CENTAUR, a Phase 2b Efficacy and Safety Study of Cenicriviroc in Adults with Non-alcoholic Steatohepatitis and Liver Fibrosis at Increased Risk of Progression to Cirrhosis

Scott Friedman[1], Arun Sanyal[2], Zachary Goodman[3], Eric Lefebvre[4], Mildred Gottwald[4], Laurent Fischer[4], Vlad Ratziu[5]

[1]Division of Liver Diseases, Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]Department of Gastroenterology, Virginia Commonwealth University, Richmond, VA, USA; [3]Center for Liver Diseases, Inova Fairfax Medical Campus, Falls Church, VA, USA; [4]Tobira Therapeutics, Inc., South San Francisco, CA, USA; [5]Hôpital Pitié Salpêtrière and Université Pierre et Marie Curie, Paris, France

Background and Aims

- Non-alcoholic fatty liver disease (NAFLD) is often associated with obesity, type 2 diabetes mellitus (T2DM), and metabolic syndrome (MetS).[1,2]
 - The advanced form of NAFLD, non-alcoholic steatohepatitis (NASH), is characterized by steatosis, hepatocellular ballooning, and lobular inflammation.[1,2]
 - NASH is often accompanied by liver fibrosis, which can progress to cirrhosis.[2]
 - Global NASH prevalence has substantially increased,[2,3] yet there are currently no approved treatments.
- C-C chemokine receptor types 2 and 5 (CCR2 and CCR5) are promising targets for treatment, as they mediate interactions that drive liver inflammation and fibrosis.[4]
 - In response to hepatocyte injury, resident liver macrophages (Kupffer cells) secrete C-C chemokine ligand type 2 (CCL2; MCP-1 [monocyte chemoattractant protein-1]), promoting recruitment and migration to the liver of pro-inflammatory monocytes, which mature into pro-inflammatory macrophages.[5,6]
 - Kupffer cells and macrophages express pro-inflammatory cytokines, which activate hepatic stellate cells (HSCs) and promote their survival, leading to collagen production or fibrogenesis.[7–10]
 - CCR2, CCR5, and their ligands, CCL2 and CCL5, mediate the intrahepatic immune-cell interactions that promote activation and migration of Kupffer cells and HSCs (**Figure 1**).[4]
- Cenicriviroc (CVC), an oral, potent, once-daily, dual CCR2/CCR5 antagonist, has demonstrated anti-inflammatory and antifibrotic effects in animal models[11,12] and reduced soluble CD14 levels, APRI (aspartate aminotransferase-to-platelet count ratio index) and FIB-4 (non-invasive hepatic fibrosis risk score) scores in HIV-positive subjects.[13] CVC demonstrated favorable tolerability in approximately 600 subjects, including those with mild or moderate hepatic impairment (Child–Pugh A and B).
- CVC is currently being evaluated in a study for the treatment of NASH in adult subjects with liver fibrosis (CENTAUR study 652-2-203; NCT02217475).
 - CVC is expected to prevent inflammation and fibrosis due to disruption of CCR2 and CCR5 signaling pathways.
 - o CCR2 inhibition would lead to decreased recruitment and migration of pro-inflammatory monocytes to the site of liver injury by activated Kupffer cells.[5,6]
 - o This consequent decrease in the number of pro-inflammatory monocytes and macrophages in the liver would thereby cause a reduction in chronic liver inflammation and downregulation of profibrotic cytokines.[5,6,9]
 - o Antagonism of CCR2 and CCR5 would also lead to reduced HSC migration, proliferation, and activation,[5,6,8,14] thereby inhibiting fibrogenesis.
- CENTAUR aims to evaluate the efficacy, pharmacokinetics (PK) and safety of CVC 150 mg for the treatment of NASH with liver fibrosis, for which CVC has received Fast Track designation by the US Food and Drug Administration (FDA).

Methods

Study design

- Phase 2b, randomized, double-blind, placebo-controlled, multinational study being conducted in 11 countries across the USA, Europe, and Asia-Pacific.
- Subjects were randomized 2:1:1 to CVC 150 mg once-daily (QD) for 2 years (Arm A), placebo QD for 1 year then CVC 150 mg QD for 1 year (Arm B), or placebo QD for 2 years (Arm C) (**Figure 2**).
 - At Baseline (Day 1), eligible subjects were assigned to the treatment arms using permuted block randomization stratified by NAFLD activity score (NAS) at Screening (4 or ≥5) and fibrosis stage (≤2 or >2).
 - Subjects will be instructed to take one tablet (CVC or matching placebo) every morning with food.
- Subjects with histological evidence of NASH, NAS ≥4, and liver fibrosis (Stages 1–3 NASH clinical research network [CRN] system) have been included in this study.
 - Other key inclusion and exclusion criteria are listed in **Table 1**.
- Disallowed medications include Vitamin E (>400 IU/day), pioglitazone, rosiglitazone, S-adenosylmethionine, pentoxifylline, and ursodiol, due to possible confounding effects on efficacy.

Study objectives

- **Primary objective:** Hepatic histological improvement in NAS at Year 1 relative to Screening biopsy (≥2-point improvement in NAS with at least a 1-point improvement in more than one category and with no concurrent worsening of fibrosis).
- **Key secondary objective:** Resolution of NASH (no hepatocellular ballooning; no more than minimal inflammation [<2 foci/200x]; any degree of steatosis) with no concurrent worsening of fibrosis at Year 2.

