Effectiveness of 8-week glecaprevir/pibrentasvir (G/P) for treatment naïve, non-cirrhotic patients with HCV infection in the TRIO Health network

STEVEN L. FLAMM[1], JENS KORT[2], STEVE MARX[2], JOHN STREZEWSKI[2], BRUCE BACON[3], MICHAEL P. CURRY[4], NAOKY TSAI[5], NICOLE WICK[6], ZOBAIR YOUNOSSI[7] AND NEZAM AFDHAL[4]

[1]Northwestern University Feinberg School of Medicine, Chicago Illinois, USA; [2]AbbVie; [3]Saint Louis University School of Medicine, St. Louis, Missouri, USA; [4]Beth Israel Deaconess Medical Center, Boston Massachusetts, USA; [5]University of Hawaii, Honolulu, Hawaii, USA; [6]Trio Health Analytics, La Jolla, California, USA; [7]Center for Liver Diseases, Department of Medicine, Inova Fairfax Hospital, Falls Church, Virginia, USA

1. BACKGROUND AND AIMS

Glecaprevir/Pibrentasvir (G/P) is the only regimen approved by the US-FDA with 8 week treatment duration for chronic HCV genotype (GT) 1-6 infection in treatment naïve (TN), non-cirrhotic (NC) patients. Real world effectiveness data from the US are lacking and the aim of this study was to close this gap.

2. METHODS

Data from 560 treatment naïve, non-cirrhotic patients who initiated 8-weeks of G/P treatment between August 2017 and April 2018 were collected electronically from providers and specialty pharmacies* of the Trio Health network and analyzed. The primary outcome assessed was Per Protocol (PP) sustained virologic response at 12 weeks post treatment (SVR12). For patient disposition, lost-to-follow-up is defined as patients who were unreachable either during or after completion of treatment.

