Rational Design of Doravirine (DOR): A Review of Development From Bench to Patients

Abstract #545

Carey Hwang, MD, PhD; Ming-Tain Lai, PhD; Daria Hazuda, PhD
Merck & Co., Inc., Kenilworth, NJ, USA

BRIEF HISTORY OF THE NNRTI CLASS

- Early/mid 1990s: discovery of first NNRTIs: 1-(2-hydroxyethoxymethyl)-6-(phenylthio)thymine (HEPT) and tetrahydroimidazo[4,5,1-jkj][1,4]benzodiazepin-2(1H)-one and -thione (TIBO)
- Late 1990s: approval of the first NNRTIs: dipyridodiazepinones (eg, NVP in 1996), bis(heteroaryl) piperazines (eg, DLV in 1997) and benzoxazinones (eg, EFV in 1998)
- Six drugs in this class are currently approved by regulatory authorities (DLV*, EFV, ETR, NVP, RPV and DOR)
- Efavirenz (EFV), the most widely used NNRTI for over a decade, was removed from the preferred list in the US guidelines due to CNS tolerability issues
- Doravirine (DOR) is a non-nucleoside reverse transcriptase inhibitor (NNRTI) that has recently been approved (August 2018) as a single entity, and as a fixed-dose combination with lamivudine (3TC) and tenofovir disoproxil fumarate (TDF) (DOR/3TC/TDF) as once-daily oral treatment for HIV-1 infection in treatment-naïve adults

NNRTI Drug Approvals to Date

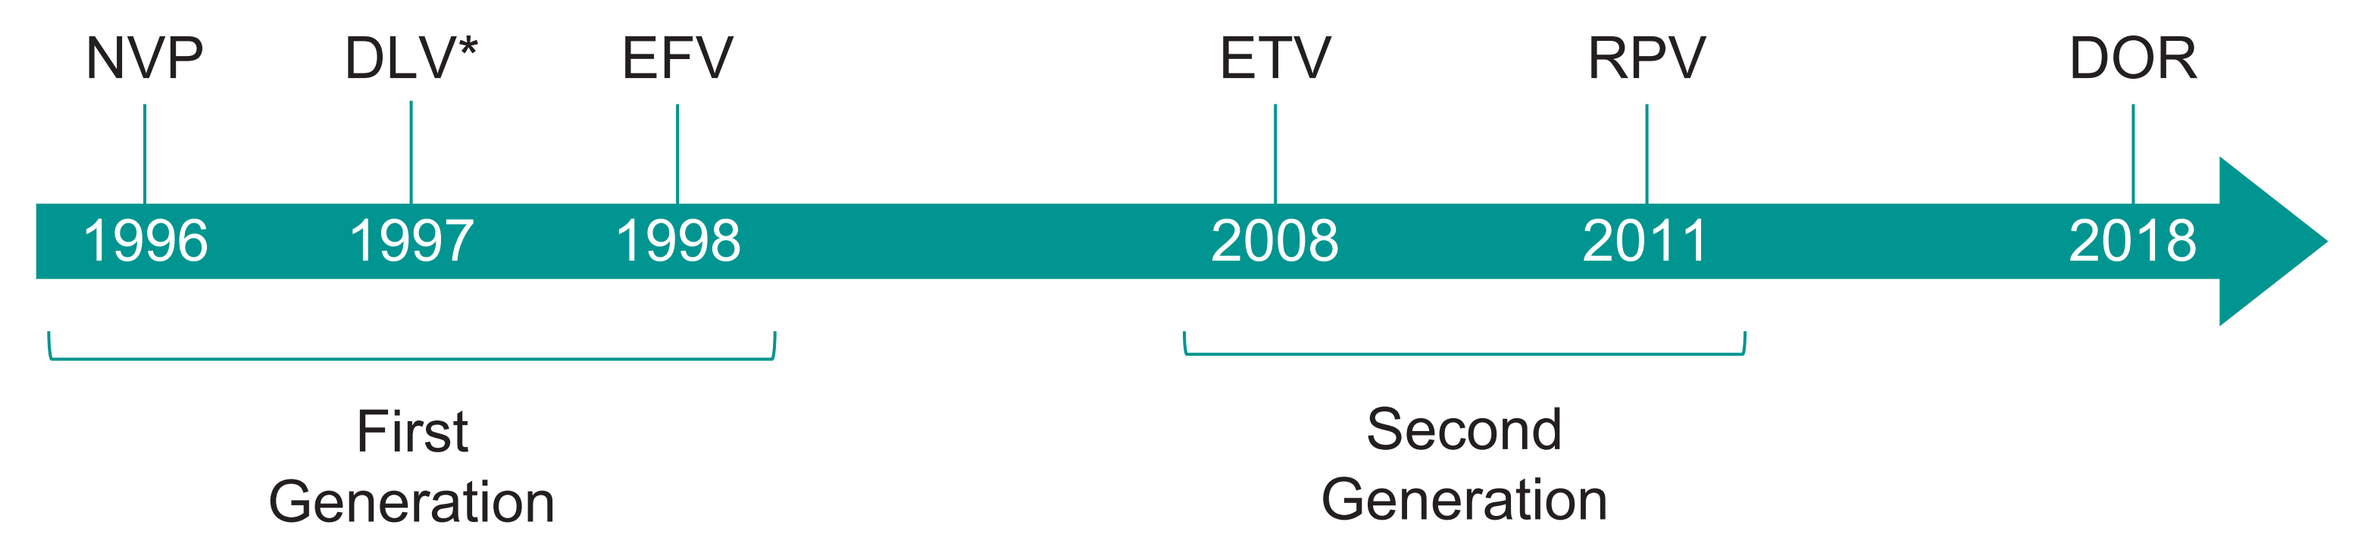
NVP
DLV*
EFV
ETV
RPV
DOR
1996
1997
1998
2008
2011
2018
First Generation
Second Generation

*Anticipate discontinuation in October 2018.
DOR=doravirine; DVL=delavirdine; EFV=efavirenz; ETR=etravirine; NNRTI=nonnucleoside reverse transcriptase inhibitor; NVP=nevirapine; RPV=rilpivirine.

MECHANISM OF ACTION AND PROPENSITY FOR RESISTANCE

- NNRTIs prevent HIV-1 replication by noncompetitively inhibiting reverse transcription of HIV-1 RNA catalyzed by reverse transcriptase (RT)
- NNRTIs bind to a unique, allosteric pocket on HIV-1 RT and induce conformational changes in the substrate-binding site, which interferes with DNA polymerase activity
 - NNRTIs, a heterogenous group of chemical structures, bind to RT at a hydrophobic pocket in the p66 subunit
 - Resistance to first-generation NNRTIs frequently led to treatment failure
- Understanding the mechanisms of NNRTI resistance, coupled with extensive structure-based modeling, led to the discovery of second-generation NNRTIs
- Of the second-generation agents, ETR is the only NNRTI approved to treat NNRTI treatment failures
 - The flexible structure of ETR allows it to rotate within the NNRTI-binding pocket, permitting multiple interactions with RT even in the presence of NNRTI mutations; thus, maintaining activity[1]
 - Due to the flexible binding mode, ETR was also shown to be more difficult to select for resistance in vitro; in clinical studies, 3 or more mutations were required to develop high-level resistance to ETR