Outcome measures

- **Efficacy evaluation:** The efficacy endpoints for this study are shown in **Figure 3**.
- **PK evaluation:** Plasma samples will be collected on Day 1 (baseline) and Months 0.5, 3, 6, 12, 15, 18, and 24.
 - Population PK endpoints include characterization of the population PK of CVC in subjects with NASH, prediction of plasma CVC exposure throughout the study, and evaluation of the covariates that could impact CVC PK, including but not limited to age, sex, weight, body mass index (BMI), race, and fibrosis stage.
- **Safety evaluation:** Adverse events (AEs), clinical laboratory tests (for liver and fasting metabolic parameters), physical examination, vital signs, and 12-lead electrocardiogram will be evaluated over 2 years.

Figures and Tables

Figure 1. Inflammatory response to hepatocyte injury leading to fibrogenesis

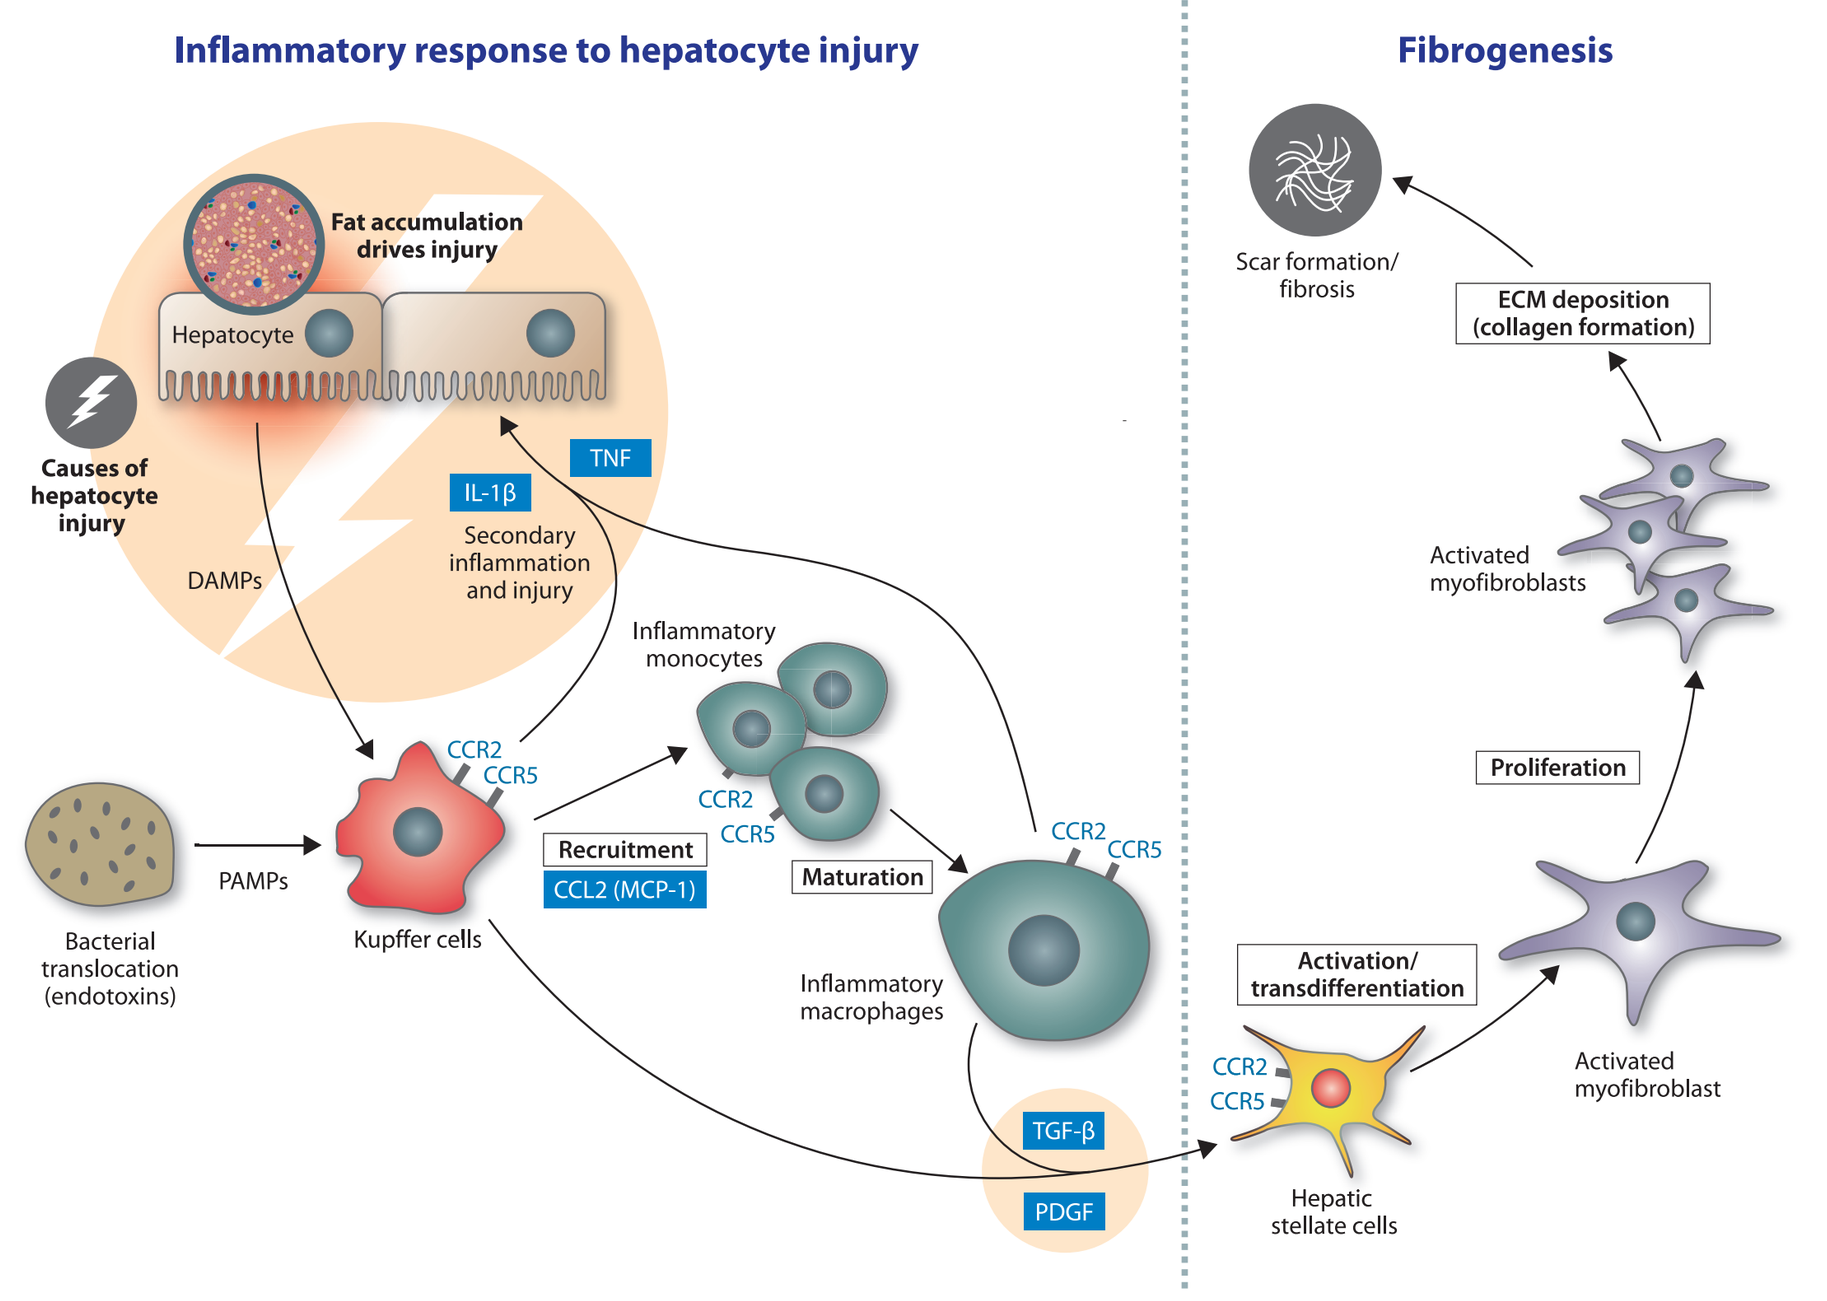
Inflammatory response to hepatocyte injury
Fibrogenesis
Fat accumulation drives injury
Hepatocyte
Causes of hepatocyte injury
DAMPs
IL-1β
TNF
Secondary inflammation and injury
Inflammatory monocytes
CCR2
CCR5
Recruitment
CCL2 (MCP-1)
Maturation
Inflammatory macrophages
Bacterial translocation (endotoxins)
PAMPs
Kupffer cells
TGF-β
PDGF
Scar formation/ fibrosis
ECM deposition (collagen formation)
Activated myofibroblasts
Proliferation
Activation/ transdifferentiation
Hepatic stellate cells
Activated myofibroblast

CCL2, C-C chemokine ligand type 2; CCR2, C-C chemokine receptor type 2; CCR5, C-C chemokine receptor type 5; DAMPs, danger-associated molecular patterns; ECM, extracellular matrix; IL-1β, interleukin 1β; MCP-1, monocyte chemoattractant protein-1; PAMPs, pathogen-associated molecular patterns; PDGF, platelet-derived growth factor; TGF-β, transforming growth factor β; TNF, tumor necrosis factor