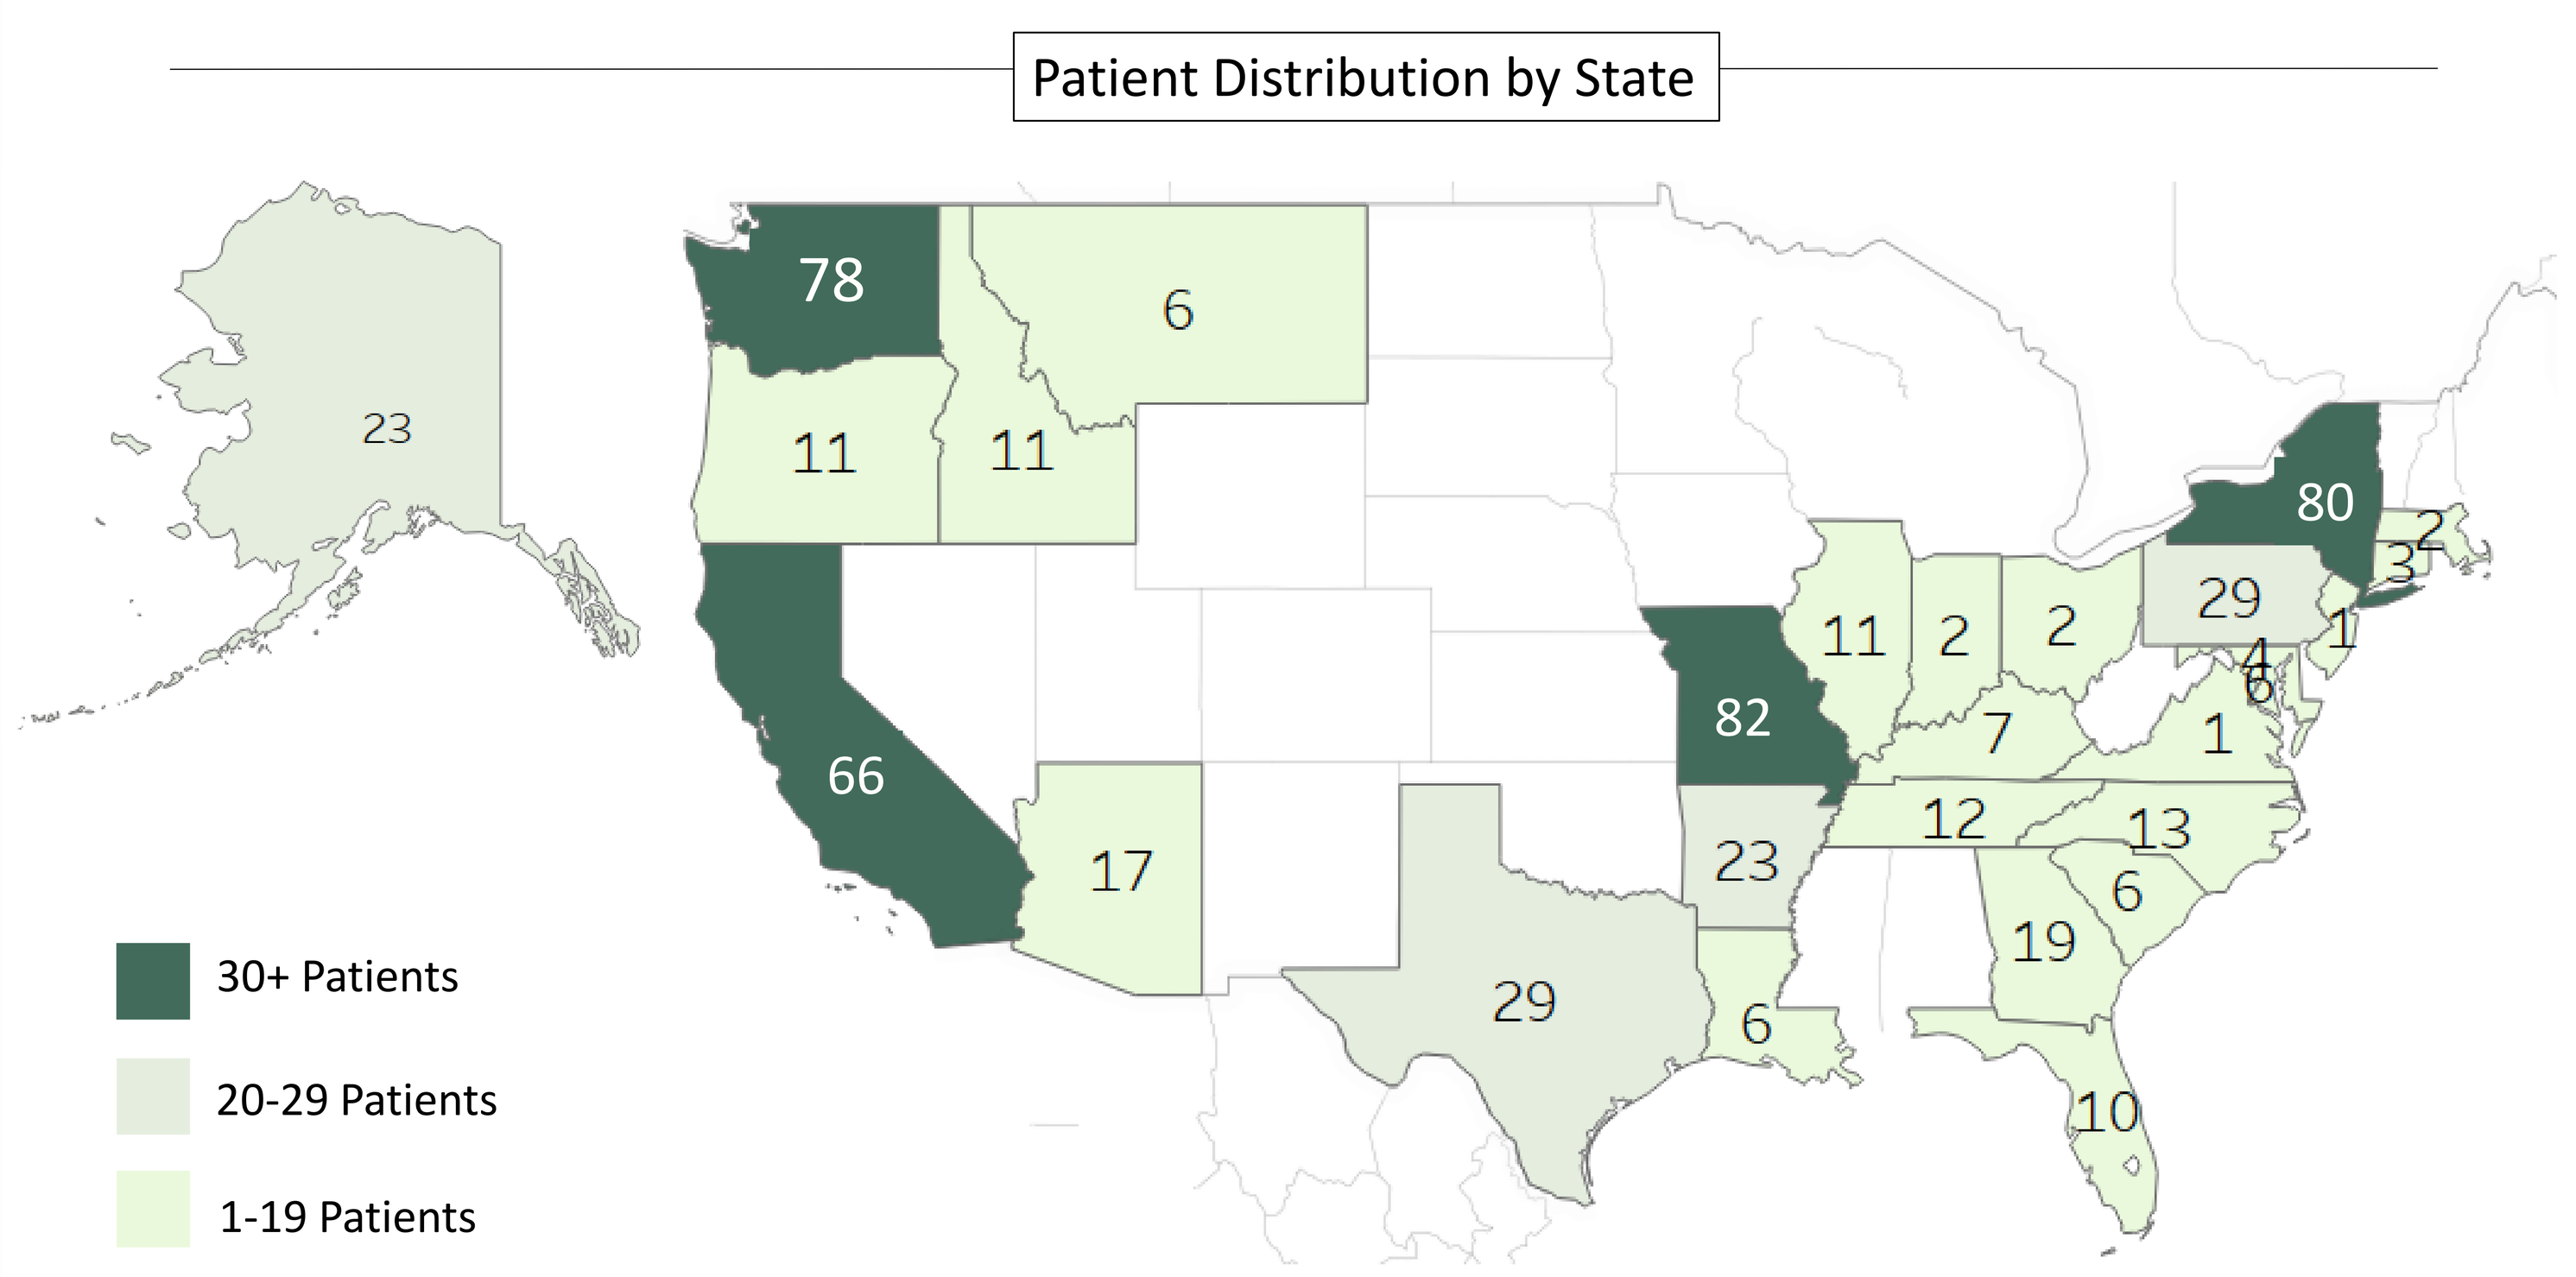
Patient Distribution by State
30+ Patients
20-29 Patients
1-19 Patients