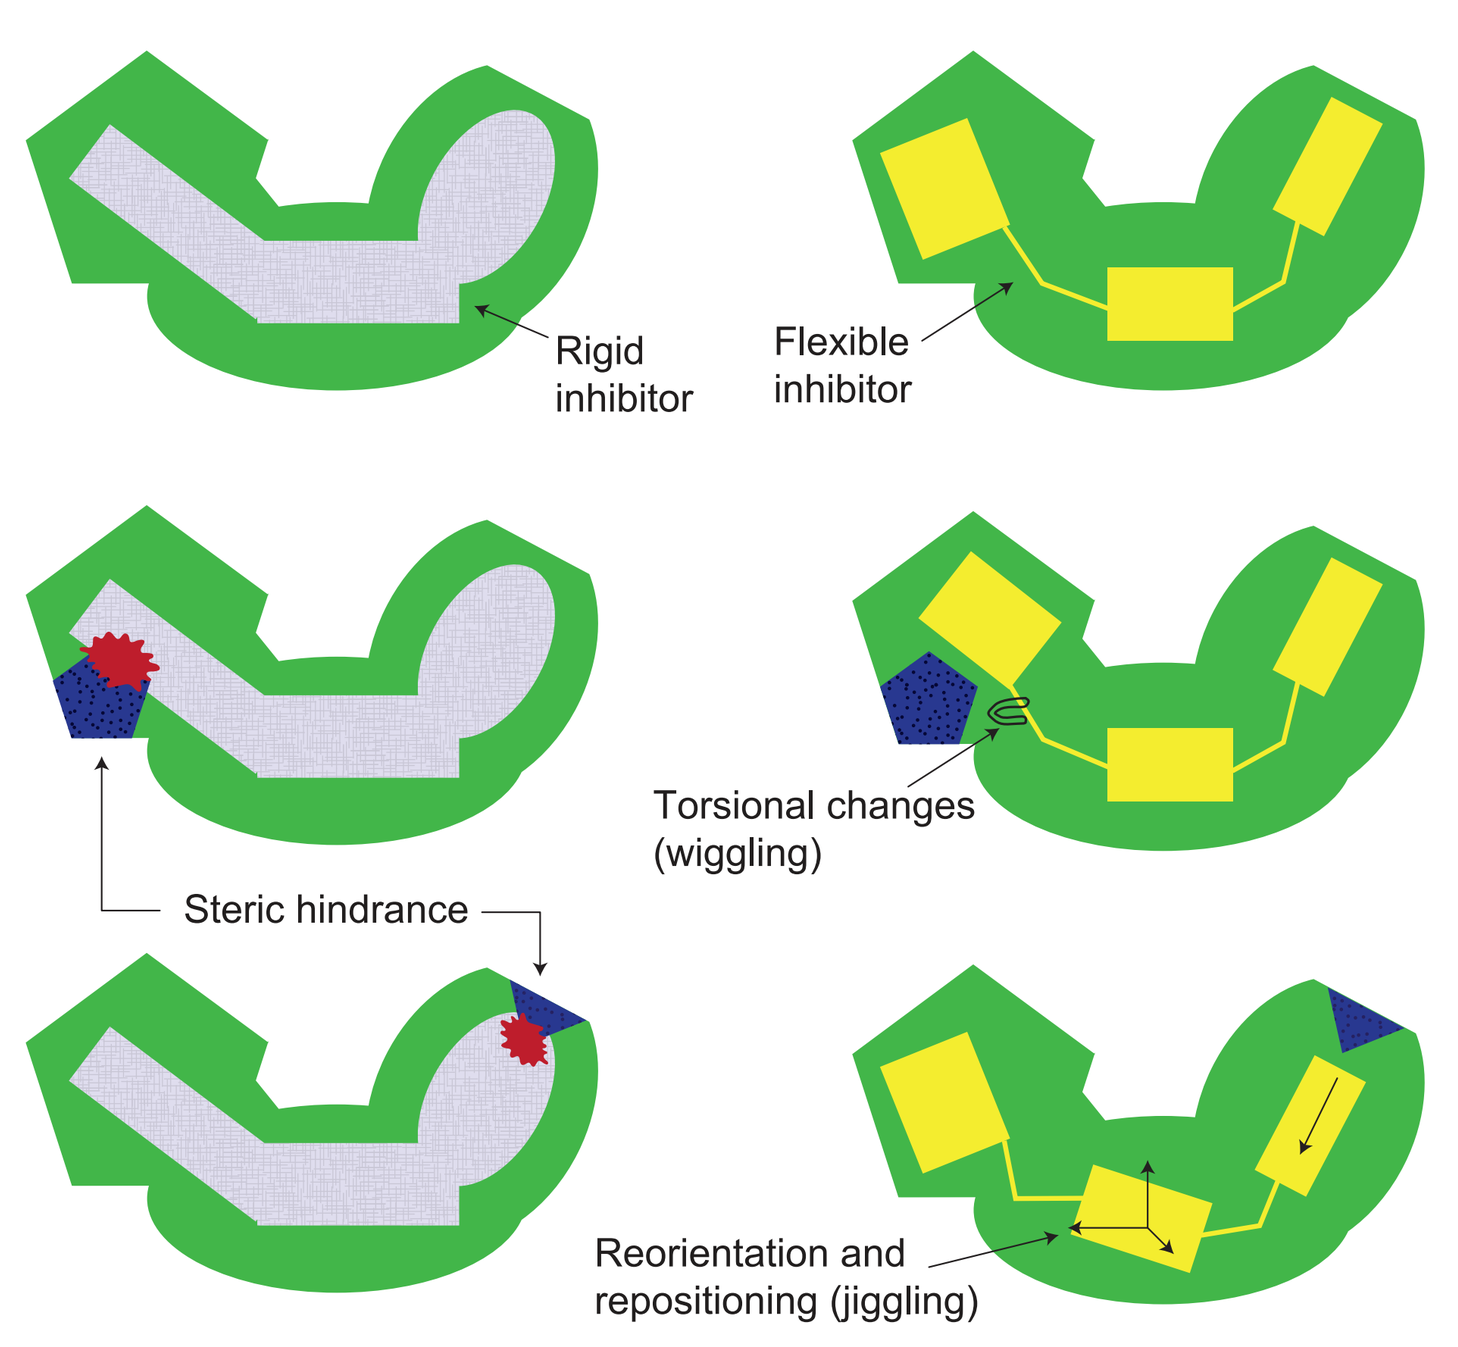
Rigid inhibitor
Flexible inhibitor
Torsional changes (wiggling)
Steric hindrance
Reorientation and repositioning (jiggling)

Reprinted with permission from Das K et al. *J Med Chem.* 2004;47(10):2550-2260.
Copyright (2004) American Chemical Society.

STRUCTURE-BASED DRUG DESIGN COUPLED WITH RESISTANCE PROFILING WERE CRITICAL TO THE DORAVIRINE DISCOVERY EFFORT

- DOR discovery effort started from a series of novel diaryl ether NNRTIs exemplified by MK-1107, a precursor of DOR, with excellent activity vs wild-type (WT) HIV-1 and NNRTI-resistant viruses[2,3]
- In contrast to ETR, comparing X-ray crystal structures of MK-1107 and related analogs bound to the WT and mutant RT enzymes led to a strategy of introducing cyclic constraints in the molecule to optimize the favorable interactions to retain activity against the key resistance mutants
- The favorable resistance profiles of key analogs, including DOR were validated using high-throughput resistance selection

FROM MK-1107 TO DORAVIRINE

- Preorganized structures of MK-1107 and related analogs bound to mutant and WT RTs provided potent compounds with markedly improved antiviral activity against WT and NNRTI-resistant viruses that were comparable or superior to more flexible analogs
- Subsequent iterations of MK-1107 delivered the optimized analog: DOR

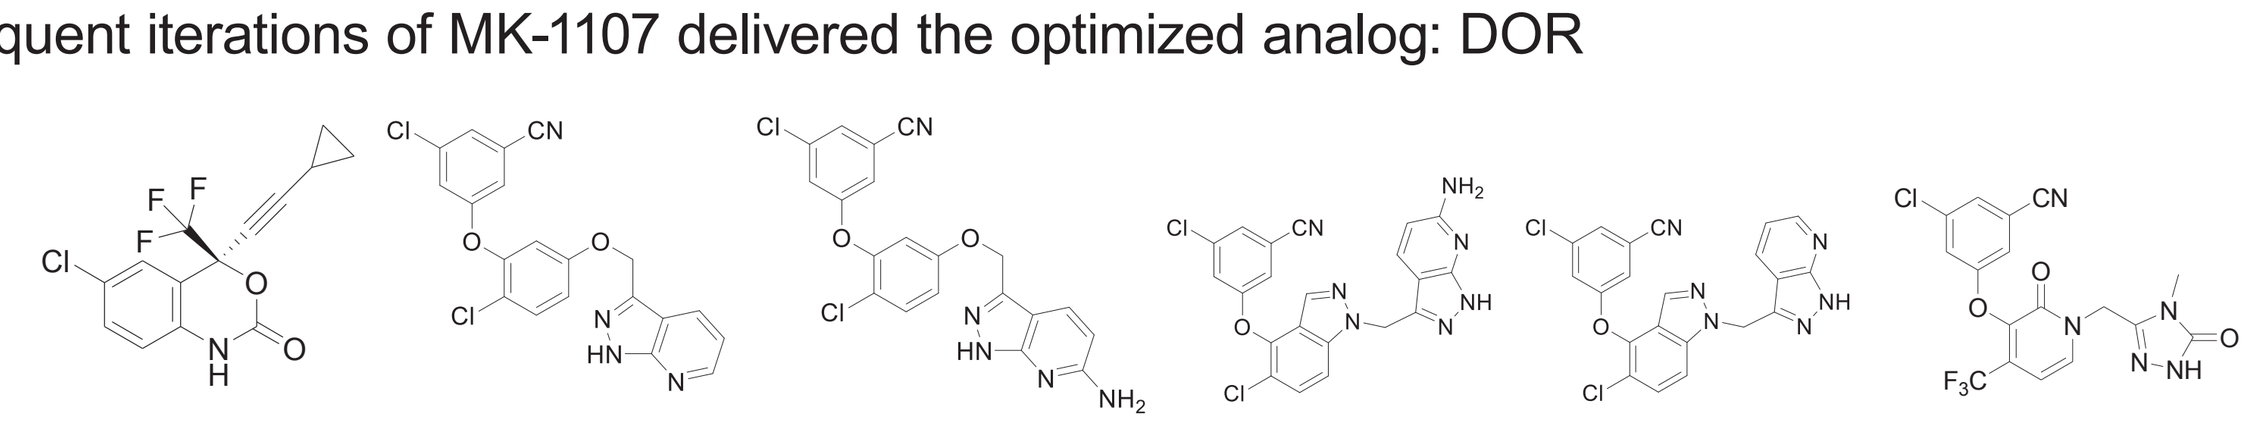

Virus	EC$_{50}$ (nM)[a] EFV[4]	MK-1107	MK-4965	MK-7445	MK-6186	DOR[4]
WT	41	34	41	77	64	19
K103N	1427	11[b]	17[b]	25[b]	113	42
Y181C	80	23[b]	28[b]	81[b]	335	25
K103N/Y181C	2943	NA	84[b]	202[b]	650	52

[a]Tested in 50% FBS; [b]Tested in 10% FBS.
EC$_{50}$=half maximal effective concentration; FBS=fasting blood sugar.