Figure 2. Study design for CENTAUR

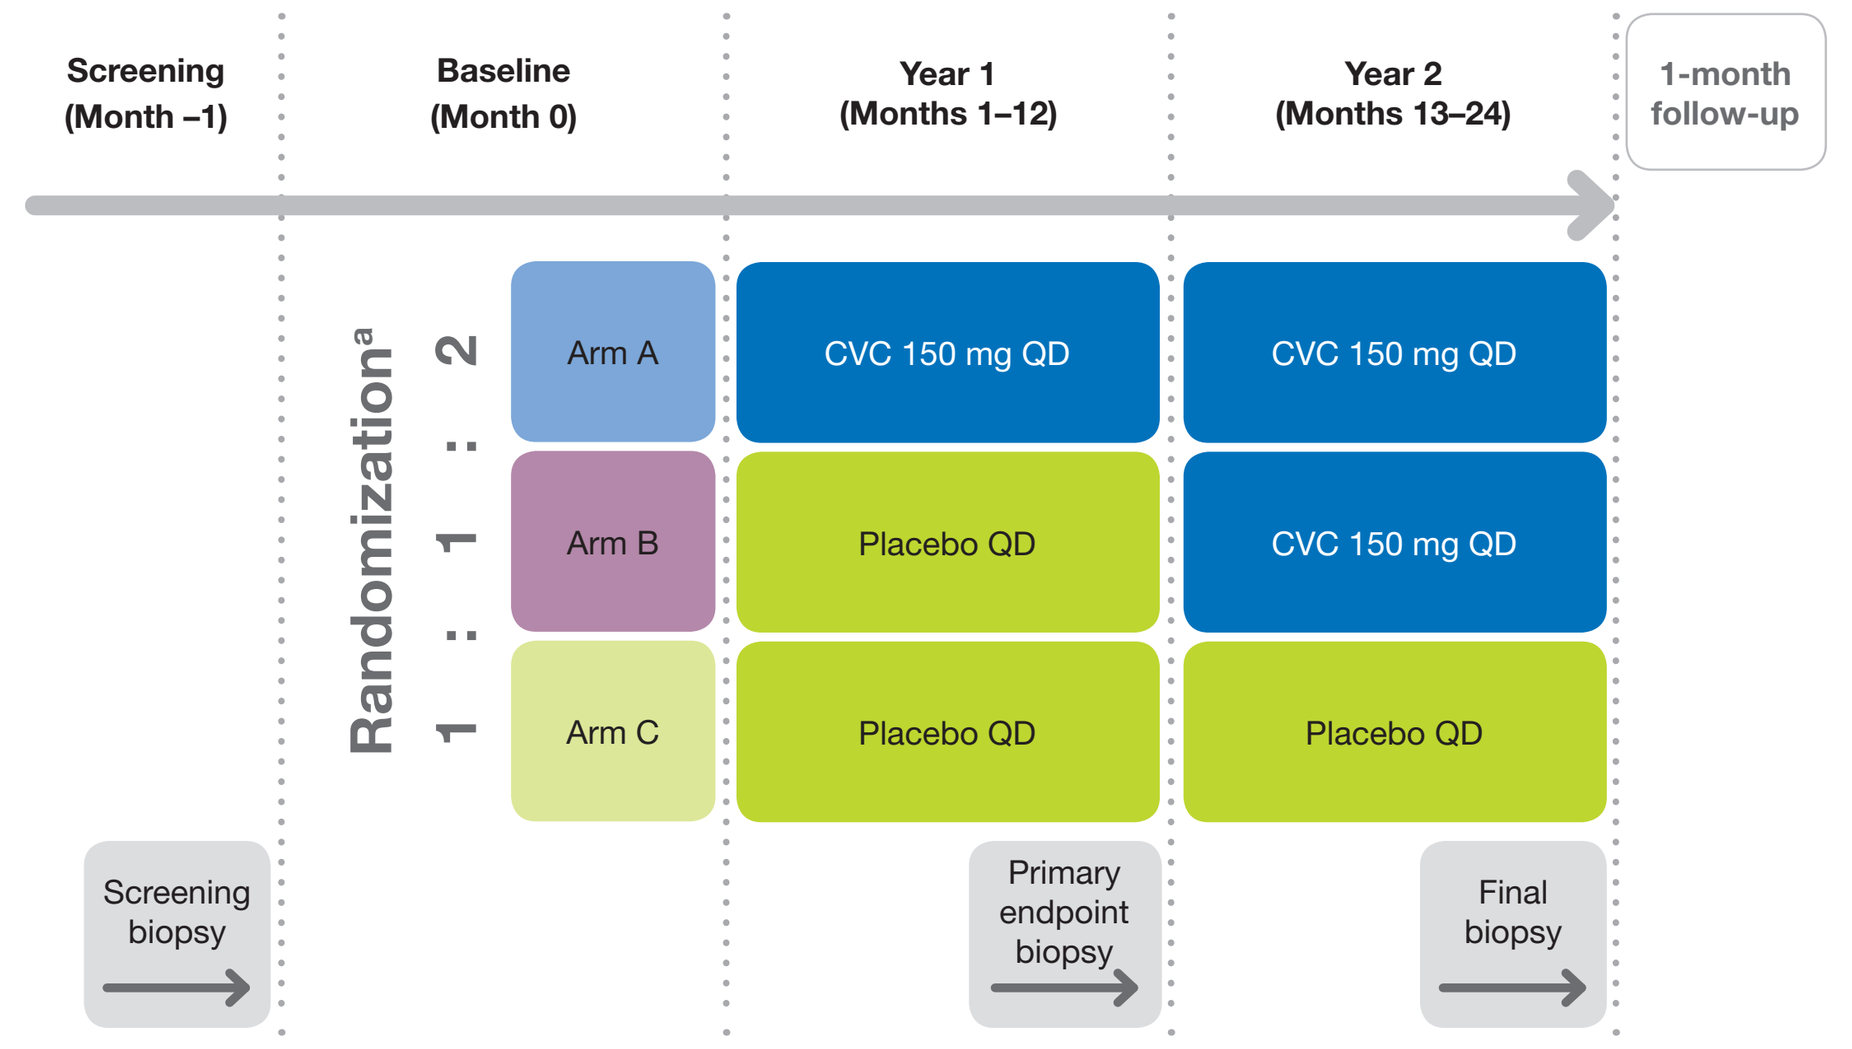
Screening (Month –1)
Baseline (Month 0)
Year 1 (Months 1–12)
Year 2 (Months 13–24)
1-month follow-up
Randomization[a]
2 : 1 : 1
Arm A
CVC 150 mg QD
CVC 150 mg QD
Arm B
Placebo QD
CVC 150 mg QD
Arm C
Placebo QD
Placebo QD
Screening biopsy
Primary endpoint biopsy
Final biopsy