*Specialty Pharmacy Partners include Premier, Quality, AllCare, TFP Wellness Systems, Grubbs NW Specialty Pharmacy, and Value Specialty Pharmacy, LLC

3. PATIENT DISPOSITION

Initiated 8 Week G/P Treatment Naive, Non-Cirrhotic n=560

- Discontinued n=7
- Lost-to-Follow-Up n=13
- SVR12 Available n=540
 - SVR12 Achieved n=537
 - SVR12 Not Achieved n=3

SVR12 PP = 99% (537/540)
SVR12 ITT = 96% (537/560)

4. PATIENT CHARACTERISTICS

Metric – no. (%) unless indicated	8 week (n=560)
Community Practice Type	476/560 (85%)
Payer Type	
Commercial	107/560 (19%)
Medicaid	247/560 (44%)
Medicare	196/560 (35%)
Male	304/560 (54%)
BMI - mean (range)	29 (15.1-68) n=503
Age – mean (range)	52 (18-87) n=560
Ethnicity	
Asian	7/560 (1%)
Black	38/560 (7%)
Hispanic or Latino	22/560 (4%)
White	168/560 (30%)
Other or Unspecified	325/560 (58%)
Concurrent PPI Use	61/560 (11%)
Liver Fibrosis[1]	
F0-2	496/560 (89%)
F3	64/560 (11%)
Chronic Kidney Disease	
No CKD	360/540 (67%)
CKD Stage 1-3	151/540 (28%)
CKD Stage 4-5	29/540 (5%)
Comorbidities	
Anxiety	98/560 (18%)
Depression	111/560 (20%)
Diabetes	55/560 (10%)
Hypertension	179/560 (32%)
Hyperlipidemia	38/560 (7%)
HIV Co-Infection	7/560 (1%)
Baseline Labs	
ALT U/L - mean (SD)	50 (37) n=560
AST U/L - mean (SD)	38 (19) n=560
FIB-4 <1.45	365/560 (65%)
FIB-4 1.45 to 3.25	195/560 (35%)
Baseline VL <800K IU/ml	226/559 (40%)
Baseline VL 800K to 6MM	221/559 (40%)
Baseline VL >6MM IU/ml	112/559 (20%)
HCV Genotype	
1A	300/560 (54%)
1B	83/560 (15%)
1 (subtype mixed or unknown)	14/560 (3%)
2	94/560 (17%)
3	57/560 (10%)
4-6[2]	12/560 (2%)

[1]FIbrosis as reported by the physician, compensated/decompensated not known; [2]No patients had GT5

5. PP & ITT SVR12 RATES BY GENOTYPE

PP SVR12 rates were 99% (GT1) or 100% (GT2, 3, 4-6). ITT SVR12 rates by HCV genotype(s) were 95% and higher. Bivariate analyses of gender, practice type, HCV genotype, eGFR, FIB-4, baseline HCV viral load and insurance type did not reveal significant association with SVR12.