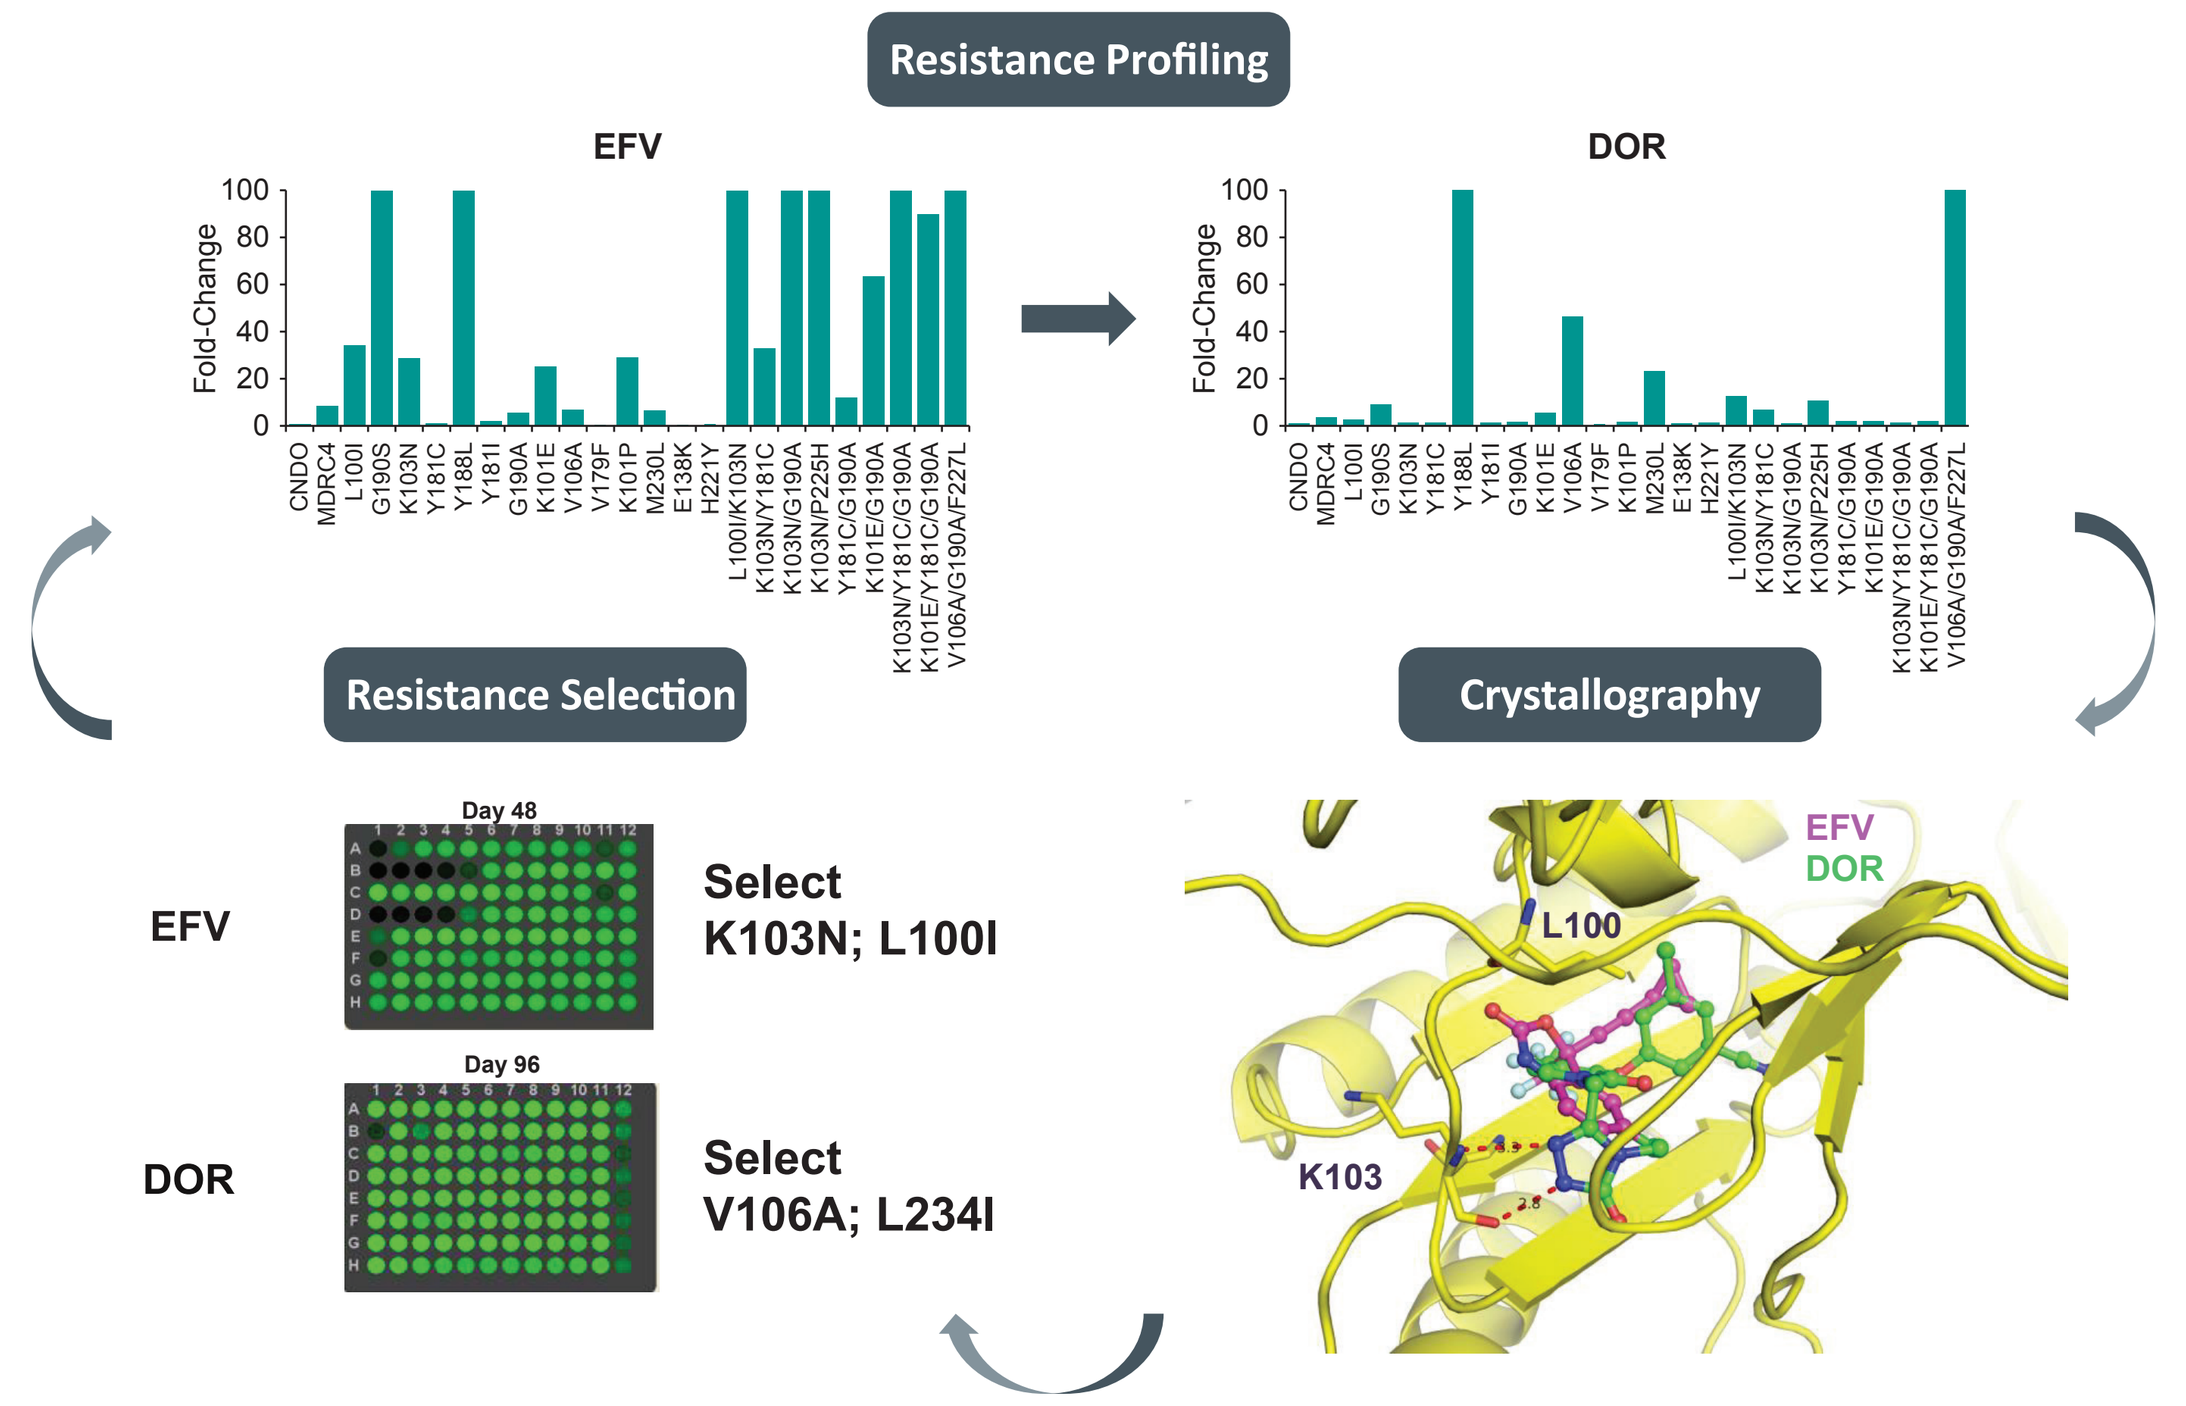
Resistance Profiling
EFV
DOR
Fold-Change
Resistance Selection
Crystallography
Day 48
Day 96
EFV
Select K103N; L100I
DOR
Select V106A; L234I
EFV
DOR
L100
K103

X-ray Structure of DOR With Reverse Transcriptase (RT)