[a]Stratification by NAS at Screening (4 or ≥5) and fibrosis stage (≤2 or >2)
CVC, cenicriviroc; NAS, non-alcoholic fatty liver disease activity score; QD, once daily

Figure 3. Efficacy endpoints for CENTAUR

Primary efficacy endpoint	Key secondary efficacy endpoint
Hepatic histological improvement in NAS at **Year 1** relative to Screening biopsy (≥2-point drop in NAS with ≥1-point improvement in ≥1 category and with no concurrent worsening of fibrosis) (Arm A versus Arms B and C)	Resolution of NASH at **Year 2** (no hepatocellular ballooning; no more than minimal inflammation [<2 foci/200x]; any degree of steatosis) with no concurrent worsening of fibrosis (Arm A versus Arm C)

Other secondary efficacy endpoints
Resolution of NASH with no concurrent worsening of fibrosis at Year 1 (Arm A versus Arms B and C)
Hepatic histological improvement in NAS at Year 2 relative to Screening biopsy (≥2-point drop in NAS with at least a 1-point improvement in more than 1 category and with no concurrent worsening of fibrosis) (Arm A versus Arm C)
Change in histologic fibrosis stage assessed using NASH CRN and Ishak scoring systems at Years 1 and 2
Change in collagen morphometry (CPA) on liver biopsy to evaluate efficacy of CVC versus placebo at Years 1 and 2
Change in hepatic stellate cell activation marker (α-SMA) at Years 1 and 2
Change from Baseline in non-invasive scores and markers of hepatic fibrosis (APRI, FIB-4, hyaluronic acid, FibroTest [FibroSure], NFS, and ELF at Months 6, 12, 18, and 24
Change from Baseline in biomarkers of hepatocyte apoptosis, assessed using CK-18 caspase-cleaved and total at Years 1 and 2

Tertiary efficacy endpoint
Change from Baseline in non-invasive liver-imaging method (e.g. TE, MRE, and ARFI) at Months 6, 12, 18, and 24 (at sites where available)

α-SMA, α-smooth muscle actin; APRI, aspartate aminotransferase-to-platelet count ratio index; ARFI, acoustic radiation force impulse; CK-18, cytokeratin 18; CPA, collagen proportionate area; CRN, clinical research network; CVC, cenicriviroc; ELF, enhanced liver fibrosis test; FIB-4, non-invasive hepatic fibrosis risk score; MRE, 2-dimensional magnetic resonance elastography; NAS, non-alcoholic fatty liver disease activity score; NASH, non-alcoholic steatohepatitis; NFS, non-alcoholic fatty liver disease fibrosis score; TE, ultrasound transient elastography

Table 1. Key inclusion and exclusion criteria for the CENTAUR study

Inclusion criteria	Description
Sex and age	Males and females; 18–75 years of age
Histological evidence of NASH	Based on screening biopsy, with a NAS of ≥4 (at least 1 point in each component)
Histological evidence of liver fibrosis	Based on screening biopsy, defined as NASH CRN system Stage 1–3 inclusive
Increased risk of disease progression	Meeting any of the following three criteria: • documented evidence of T2DM • high BMI (>25 kg/m²) with at least one criteria of MetS, as defined by the NCEP – Central obesity: waist circumference ≥40" (male), ≥35" (female) – Dyslipidemia: TG ≥150 mg/dL – Dyslipidemia: HDL-c <40 mg/dL (male), <50 mg/dL (female) – Blood pressure ≥130/85 mmHg (or treated for hypertension) – Fasting plasma glucose ≥110 mg/dL • bridging fibrosis (NASH CRN Stage 3) and/or definite NASH (NAS ≥5)
Exclusion criteria	**Description**
Liver co-morbidities	• History of cirrhosis and/or hepatic decompensation, including ascites, hepatic encephalopathy, or variceal bleeding • HBsAg positive • HCVAb positive, with two exceptions if all other eligibility criteria are met – Subjects previously treated for HCV with at least a 1-year period since documented sustained virologic response at Week 12 (post treatment) – Subjects with the presence of HCVAb but negative HCV RNA without treatment (i.e. spontaneous clearance) • Other known causes of chronic liver disease, including alcoholic liver disease • Prior or planned liver transplantation

