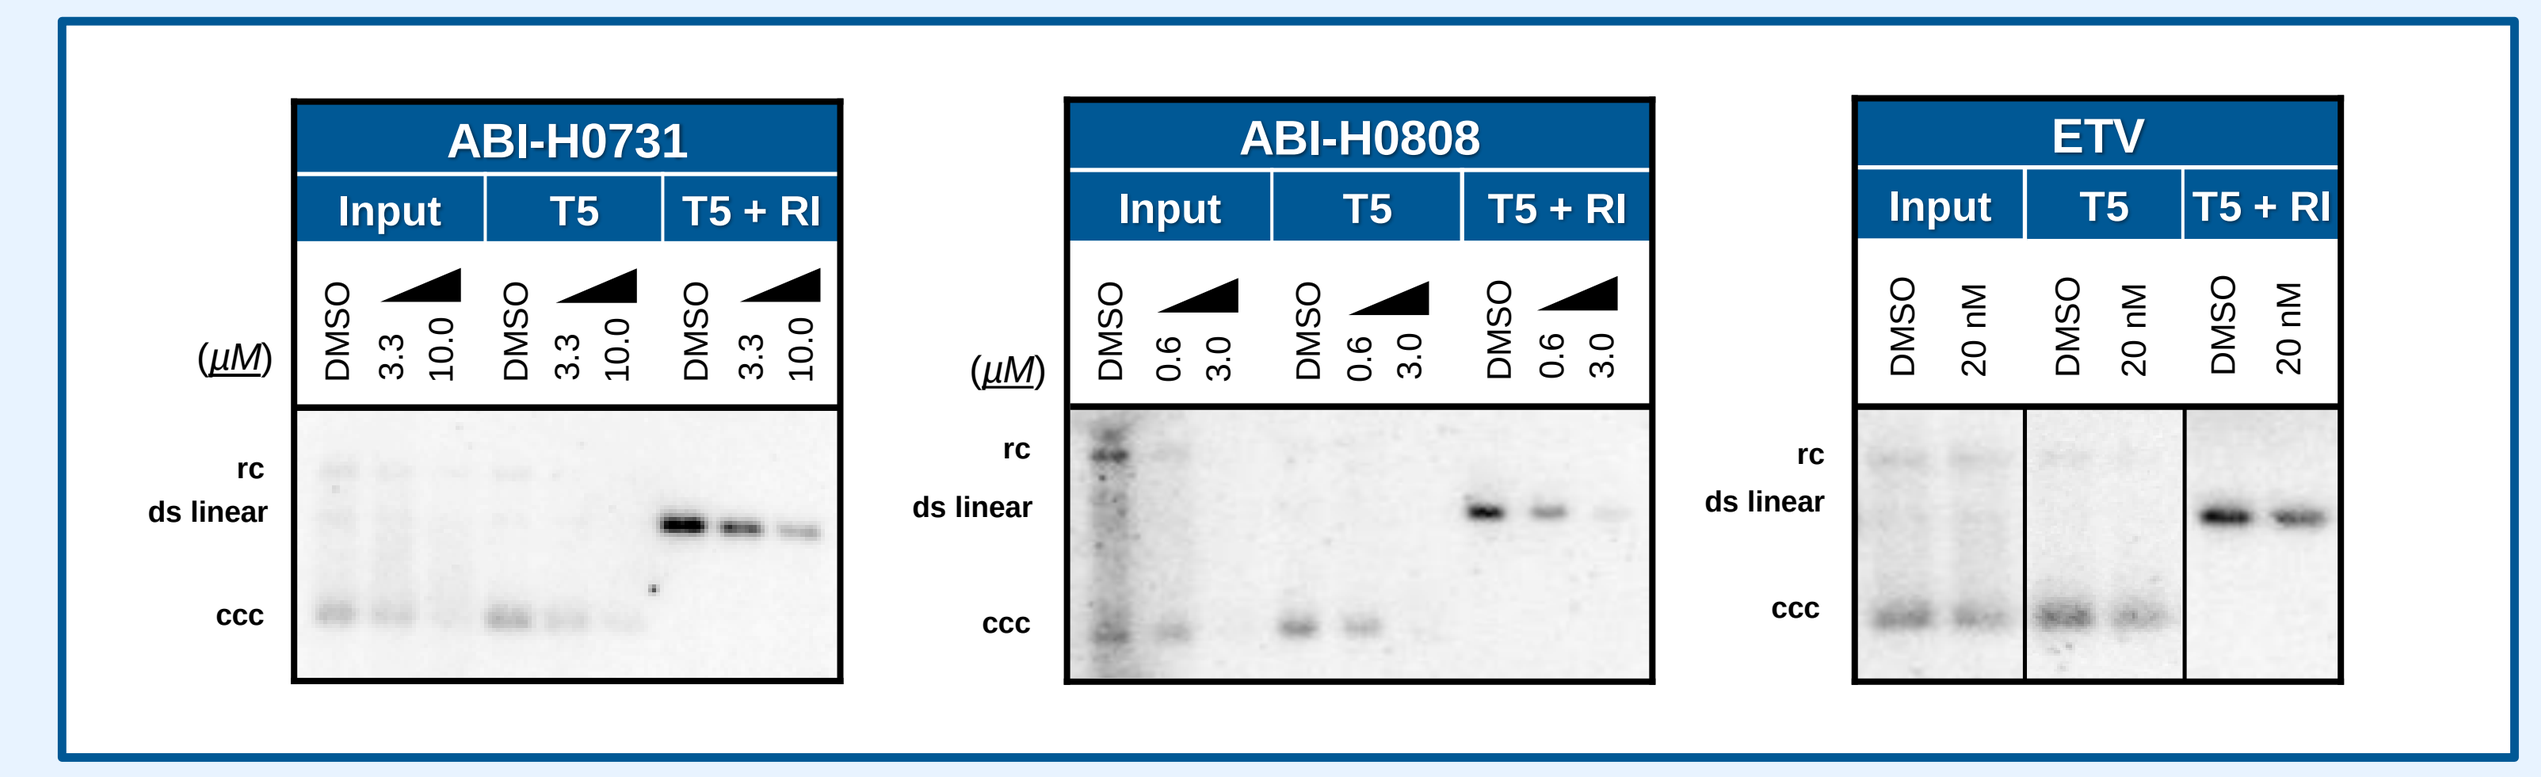
ABI-H0731
Input T5 T5 + RI
(µM)
DMSO 3.3 10.0
ABI-H0808
DMSO 0.6 3.0
ETV
DMSO 20 nM
rc
ds linear
ccc

- CpAMs reduced HBV cccDNA formation in both NTCP-HepG2 cells and PHH
- ETV (EC_{50} concentrations 125-200x) had minimal effect on cccDNA levels

CpAM Effect on cccDNA is Sustainable

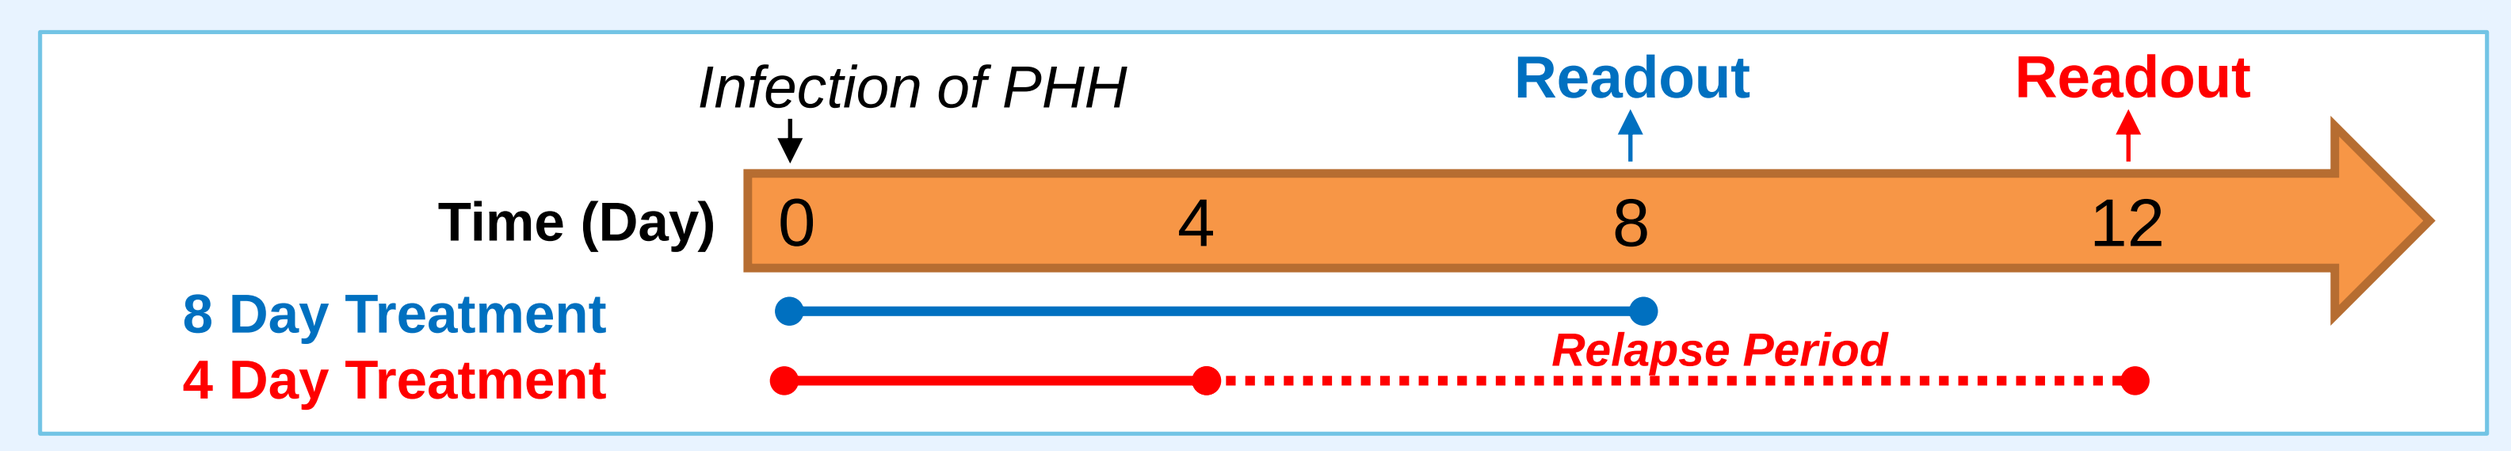
Infection of PHH
Readout
Readout
Time (Day)
0 4 8 12
8 Day Treatment
4 Day Treatment
Relapse Period

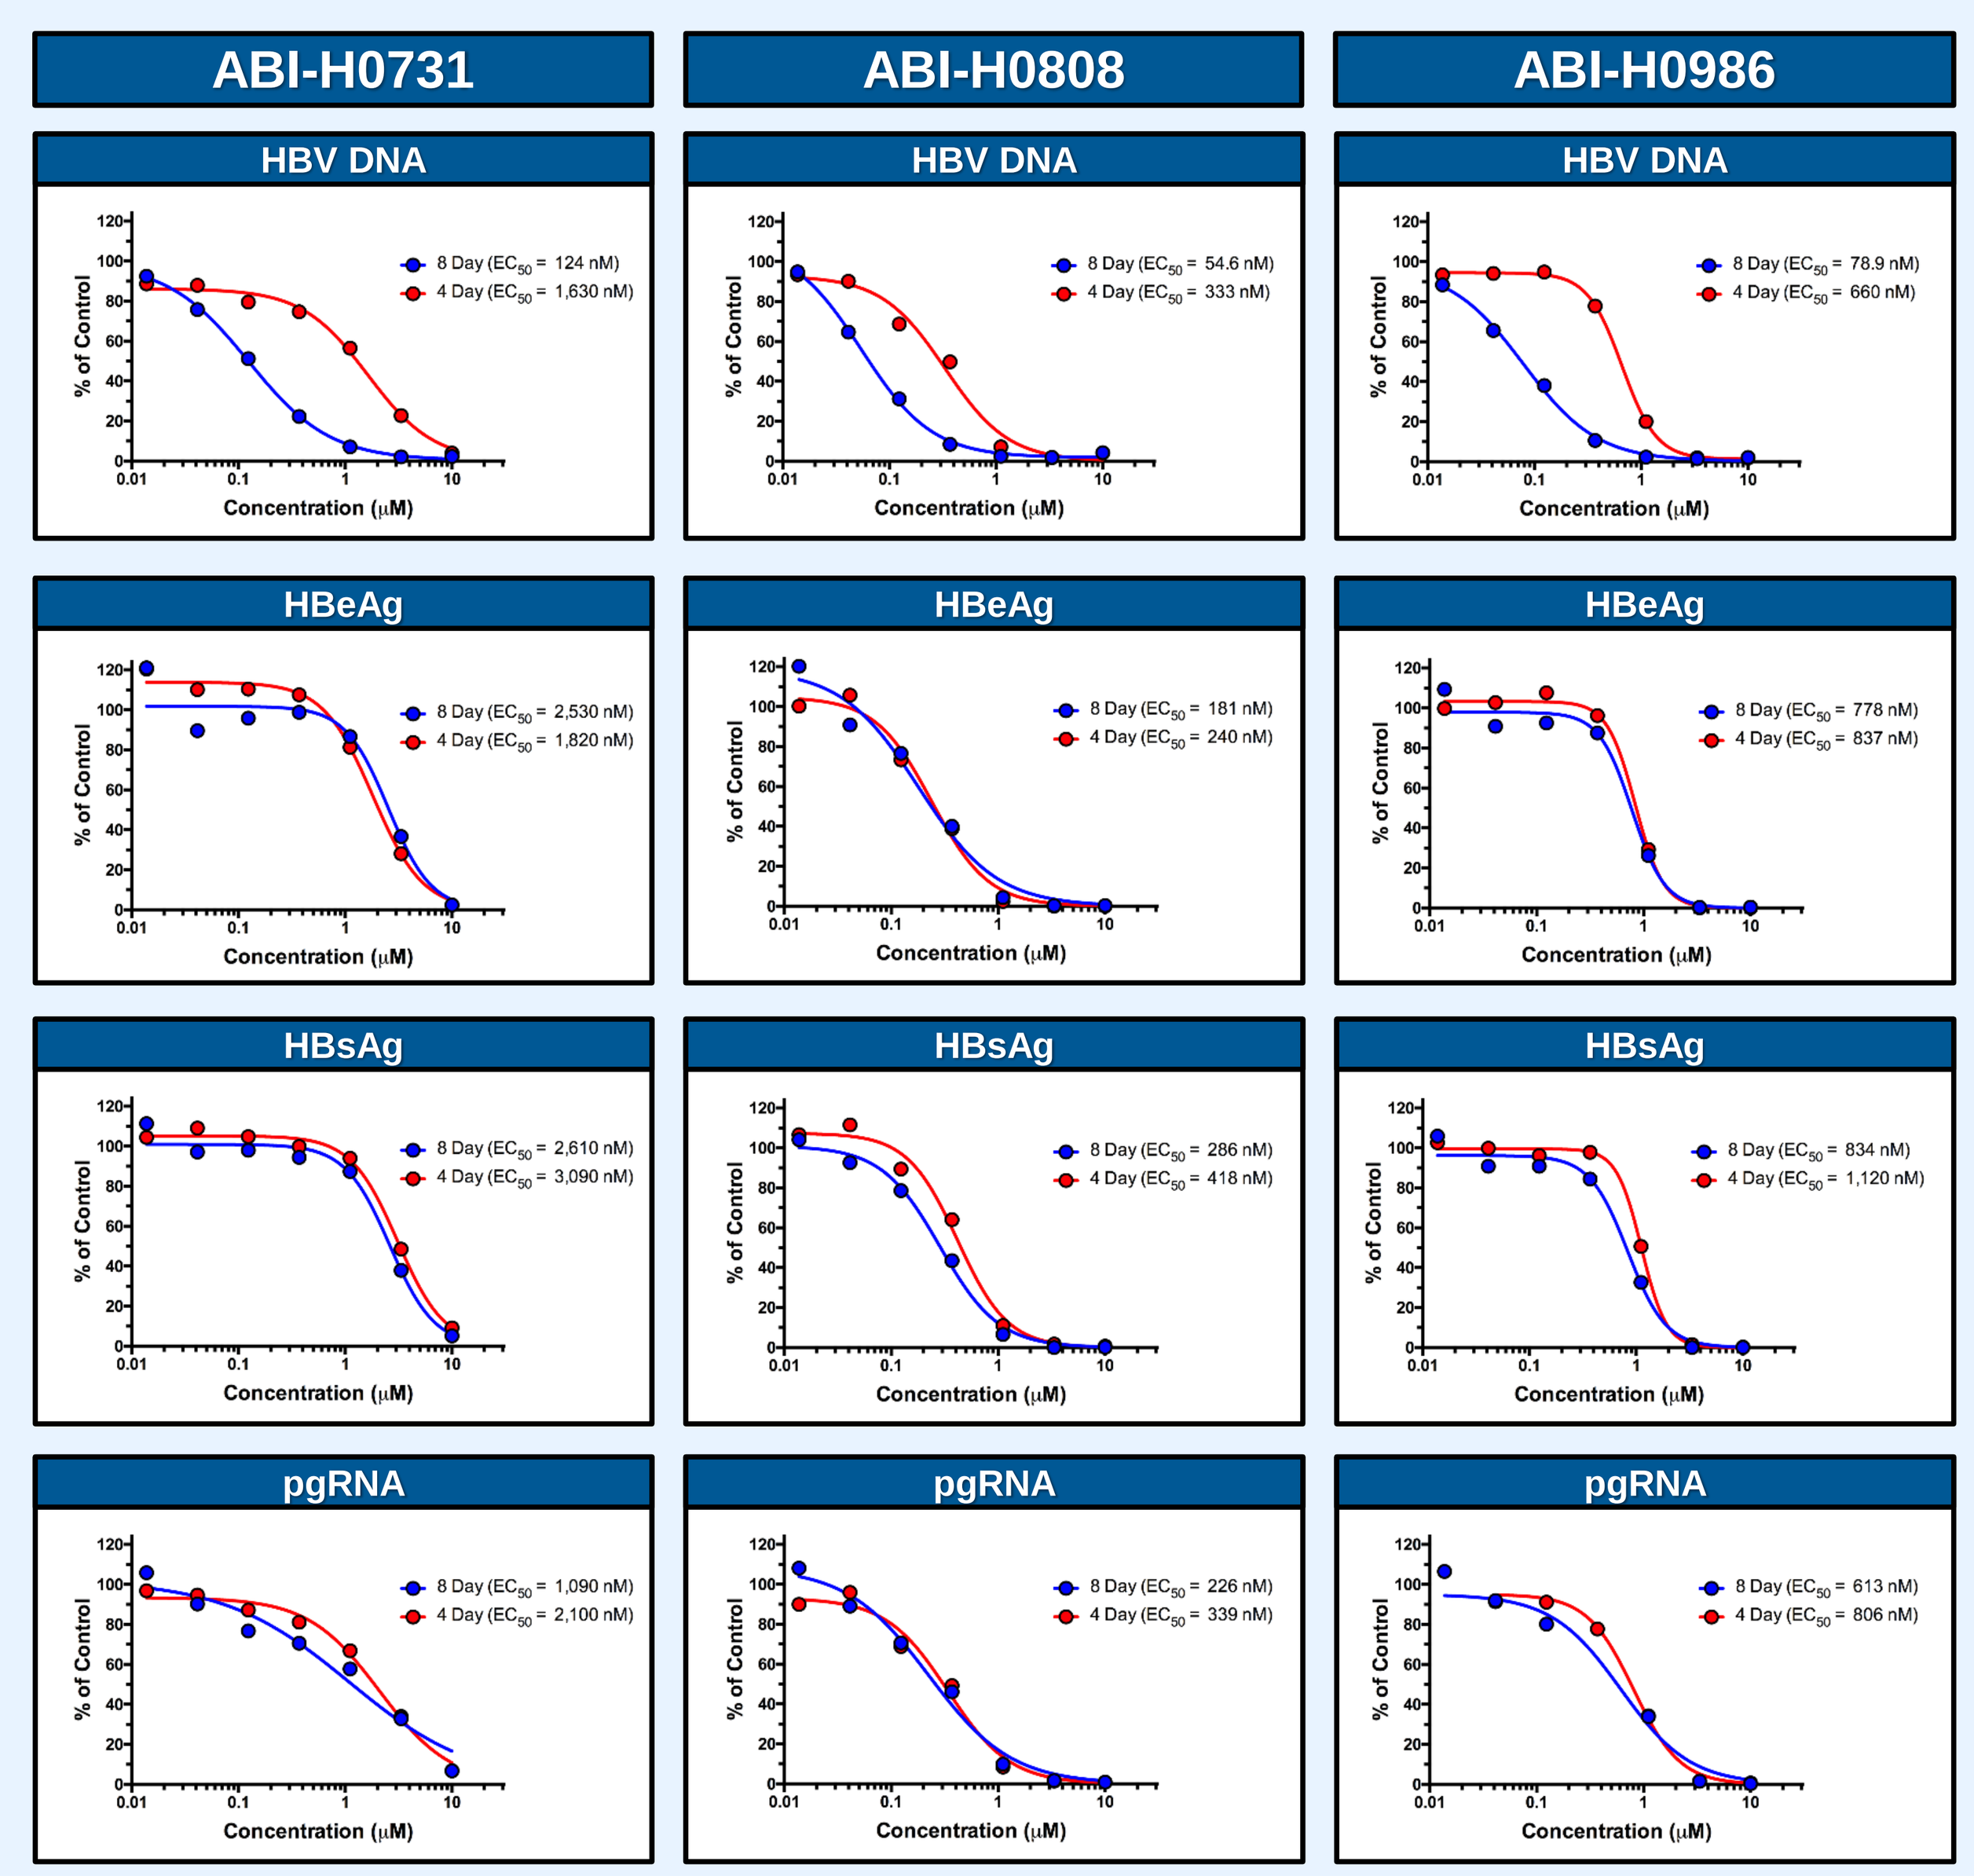
ABI-H0731
ABI-H0808
ABI-H0986
HBV DNA
HBeAg
HBsAg