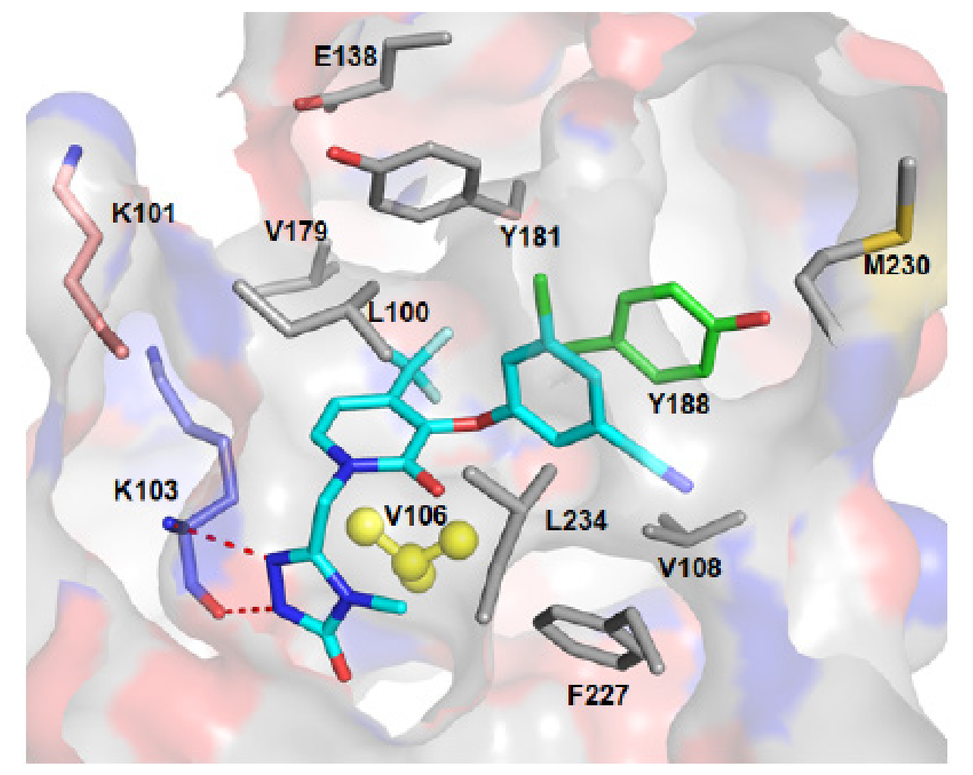
E138
K101
V179
Y181
M230
L100
Y188
K103
V106
L234
V108
F227

Doravirine interacts with backbone of K103 (not side chain)
No interaction with Y181, L100, E138, K101 but V106 is key[5]

Reprinted with permission from Feng M, et al. *Antimicrob Agents Chemother.* 2015;59(1):590-598. Copyright (2015) American Society for Microbiology.

- Structural studies suggested a key hydrophobic interaction between the V106 side chain and the central phenyl ring of DOR[3]
- Resistance profiling studies with DOR and related analogs demonstrated that they were less effective against viruses that contained the triple NNRTI resistance mutation pattern, V106A/G190A/F227L[3]
- DOR resistance was slower to appear in vitro as compared with first-generation NNRTIs, such as EFV[5]

DOR Displays Greater Activity vs RPV, EFV, and ETR Against WT and NNRTI-Resistant Viruses[6,7]

NNRTI	Antiviral Activity IC$_{50}$ (nM)[a] WT	K103N	Y181C	K103N/Y181C	Protein Binding (%)
DOR	12	21	31	33	76.0
RPV	58	56	169	318	99.6
EFV	30	1173	90	3119	99.6
ETR	67	68	383	478	99.9

[a]In vitro multiple cycle infectivity assay conducted in the presence of 100% normal human serum.

PRECLINICAL AND PHARMACOKINETIC STUDIES OF DORAVIRINE

- DOR exhibited favorable pharmacokinetic profiles in rats and dogs, suggesting the potential for once-daily dosing in humans[3]
- Preclinical studies also suggested the potential for DOR to be administered at low doses conducive to the development of a fixed-dose coformulation with other antiretroviral drugs[3]
- DOR is a substrate for CYP3A4, but unlike other NNRTIs, DOR does not impact the expression of CYP3A4 or other major drug-metabolizing enzymes or transporters[8]
 - No clinically meaningful interactions in humans between DOR (with tenofovir disoproxil fumarate [TDF]) and atorvastatin, oral contraceptives, or pantoprazole[9,10]
- No clinically meaningful effect of food on DOR[11]

Disposition of DOR[12]

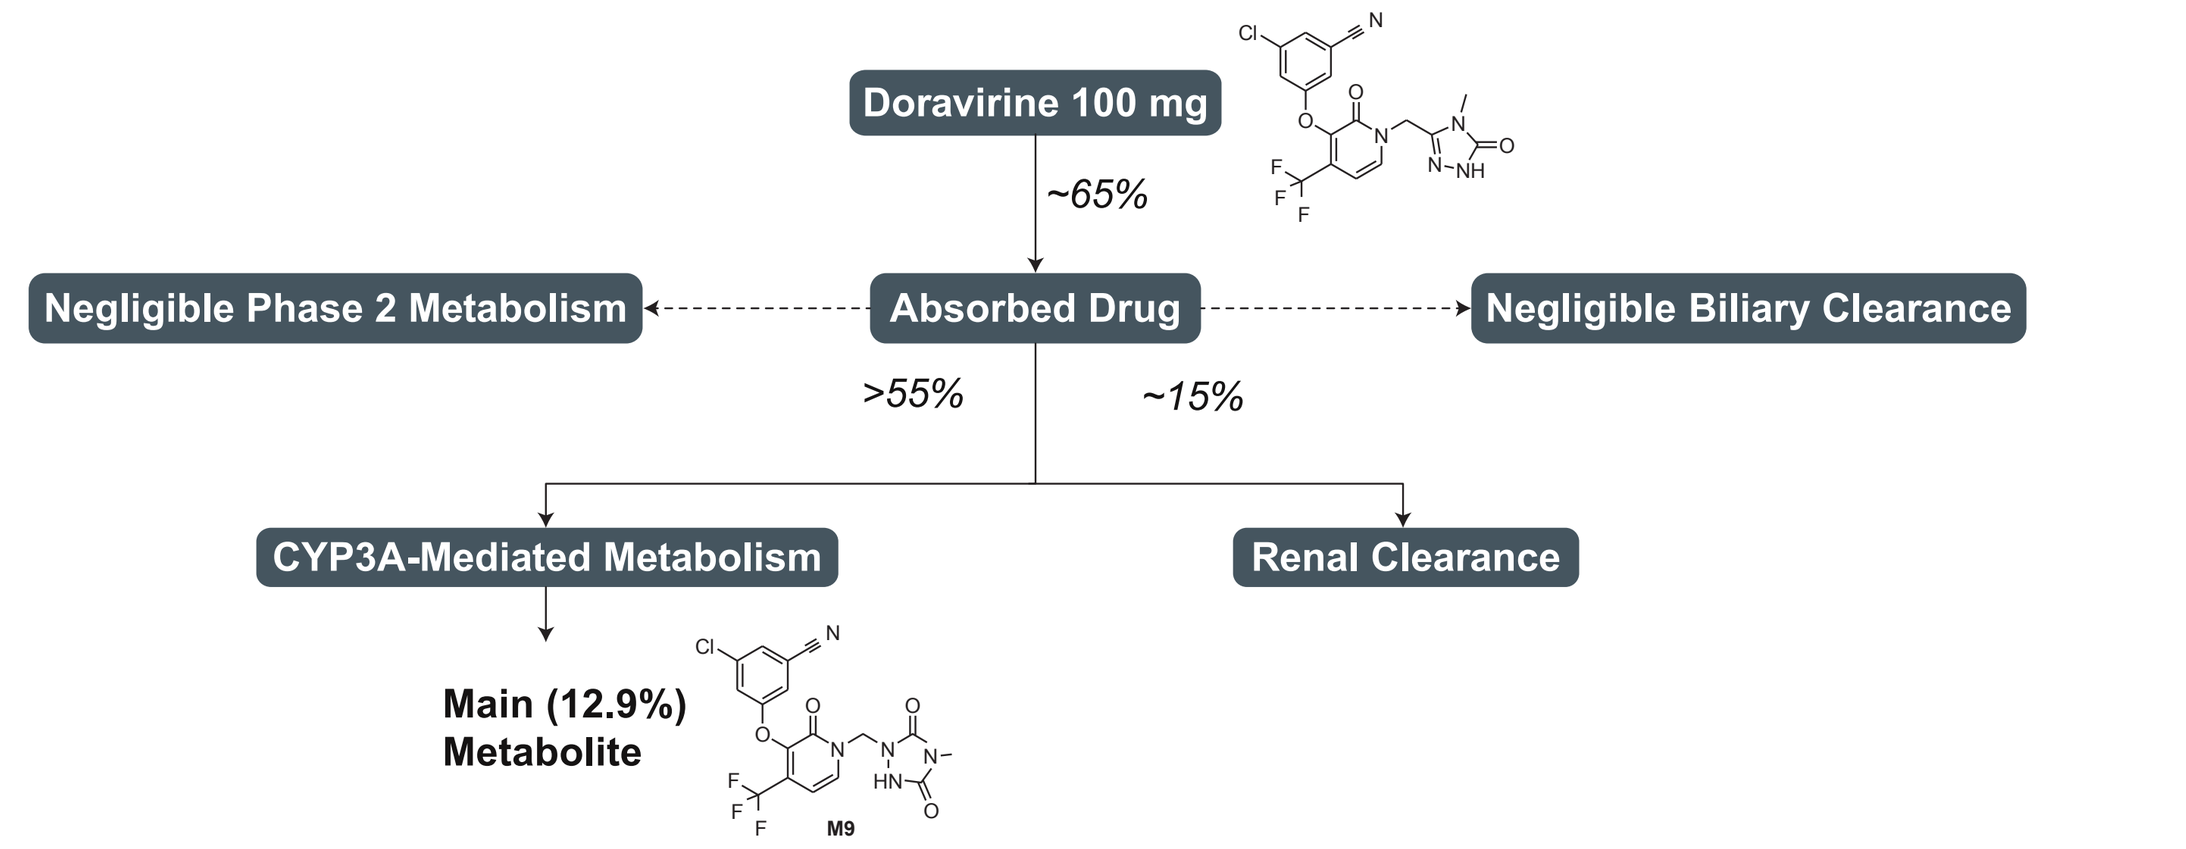
Doravirine 100 mg
~65%
Negligible Phase 2 Metabolism
Absorbed Drug
Negligible Biliary Clearance
>55%
~15%
CYP3A-Mediated Metabolism
Renal Clearance
Main (12.9%) Metabolite
M9