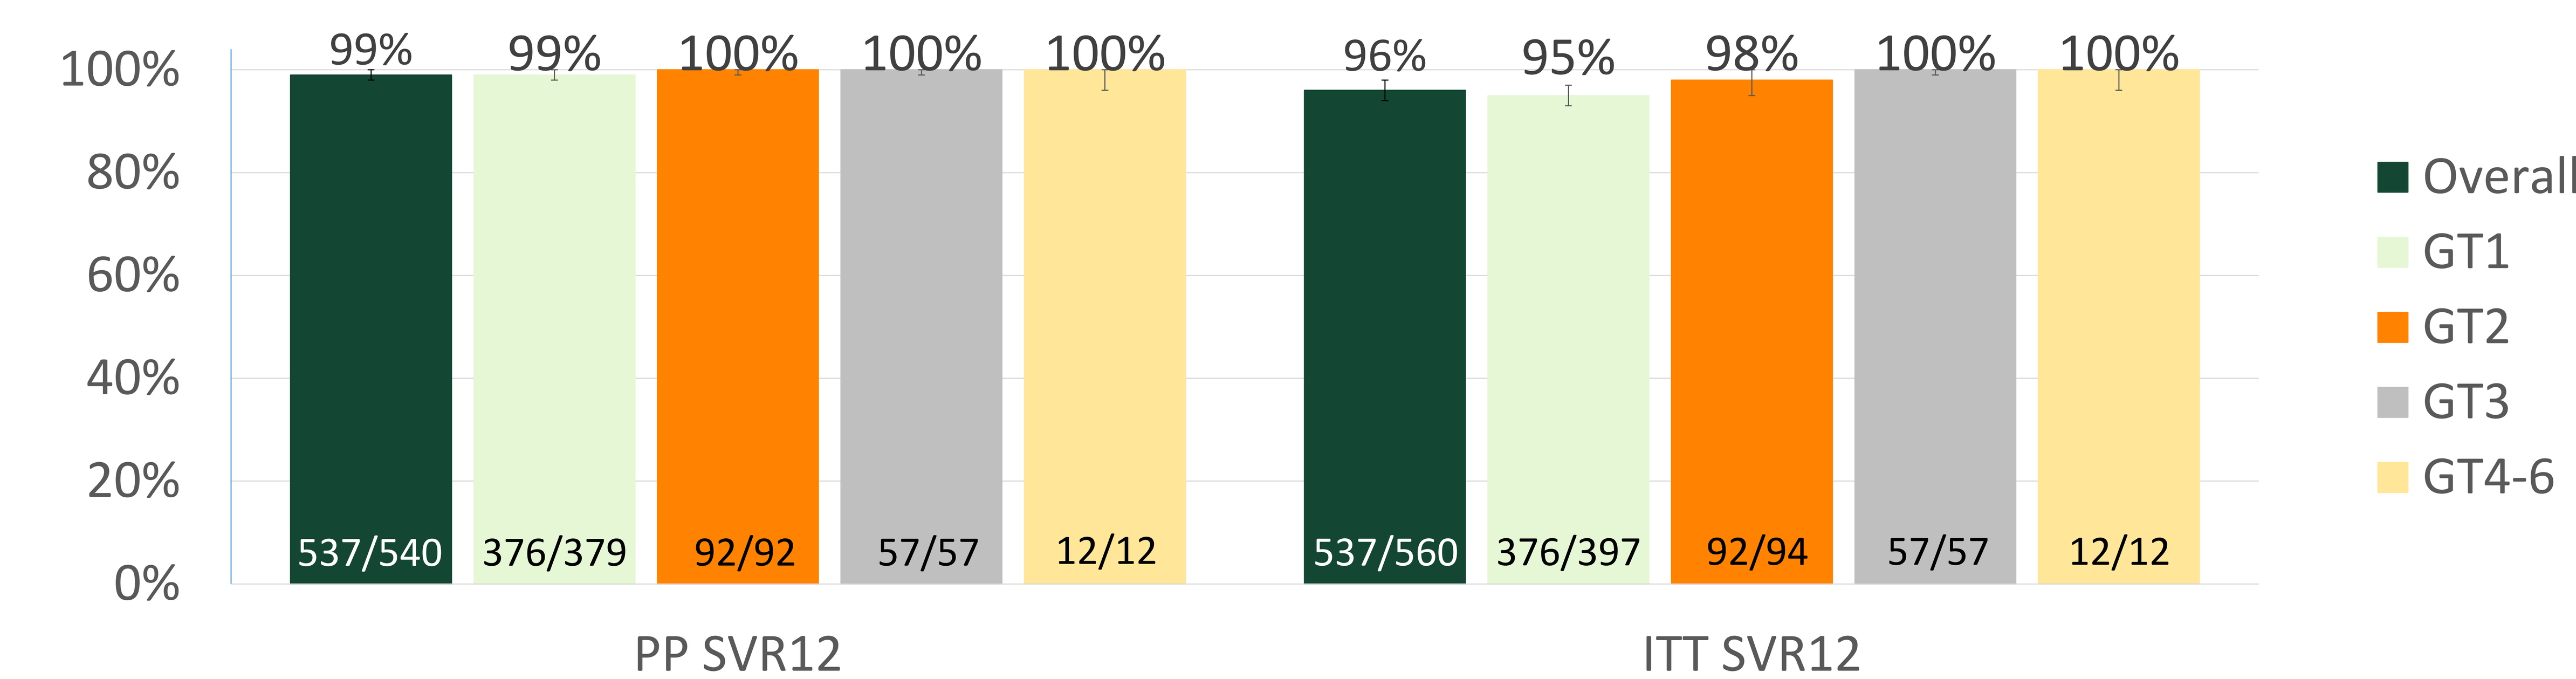
100%
80%
60%
40%
20%
0%
99%
99%
100%
100%
100%
537/540
376/379
92/92
57/57
12/12
96%
95%
98%
100%
100%
537/560
376/397
92/94
57/57
12/12
PP SVR12
ITT SVR12
Overall
GT1
GT2
GT3
GT4-6