pgRNA
8 Day (EC_{50} = 124 nM)
4 Day (EC_{50} = 1,630 nM)
8 Day (EC_{50} = 54.6 nM)
4 Day (EC_{50} = 333 nM)
8 Day (EC_{50} = 78.9 nM)
4 Day (EC_{50} = 660 nM)
8 Day (EC_{50} = 2,530 nM)
4 Day (EC_{50} = 1,820 nM)
8 Day (EC_{50} = 181 nM)
4 Day (EC_{50} = 240 nM)
8 Day (EC_{50} = 778 nM)
4 Day (EC_{50} = 837 nM)
8 Day (EC_{50} = 2,610 nM)
4 Day (EC_{50} = 3,090 nM)
8 Day (EC_{50} = 286 nM)
4 Day (EC_{50} = 418 nM)
8 Day (EC_{50} = 834 nM)
4 Day (EC_{50} = 1,120 nM)
8 Day (EC_{50} = 1,090 nM)
4 Day (EC_{50} = 2,100 nM)
8 Day (EC_{50} = 226 nM)
4 Day (EC_{50} = 339 nM)
8 Day (EC_{50} = 613 nM)
4 Day (EC_{50} = 806 nM)
% of Control
Concentration (µM)

- While CpAM removal induced a relapse in viral replication, the suppression of HBV antigens and pgRNA was sustained, indicating that early treatment with CpAMs block cccDNA formation

Summary

- Assembly Biosciences has discovered a proprietary series of selective and potent CpAMs
- CpAMs target core protein and interfere with multiple steps in the viral cycle
- CpAMs exhibit broad genotypic activity and retain potency against a Nuc resistant variant