SAFETY AND TOLERABILITY

- Quantitative EEG methods were developed in preclinical species and validated in humans with EFV to derisk potential CNS toxicity of novel NNRTIs
- Quantitative preclinical electroencephalography (EEG) studies of MK-6186, a closely related analog of DOR, found no measurable central nervous system (CNS) toxicity
- Adverse CNS effects are observed with EFV
 - Mechanism(s) responsible for the adverse CNS effects of EFV remain unclear, but several clinical studies have demonstrated associated changes in brain EEG and sleep architecture[13]
- In clinical studies, DOR has been shown to have fewer neuropsychiatric AEs vs EFV with a CNS profile indistinguishable from DRV[14,15]

Neuro Target In Vitro Activities

K$_i$ or IC$_{50}$ <10µM — EFV Target	K$_i$ (µM)	RPV Target	K$_i$ (µM)	DOR Target	K$_i$ (µM)
Norepinephrine transporter	4.1	Monoamine oxidase (MAO-A)	2.3 (IC$_{50}$)	Serotonin (5-HT$_{2B}$)	1.6
Dopamine transporter	5.6	Phosphodiesterase (PDE3)	8.0 (IC$_{50}$)		
Progesterone (PR-B)	2.3	Protein Ser/Thr Kinase MAPK3 (ERK1)	2.4 (IC$_{50}$)		
Serotonin (5-HT$_{2A}$)	1.1	Protein Ser/Thr Kinase, MARK3	3.1 (IC$_{50}$)		
Serotonin (5-HT$_{2B}$)	6.3	Protein Ser/Thr Kinase, PKA, nonselective	2.6 (IC$_{50}$)		
Serotonin (5-HT$_{2C}$)	0.34	Protein Ser/Thr Kinase, PKC, nonselective	2.6 (IC$_{50}$)		
Serotonin (5-HT$_6$)	0.28	Adenosine A$_1$	4.1		
Androgen AR	4.0	Adenosine A$_2$	3.8		
		Adenosine transporter	0.59		
		Adrenergic β$_1$	6.6		
		Adrenergic β$_3$	6.2		
		Norepinephrine transporter	6.6		
		Cannabinoid (CB$_1$)	1.7		
		Histamine (H$_4$)	2.3		
		Progesterone PR-B	0.43		
		Serotonin (5-HT$_{2B}$)	1.1		
		Androgen receptor	4.5		
		Prostanoid thromboxane A$_2$ (TP)	4.9		
		Prostanoid FP	4.6 (IC$_{50}$)		

CLINICAL TRIAL OBSERVATIONS

- The clinical development program for DOR and the combination, DOR/3TC/TDF includes:
 - 36 phase 1 trials
 - 3 phase 2b trials: P007 (NCT01632345), DRIVE-BEYOND (NCT02629822), DRIVE-CLEAR (NCT02652260)
 - 3 phase 3 trials: DRIVE-FORWARD (NCT02275780),[14] DRIVE-AHEAD (NCT02403674),[15] DRIVE-SHIFT (NCT02397096)
- DRIVE-FORWARD: DOR was noninferior to darunavir plus ritonavir (DRV+r), both given with 2 nucleoside reverse transcriptase inhibitors (NRTIs) in treatment-naïve patients[14]
 - HIV-1 RNA <50 copies/mL at Week 48: 83.8% DOR vs 79.9% DRV+r; difference = 3.9% (95% CI: -1.6 to 9.4)
 - CNS tolerability was comparable, with a more favorable lipid profile for DOR
- DRIVE-AHEAD: DOR/3TC/TDF was non-inferior to EFV/FTC/TDF[15]
 - HIV-1 RNA <50 copies/mL at Week 48 : 84.3% DOR vs 80.8% EFV; difference = 3.5% (95% CI: -2.0 to 9.0)
 - DOR had significantly fewer neuropsychiatric events and a more favorable lipid profile compared with EFV

Proportion of Participants With HIV-1 RNA <50 copies/mL at Week 48 (FDA Snapshot Approach). A. DRIVE-FORWARD[14]; B. DRIVE-AHEAD[15]