6. TREATMENT FAILURES

Reasons for not achieving SVR12 were lost-to-follow-up (2%, 13/560), virologic failure (0.5%, 3/560), and discontinuation (1%, 7/560). The 3 virologic failures were all GT1A. Reasons for discontinuation were medical care unrelated to treatment, patient choice, MD choice, insurance denial/positive drug or alcohol screening, and side effects*.

Type	Patient	Baseline VL	Genotype	FIB-4	Comorbids	Insurance	Practice Type
Discontinued	69, F, Black	1,190,000	2Mixed	<1.45	CKD Stage 5	Medicaid	Community
Discontinued	58, M	839,697	1A	1.45 <= 3.25		Medicaid	Community
Discontinued	59, M	4,790,000	1A	1.45 <= 3.25	Diabetes, CKD Stage 5	Medicare	Community
Discontinued	62, M	2,522,565	1A	1.45 <= 3.25	Anxiety, Depression, HTN, CKD Stage 3	Medicaid	Community
Discontinued	55, M	2,826,821	2B	<1.45	Anxiety, Depression, CKD Stage 4	Medicaid	Community
Discontinued	49, M	1,806	1A	1.45 <= 3.25		Medicaid	Academic
Discontinued	62, F, Black	Not Reported	1A	1.45 <= 3.25	Depression, Diabetes, HLD, HTN	Medicaid	Academic
Lost-to-Follow-Up	63, F	1,397,548	1B	1.45 <= 3.25		Medicare	Community
Lost-to-Follow-Up	60, M, Asian	66,333	1B	1.45 <= 3.25		Medicare	Academic
Lost-to-Follow-Up	51, M	682,235	1A	<1.45	Depression	Medicare	Academic
Lost-to-Follow-Up	76, M, White	5,070,000	1A	1.45 <= 3.25	Diabetes, CKD Stage 3	Medicare	Community
Lost-to-Follow-Up	30, F	169,533	1A	<1.45	Anxiety, Depression	Medicaid	Community
Lost-to-Follow-Up	39, M	454,938	1A	<1.45	HTN	Medicare	Community
Lost-to-Follow-Up	28, F	16,300,000	1A	<1.45		Medicaid	Community
Lost-to-Follow-Up	54, M	30,200,000	1	<1.45		Medicaid	Community
Lost-to-Follow-Up	66, F	4,080,000	1A	<1.45	Anxiety, HTN, CKD Stage 3	Medicare	Academic
Lost-to-Follow-Up	28, M	19,900,000	1A	<1.45		Medicaid	Community
Lost-to-Follow-Up	69, M	1,324,618	1A	1.45 <= 3.25	HTN	Medicare	Community
Lost-to-Follow-Up	50, M, White	11,196,721	1A	<1.45	Diabetes	Medicaid	Community
Lost-to-Follow-Up	46, F	72,752	1A	<1.45	Anxiety, Depression	Medicaid	Community
SVR12 Not Achieved	55, M	1,440,000	1A	<1.45		Commercial	Community
SVR12 Not Achieved	40, M	12,100,000	1A	1.45 <= 3.25	Anxiety, Depression	Commercial	Community
SVR12 Not Achieved	58, M, Black	12,500,000	1A	<1.45		PA	Community