BMI, body mass index; CRN, clinical research network; HBsAg, hepatitis B surface antigen; HCV, hepatitis C virus; HCVAb, hepatitis C virus antibody; HDL-c, high-density lipoprotein-cholesterol; MetS, metabolic syndrome, NAS, non-alcoholic fatty liver disease activity score; NASH, non-alcoholic steatohepatitis; NCEP, National Cholesterol Education Program; T2DM, type 2 diabetes mellitus; TG, triglyceride

Table 2. Demographics and baseline characteristics of subjects enrolled in the CENTAUR study

Demographics	
Mean age, years (±SD) Sex, % m : % f Mean BMI, kg/m² (±SD)	54.1 (10.59) 47.4 : 52.6 33.86 (6.49)
Criteria of MetS, N (%)	
Waist circumference ≥40" (m), ≥35" (f) TG ≥150 mg/dL HDL-c <40 mg/dL (m), <50 mg/dL (f) Blood pressure ≥130/85 mmHg Fasting plasma glucose ≥110 mg/dL	247 (85.5) 142 (49.1) 188 (65.1) 146 (50.5) 140 (48.4)
Subjects meeting ≥3 MetS criteria	194 (67.1)
Insulin resistance parameters, mean (±SD)	
HbA1c, % HOMA-IR Adipo-IR	6.54 (1.27) 8.67 (9.99) 15.42 (11.16)
Most common concomitant medications, N (%)	
Biguanides (metformin) Proton pump inhibitors (acid reducers) HMG CoA reductase inhibitors (statins)	123 (42.6) 114 (39.4) 92 (31.8)

Adipo-IR, adipose tissue insulin resistance; ALT, alanine aminotransferase; BMI, body mass index; f, female; HbA1c, hemoglobin A1c; HDL-c, high-density lipoprotein-cholesterol; HOMA-IR, homeostatic model assessment of insulin resistance; m, male; MetS, metabolic syndrome; SD, standard deviation; TG, triglyceride

Figure 4. Liver histology characteristics of subjects enrolled in CENTAUR (stratification factors)