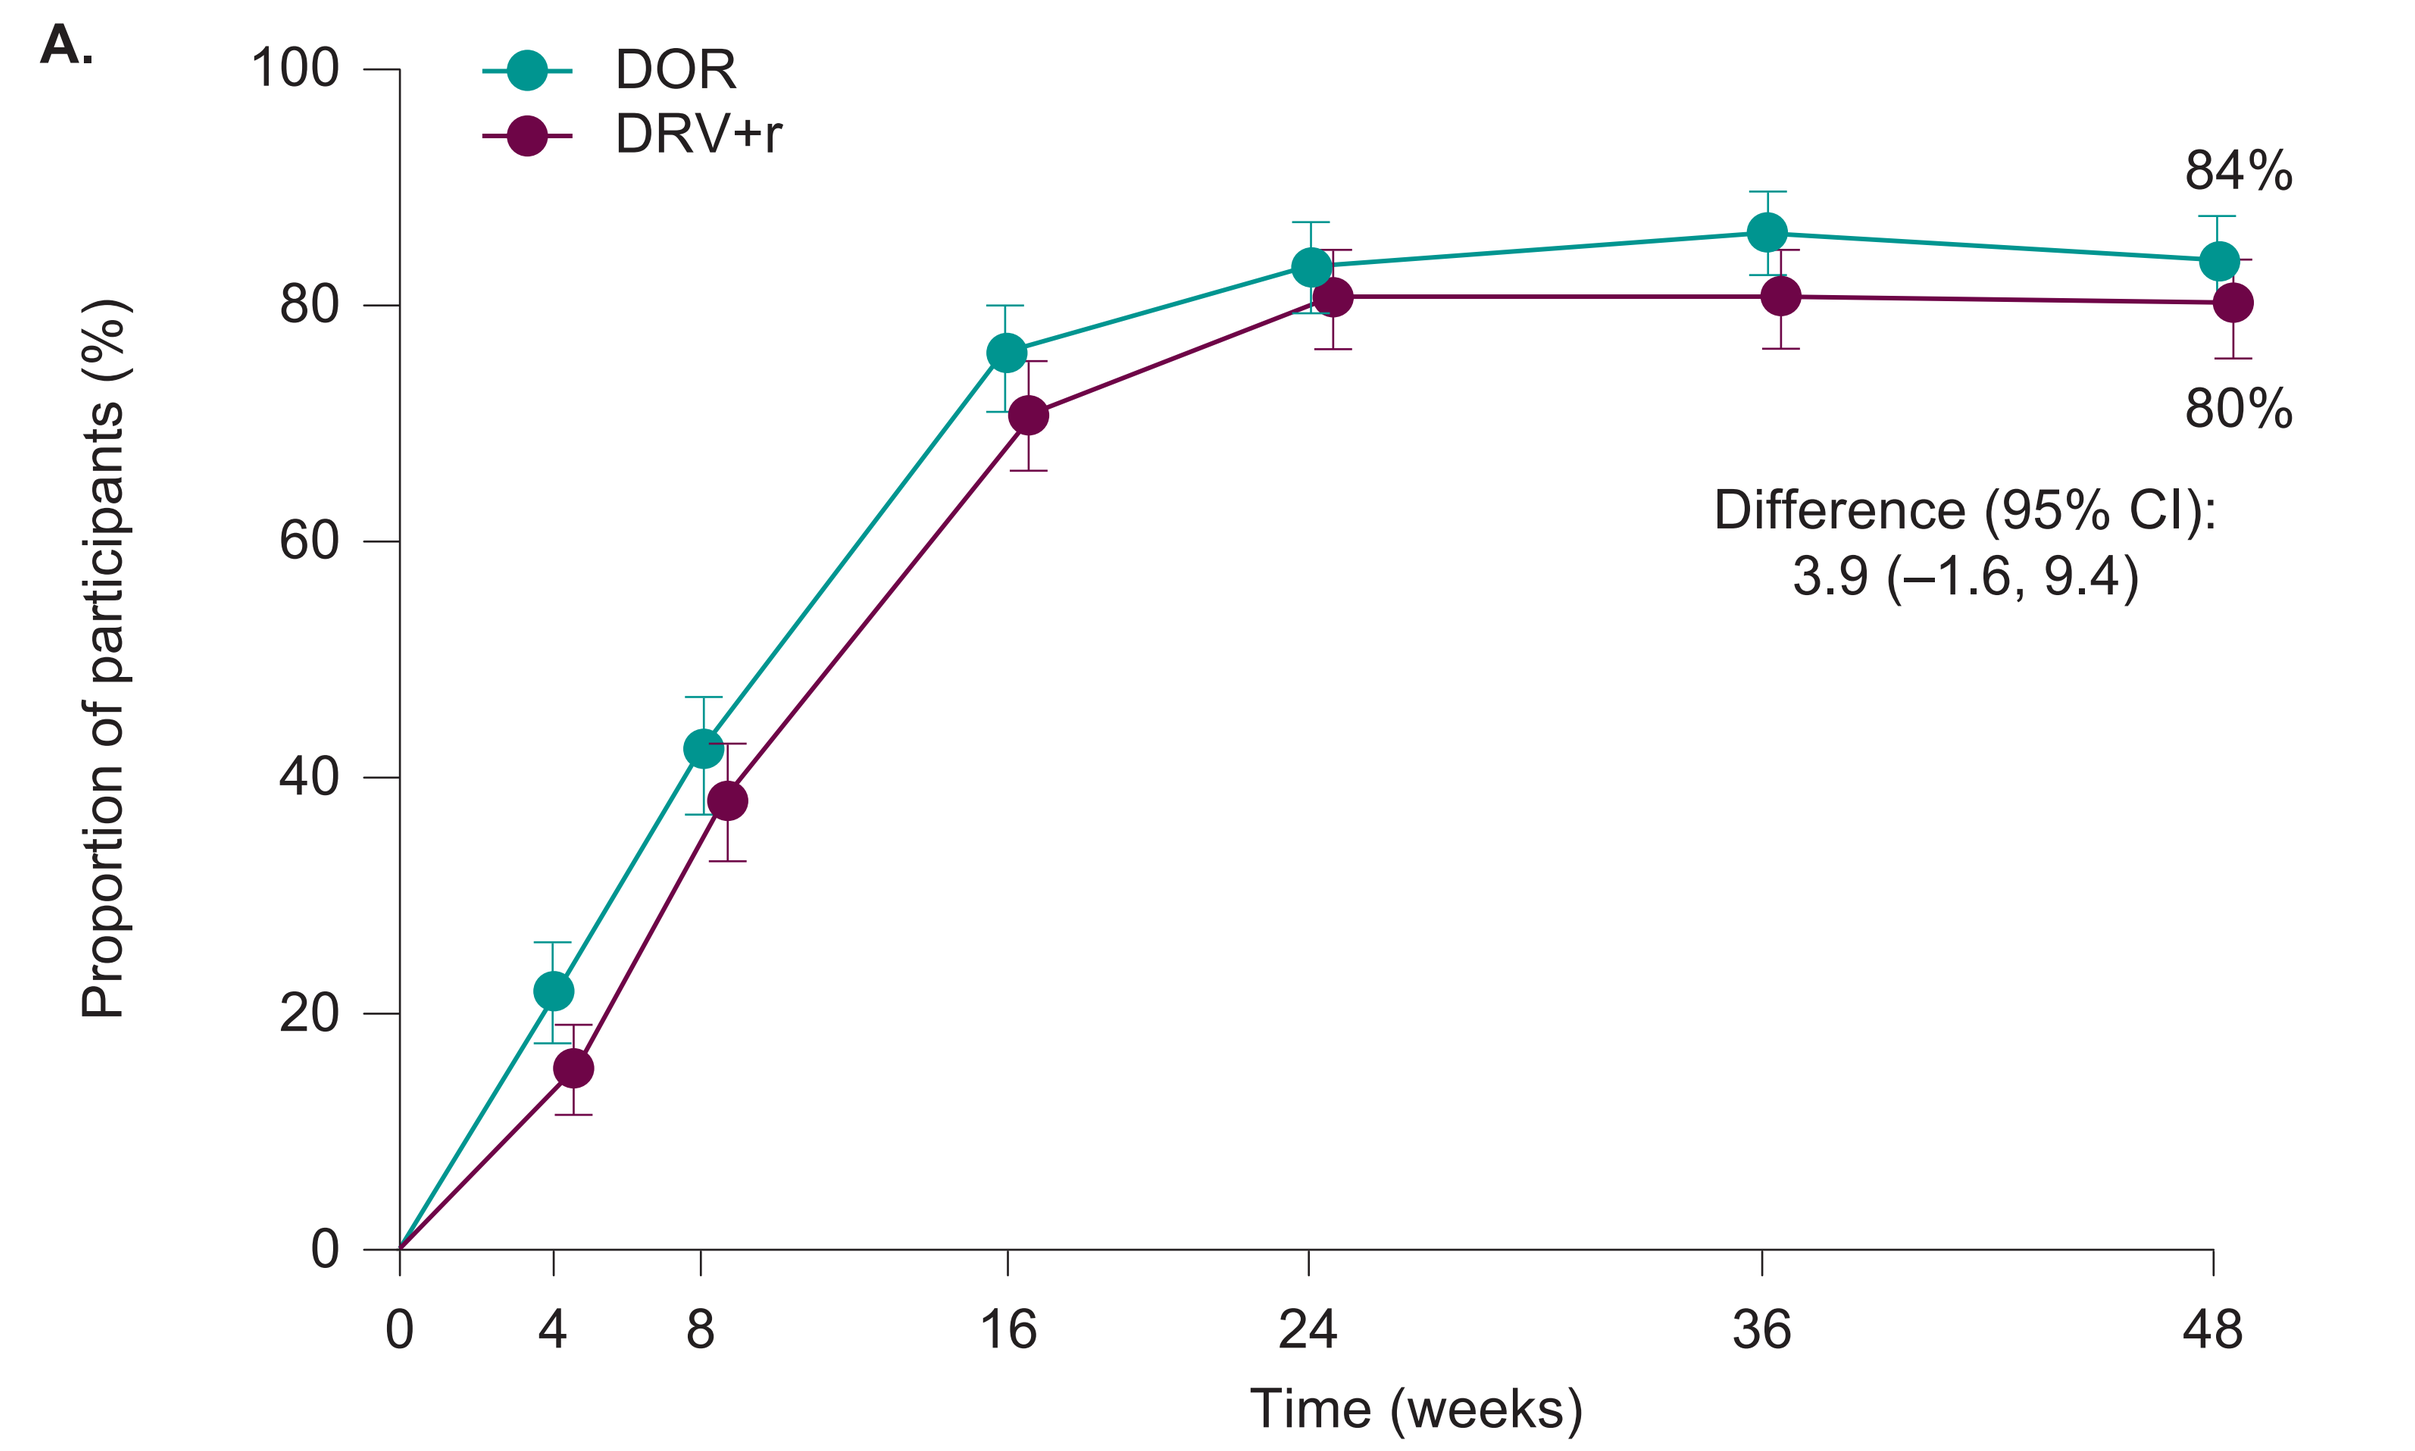
A.
DOR
DRV+r
84%
80%
Difference (95% CI):
3.9 (–1.6, 9.4)
Proportion of participants (%)
Time (weeks)