HTN=Hypertension; HLD=Hyperlipidemia; CKD=Chronic Kidney Disease; PA=Patient Assistance. *Data are insufficient to determine whether side effects are related or unrelated to treatment.

7. SUMMARY

Glecaprevir/Pibrentasvir (G/P) is the only regimen approved by the US-FDA with 8 week treatment duration for chronic HCV genotype 1-6 infection in treatment naïve, non-cirrhotic patients. Data from 560 treatment naïve, non-cirrhotic patients who initiated 8-weeks of G/P treatment between August 2017 and April 2018 were collected and analyzed.

Overall SVR12 rate of 96% (537/560) and 99% (537/540) by ITT and PP analyses respectively show high effectiveness of the 8 week G/P regimen in a diverse treatment naïve, non-cirrhotic US population with HCV GT 1,2,3,4 and 6 infection. The virologic failure rate was 0.5% (3/560) and other reasons for not achieving SVR12 were lost-to-follow-up (2%, 13/560) and discontinuation (1%, 7/560).

The design, study conduct, and financial support for the study were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of this publication.

Dr. Flamm advises for, is on the speakers' bureau for, and received grants from Gilead and AbbVie. He advises for and is on the speakers' bureau for Merck. Jens Kort, Steve Marx, and John Strezewski, are employees of AbbVie and may own stock. Dr. Bacon consults for, is on the speakers' bureau for, and received grants from Merck. He advises for, is on the speakers' bureau for, and received grants from AbbVie and Gilead. He advises for and is on the speakers' bureau for Janssen. He advises for and received grants from Bristol-Myers Squibb. He is on the speakers' bureau for Valeant. Dr. Curry consults for and received grants from Gilead. He consults for Trio Health, AbbVie, and Bristol-Myers Squibb. Dr. Tsai consults for, advises for, is on the speakers' bureau for, and received grants from Gilead, Bristol-Myers Squibb, and Merck. N. Wick is employed by Trio Health and received grants from Gilead, AbbVie, and Merck. Dr. Younossi consults for, advises for, and received grants from Gilead. He consults for and advises for AbbVie, Bristol-Myers Squibb, and Intercept. Dr. Afdhal consults for, advises for, and received grants from Gilead. He consults for and advises for Merck, Echosens, GlaxoSmithKline, Ligand, Janssen, Roivant, Co-Crystal, and Shionogi. He received grants from AbbVie and Bristol-Myers Squibb. He is employed by and owns stock in Spring Bank and Trio Healthcare.