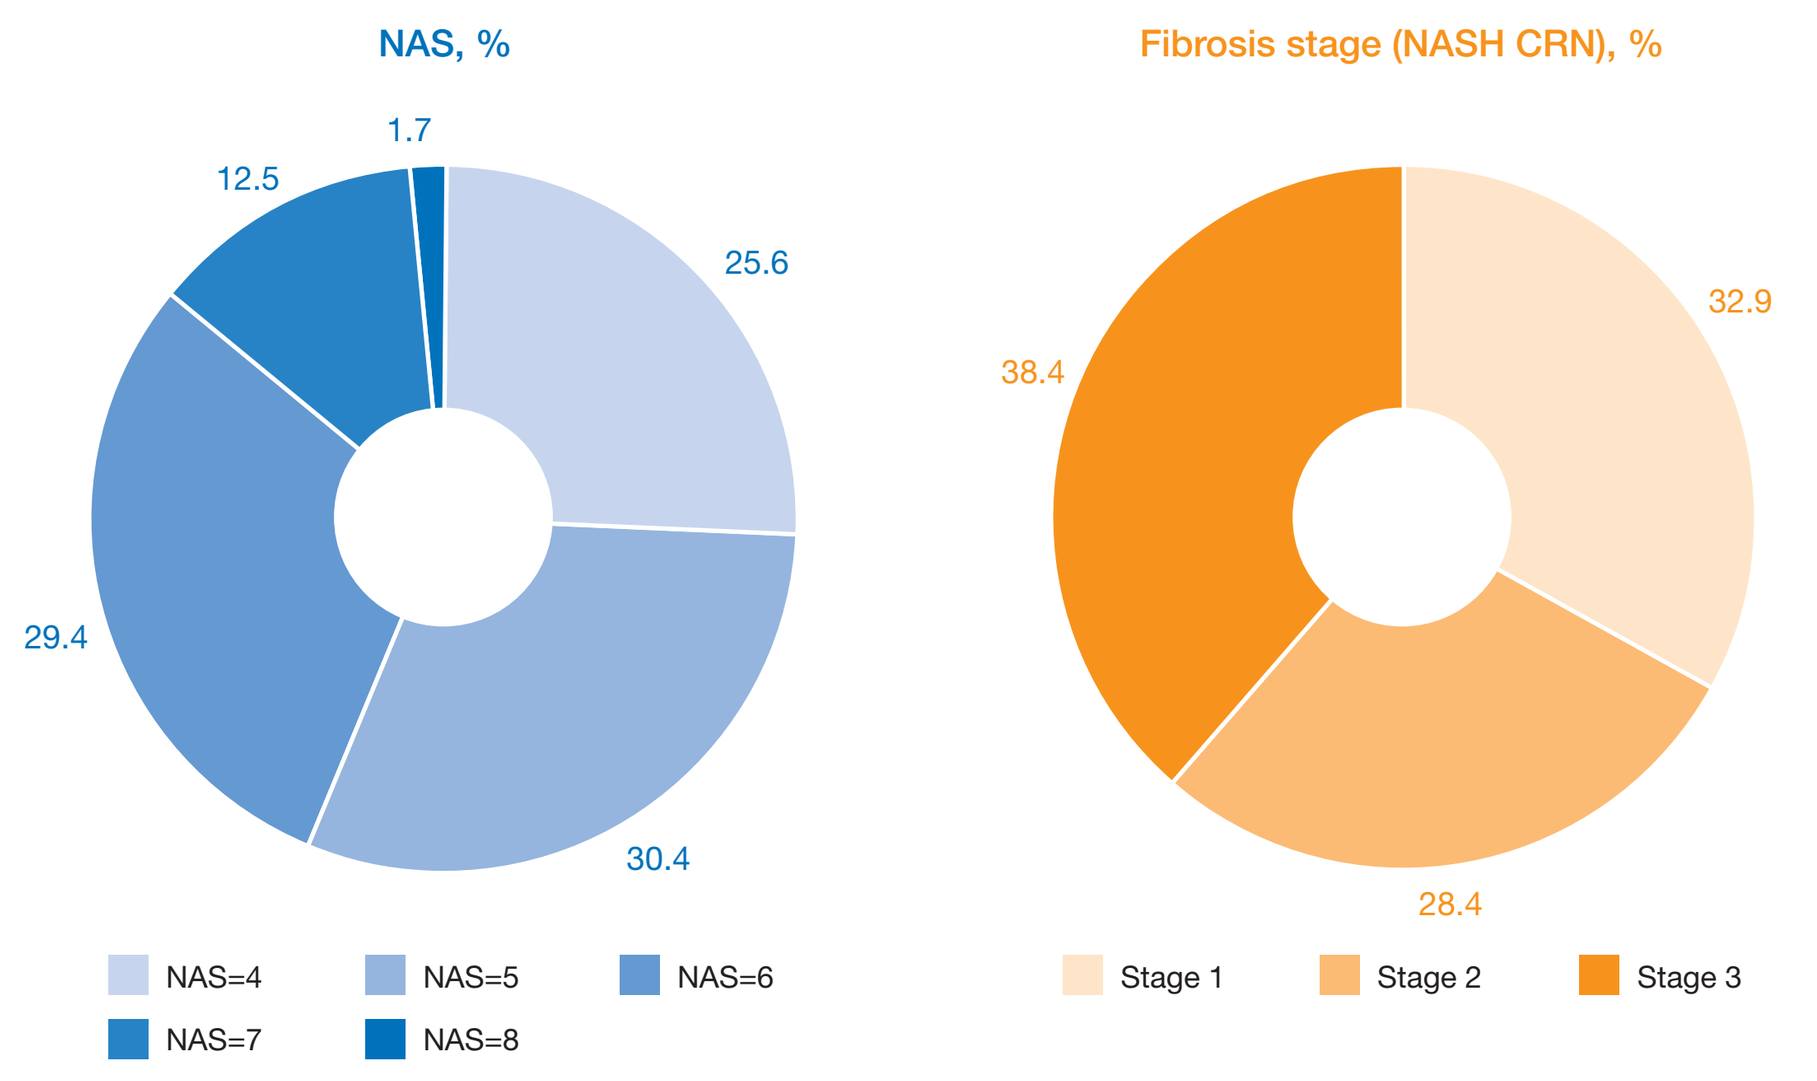
NAS, %
1.7
12.5
25.6
29.4
30.4
NAS=4
NAS=5
NAS=6
NAS=7
NAS=8
Fibrosis stage (NASH CRN), %
38.4
32.9
28.4
Stage 1
Stage 2
Stage 3

CRN, clinical research network; NAS, non-alcoholic fatty liver disease activity score; NASH, non-alcoholic steatohepatitis

Statistical Methods

- **Efficacy evaluation:** The primary (Arm A versus Arms B and C at Year 1) and key secondary (Arm A versus Arm C at Year 2) efficacy endpoints will be analyzed using the Cochran-Mantel-Haenszel test. Descriptive statistics and two-sided 95% confidence intervals will be used for secondary efficacy endpoints.
- **PK evaluation:** Measured plasma CVC concentrations will be used to conduct non-linear mixed-effects modeling of population PK.
- **Safety evaluation:** The occurrence of treatment-emergent AEs will be summarized by treatment group using MedDRA (Medical Dictionary for Regulatory Activities) preferred terms, system–organ classifications, and severity.
 - Clinical laboratory tests will be presented as descriptive summaries by study visit, and laboratory abnormalities will be graded according to NCI CTCAE (National Cancer Institute's Common Terminology Criteria for Adverse Events) version 4.03 and summarized by treatment group and severity grade.

Results

Enrollment of subjects

- A total of 812 subjects were screened and 289 were enrolled in 11 countries (166 in the USA, 94 in the European Union, 29 in the Asia-Pacific region).
- The demographics and baseline characteristics of enrolled subjects are summarized in **Table 2**.
 - The mean age of enrolled subjects is 54.1 years (standard deviation [SD] 10.59), mean BMI is 33.86 kg/m² (SD 6.49), and 52.6% are female.
 - Most subjects (88.6%) have BMI >25 kg/m² with ≥1 MetS criteria. The majority of subjects have T2DM (52.9%), and the mean hemoglobin A1c (HbA1c) of diabetic subjects is 7.21% (SD 1.27).
 - The mean homeostatic model assesments of insulin resistance (HOMA-IR) and adipose tissue insulin resistance (Adipo-IR) of all subjects are 8.67 (SD 9.99) and 15.42 (SD 11.16), respectively.