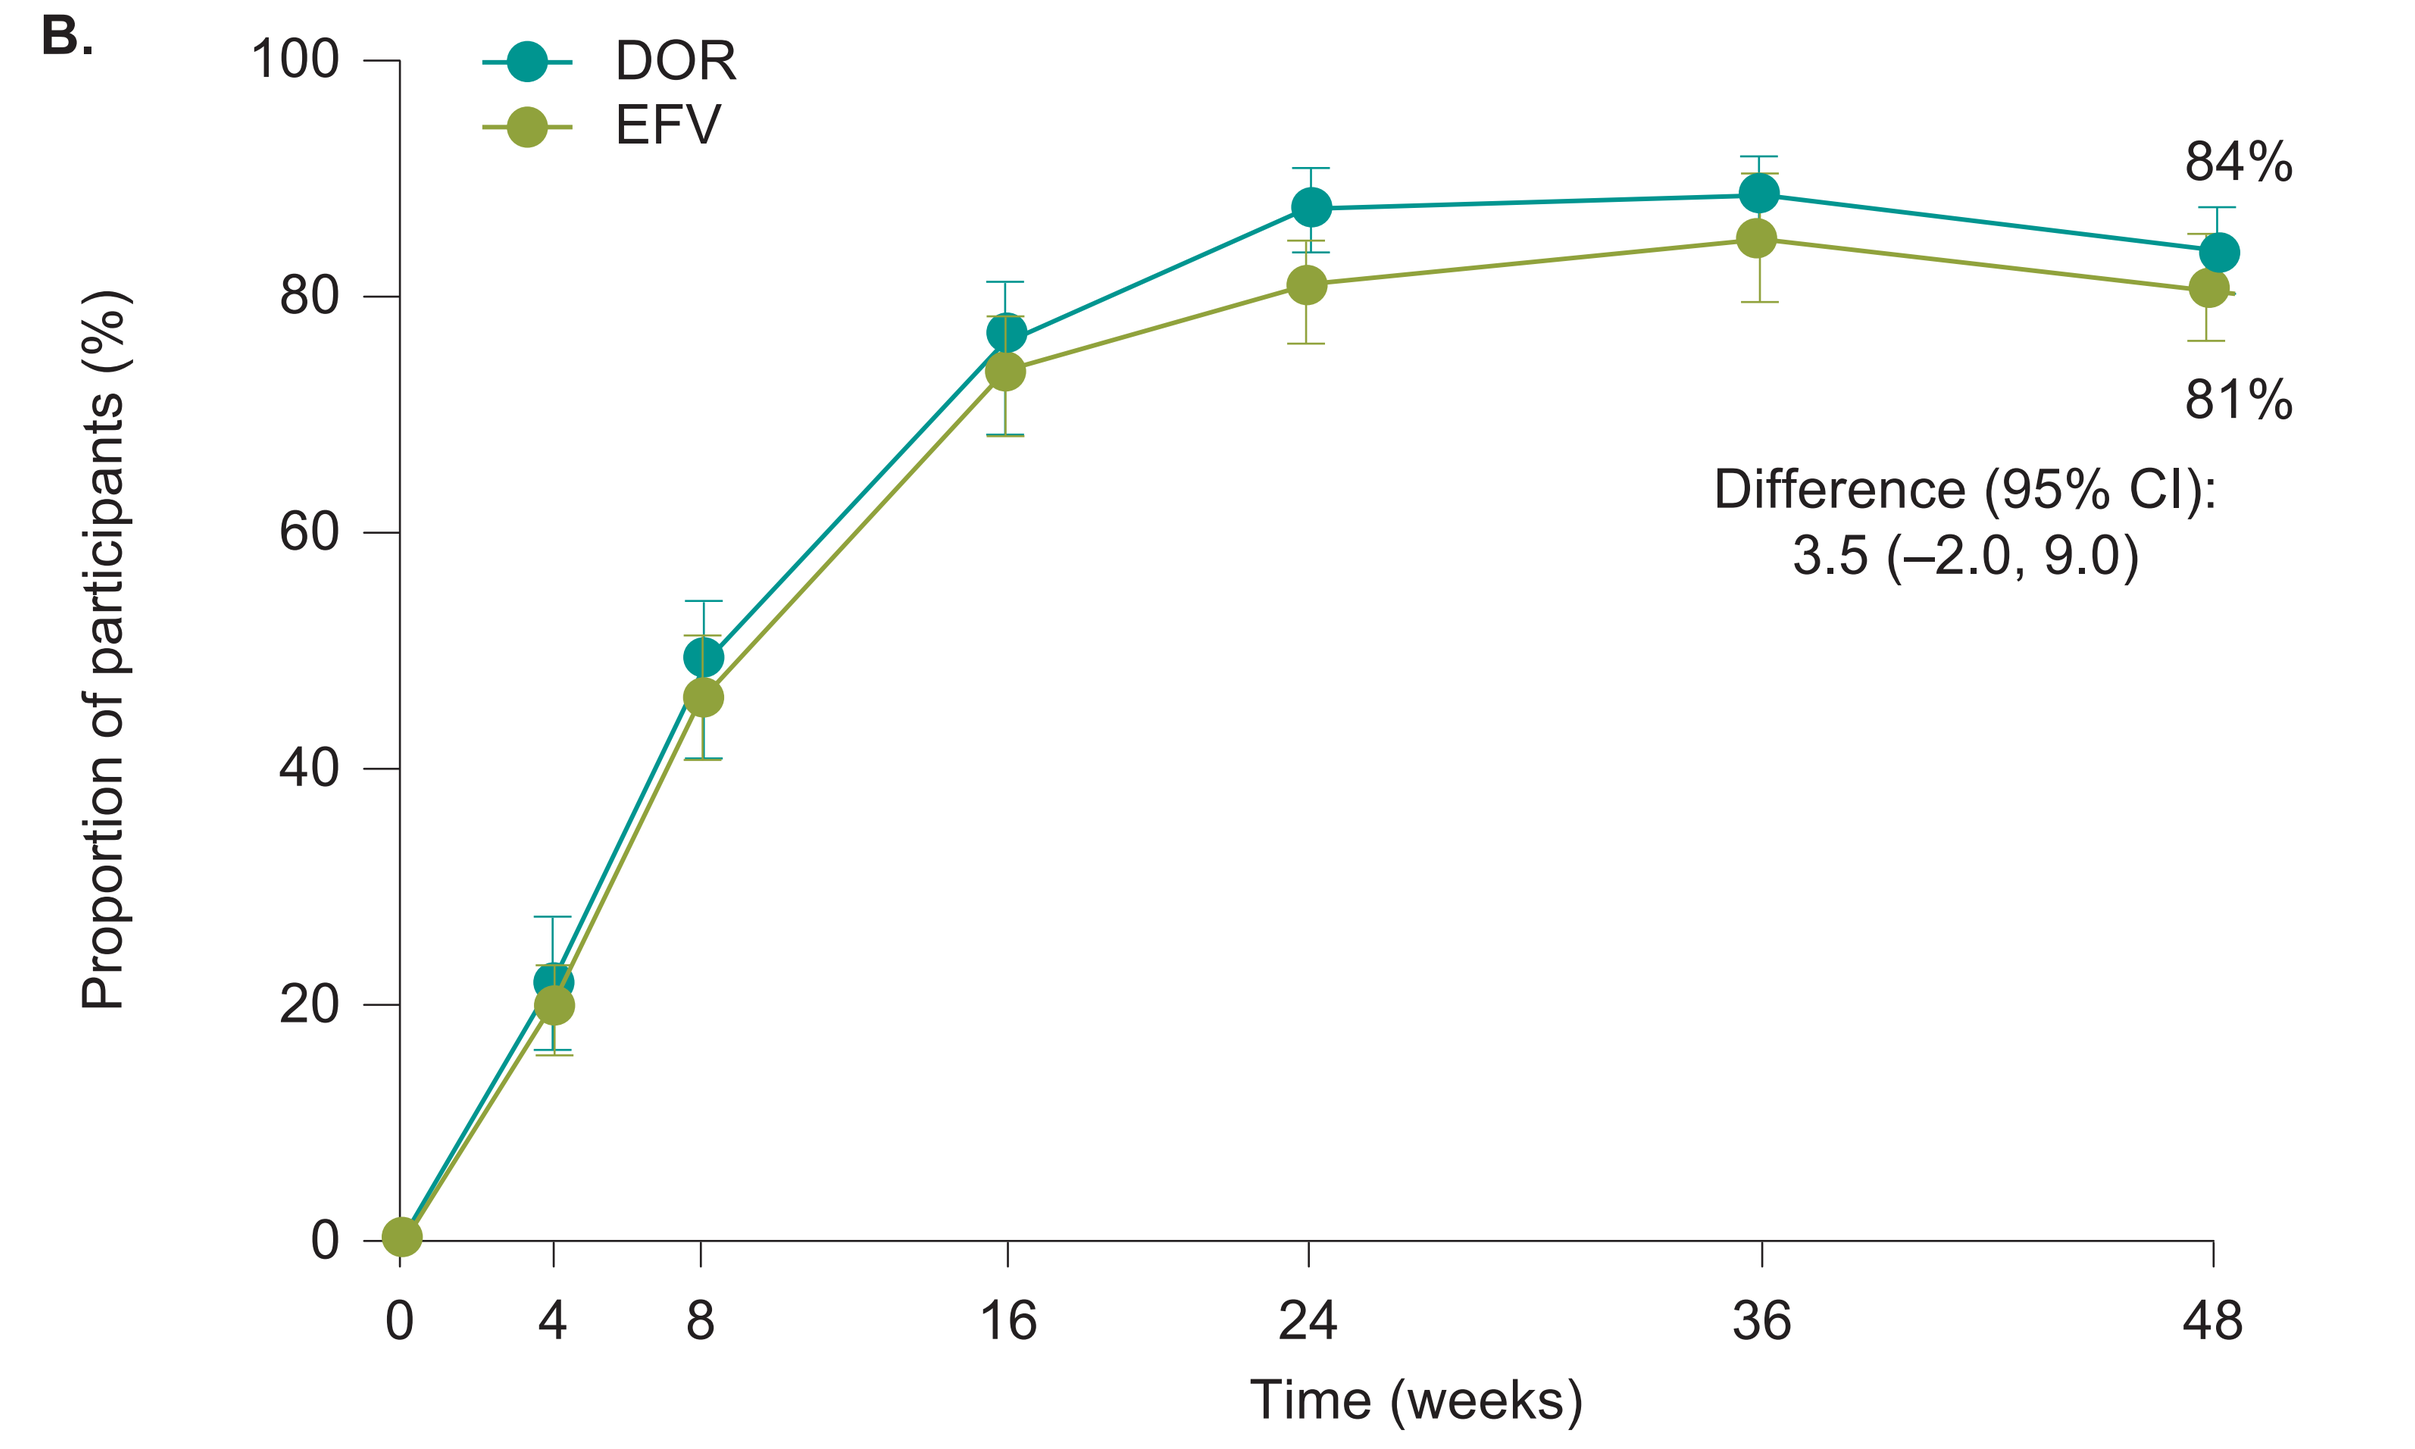
B.
DOR
EFV
84%
81%
Difference (95% CI):
3.5 (–2.0, 9.0)
Proportion of participants (%)
Time (weeks)

DRIVE-AHEAD: Proportion of Participants With Predefined Neuropsychiatric Adverse Events[a] at Week 48[15]