- Treatment with CpAMs early after HBV infection inhibits the formation of cccDNA based on surrogate markers HBeAg, HBsAg and pgRNA levels, and direct cccDNA detection (Southern Blot)
- The suppression of HBV antigens and pgRNA expression is sustainable following CpAM withdrawal
- CpAMs represent a novel class of HBV antivirals and Assembly Biosciences' first candidate is progressing into clinical development

References

1. Bourne, C. et al. Small-molecule effectors of hepatitis B virus capsid assembly give insight into virus life cycle. J. Virol. (2008)
2. Stray, S. J. et al. A heteroaryldihydropyrimidine activates and can misdirect HBV capsid assembly. Proc. Natl. Acad. Sci. USA (2005)
3. Siegfried G, et al. Bromo-phenyl substituted thiazolyl dihydropyrimidines. US patent application publication no. US 2012/0282221 A1 (2012)
4. Wang et al. In vitro inhibition of HBV replication by a novel compound, GLS4, and its efficacy against adefovir- dipivoxil-resistant HBV mutations. Antivir. Ther. (2012)
5. Innaimo, S. F. et al. Identification of BMS-200475 as a Potent and Selective Inhibitor of Hepatitis B Virus. J. Antimicrob. Chemother. (1997)
6. Ladner, S. K. et al. Inducible expression of human hepatitis B virus (HBV) in stably transfected hepatoblastoma cells: a novel system for screening potential inhibitors of HBV replication. Antimicrob. Agents Chemoter. (1997)
7. Cai, D et al. Establishment of an Inducible HBV Stable Cell Line that Expresses cccDNA-dependent Epitope-tagged HBeAg for Screening of cccDNA Modulators. Antivir. Res. (2016)
8. Cai et al. A southern blot assay for detection of hepatitis B virus covalently closed circular DNA from cell cultures. Methods Mol. Bio. (2013)