Liver histology characteristics

- The results of screening liver biopsies of enrolled subjects are summarized in **Figure 4**.
 - Subjects had a total mean NAS of 5.3 (SD 1.05) with mean scores of 1.5 (SD 0.50) for hepatocellular ballooning, 1.4 (SD 0.57) for steatosis and 2.4 (SD 0.57) for lobular inflammation.
 - The majority of subjects have NAS ≥5 (74.0%) and moderate-to-bridging fibrosis, i.e. NASH CRN Stages 2–3 (66.8%).

Discussion

- CENTAUR is the first prospective study evaluating an oral agent to exclusively enroll subjects with NASH and liver fibrosis at increased risk of progression to cirrhosis.
 - Fibrosis stage has been shown to be associated with long-term overall mortality, liver transplantation, and liver-related events. Therefore, subjects with liver fibrosis are likely to benefit most from treatment in order to achieve resolution of NASH and prevention of fibrosis progression.[15]
 - Importantly, inclusion of a high-risk population with NASH and fibrosis is also expected to reduce the likelihood of spontaneous improvement, allowing detection of a greater difference between CVC treatment and placebo.
- The key endpoints of this study are in line with those discussed in a joint meeting between the American Association for the Study of Liver Diseases and the US Food and Drug Administration in 2011, and those used in the PIVENS and FLINT studies.[16–18]
 - The key secondary endpoint, 'resolution of NASH with no concurrent worsening of fibrosis', is potentially an acceptable surrogate endpoint for FDA accelerated approval under subpart H.[19]
- CENTAUR will allow comparison of the benefits of shorter (1-year) versus longer (2-year) treatment with CVC and assessment of correlations between decreased inflammation and fibrosis.

Acknowledgment: Editorial assistance was provided by Rebekah Chan PhD, Alpharmaxim Healthcare Communications.

Disclosures: Scott Friedman is a consultant to Tobira Therapeutics, Inc., has stock options from the company and has previously received research support for animal studies of cenicriviroc; Zachary Goodman has received research support from Tobira Therapeutics, Inc., Gilead, Galectin, Intercept, Conatus, Cempra and Alexion; Eric Lefebvre, Mildred Gottwald and Laurent Fischer are employees and stockholders of Tobira Therapeutics, Inc.; Vlad Ratziu has received consultancy fees from Tobira Therapeutics, Inc., Intercept, Genfit, Galmed, Boehringer Ingelheim, Zafgen, Abbott, Isis, AstraZeneca, Pfizer and Novartis. Arun Sanyal has no conflicts of interest to disclose.

Study funded by Tobira Therapeutics, Inc.

References: **1.** Chalasani N, *et al*. *Am J Gastroenterol* 2012;107(6):811–826; **2.** World Gastroenterology Organisation (WGO). WGO Global Guidelines. Nonalcoholic Fatty Liver Disease and Nonalcoholic Steatohepatitis. 2012. http://www.worldgastroenterology.org/UserFiles/file/guidelines/nafld-nash-english-2012.pdf. Accessed 11 November 2015; **3.** Vernon G, *et al*. *Aliment Pharmacol Ther* 2011;34(3):274–285; **4.** Saiman Y, Friedman SL. *Front Physiol* 2012;3:213; **5.** Karlmark KR, *et al*. *Hepatology* 2009;50(1):261–274; **6.** Miura K, *et al*. *Am J Physiol Gastrointest Liver Physiol* 2012;302(11):G1310–G1321; **7.** Rivera CA, *et al*. *Am J Physiol Gastrointest Liver Physiol* 2001;281(1):G200–G207; **8.** Duffield JS, *et al*. *J Clin Invest* 2005;115(1):56–65; **9.** De Minicis S, *et al*. *Gastroenterology* 2007;132(5):1937–1946; **10.** Pradere JP, *et al*. *Hepatology* 2013;58(4):1461–1473; **11.** Lefebvre E, *et al*. *Hepatology* 2013;58(S1):221A–222A. Abstract 30; **12.** Hong F, *et al*. *Hepatology* 2013;58(6 Suppl):1381A–1382A. Abstract LB-7; **13.** Thompson M, *et al*. *Hepatology* 2014;60(Suppl 1):424A. Abstract 455; **14.** Schwabe RF, *et al*. *Am J Physiol Gastrointest Liver Physiol* 2003;285(5):G949–G958; **15.** Angulo P, *et al*. *Gastroenterology* 2015;149(2):389–397; **16.** Sanyal AJ, *et al*. *Hepatology* 2011;54(1):344–353; **17.** Sanyal AJ, *et al*. *N Engl J Med* 2010;362(18):1675–1685; **18.** Neuschwander-Tetri BA, *et al*. *Lancet* 2015;385(9972):956–965; **19.** Sanyal AJ, *et al*. *Hepatology* 2015;61(4):1392–1405

Poster 95 presented at HEP DART 2015, Frontiers in Drug Development for Viral Hepatitis, December 6–10 2015, Grand Wailea, HI, USA