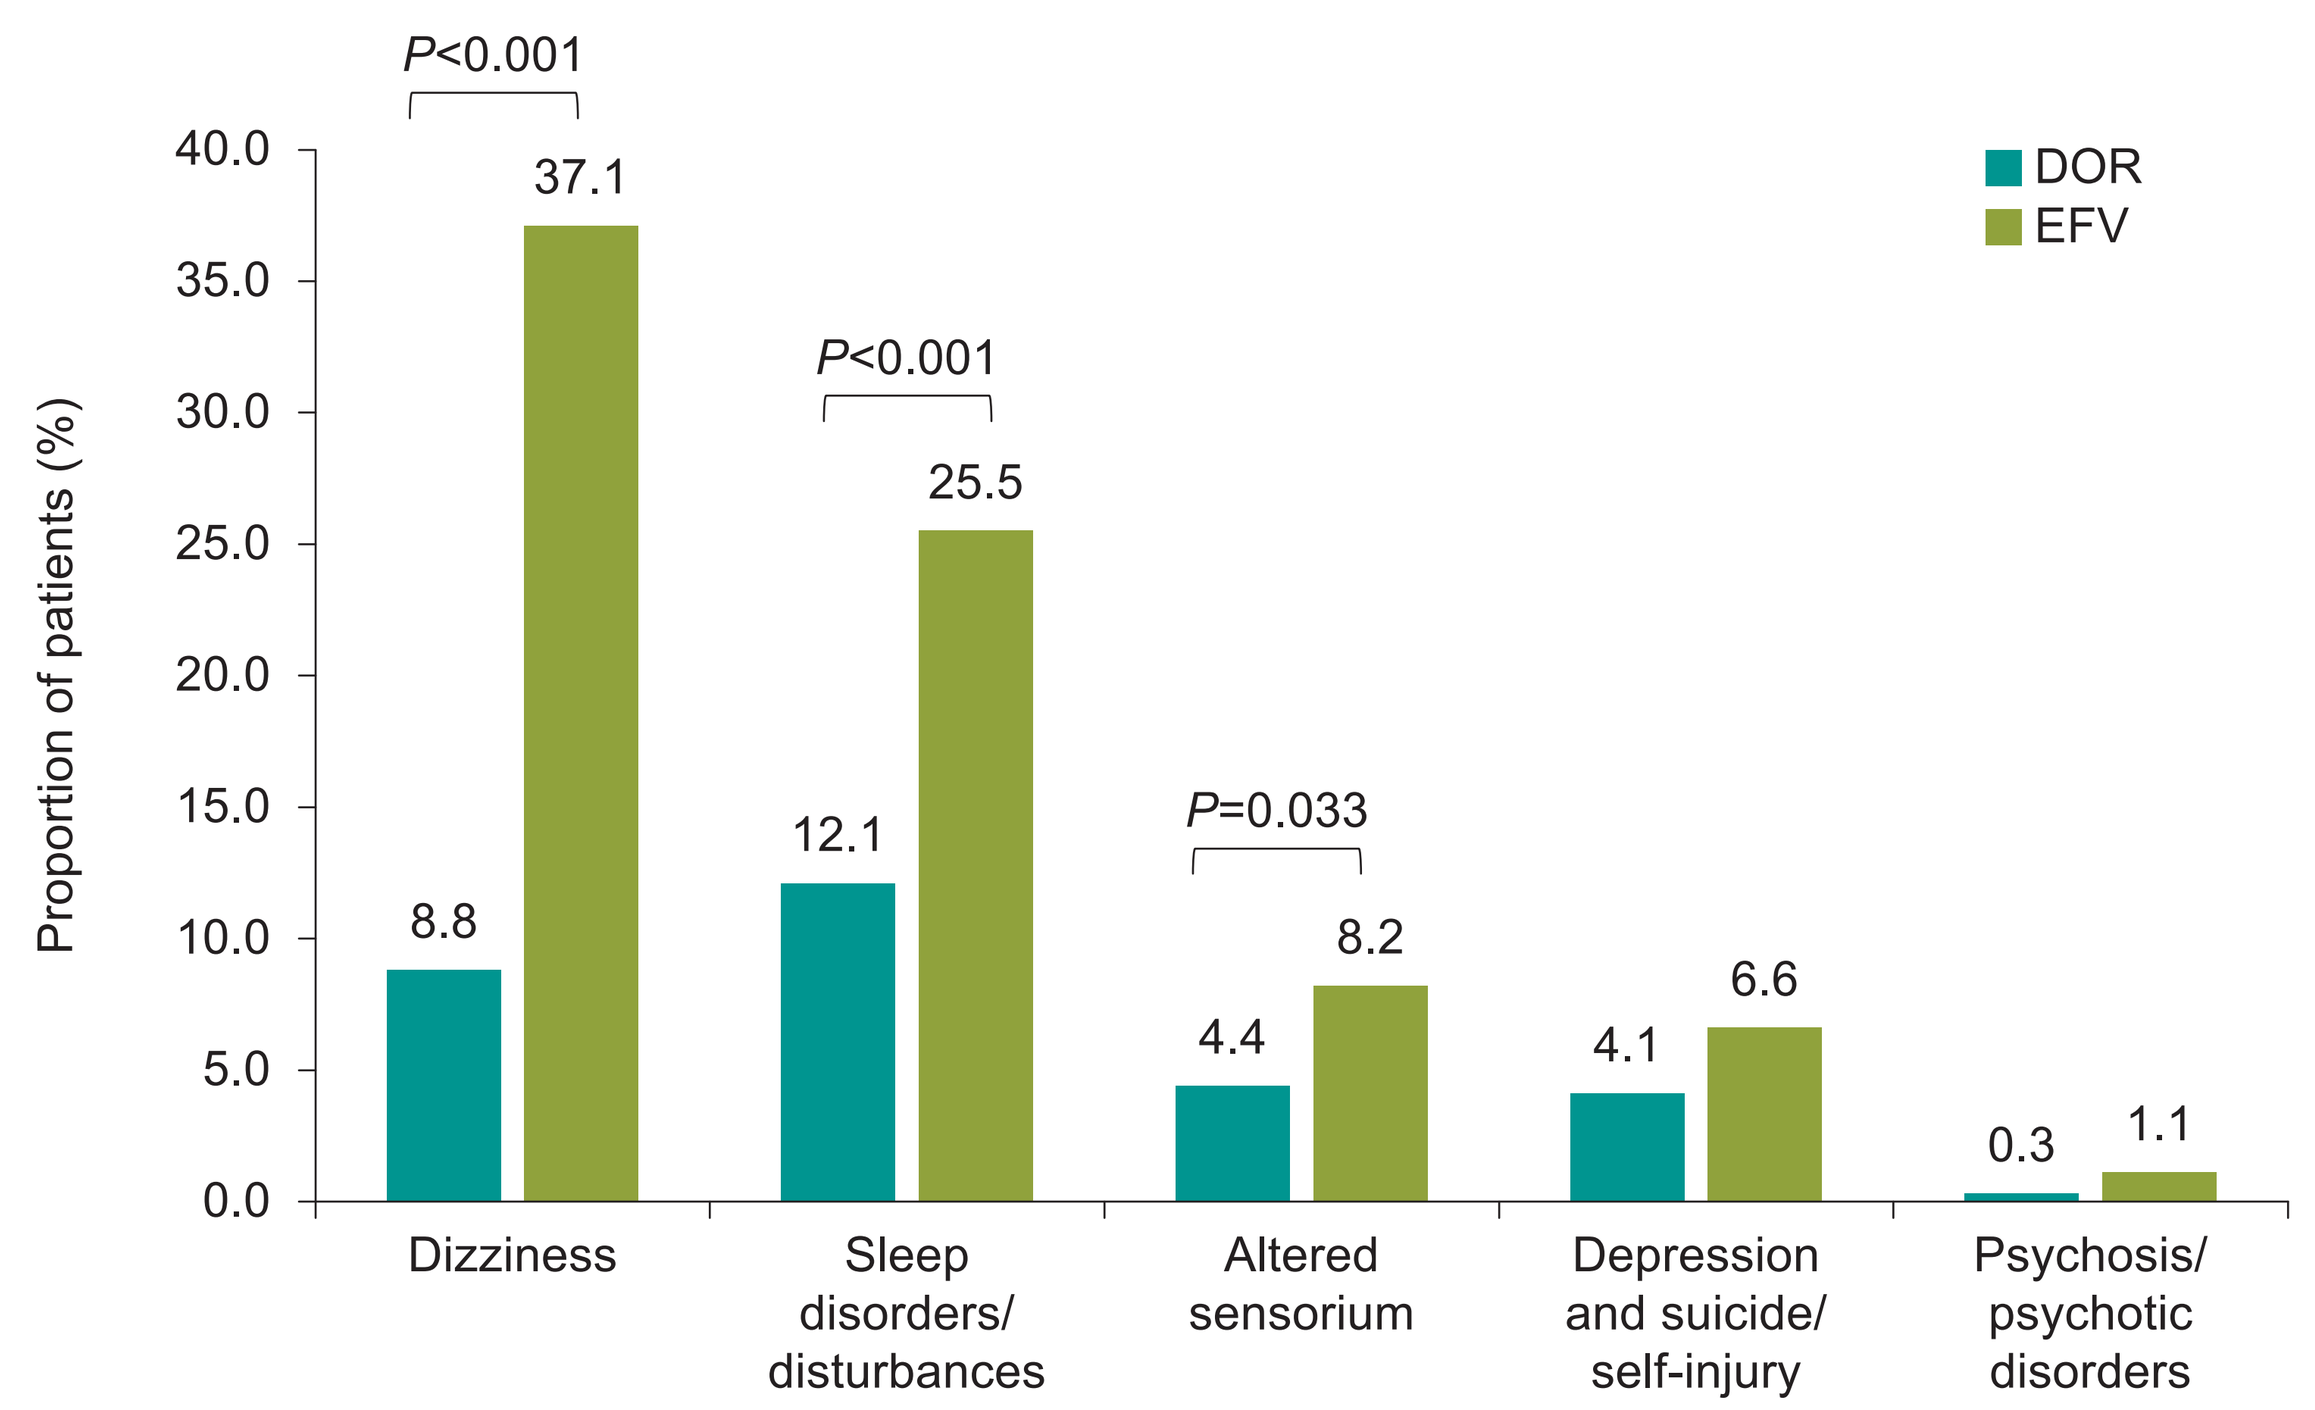
P<0.001
P<0.001
P=0.033
DOR
EFV
Proportion of patients (%)
8.8
37.1
12.1
25.5
4.4
8.2
4.1
6.6
0.3
1.1
Dizziness
Sleep disorders/ disturbances
Altered sensorium
Depression and suicide/ self-injury
Psychosis/ psychotic disorders

[a]Statistical testing for parameters other than dizziness and sleep disorders/disturbances, and altered sensorium were not prespecified.

CONCLUSIONS

- Two decades of research on NNRTIs
 - Numerous failures but wealth of preclinical and clinical experience
 - Helped to build a logical, step-by-step approach to DOR development that addressed many of the critical liabilities of the NNRTI class
- DOR was rationally designed
 - Unique NNRTI
 - Distinct resistance profile
 - Excellent tolerability
 - Noninferior efficacy to EFV and DRV+r
 - Superior neuropsychiatric profile compared with EFV
 - Superior lipid profile compared with DRV+r and EFV

References

1. Das K, Clark AD Jr, Lewi PJ, et al. *J Med Chem.* 2004;47(10):2550-2560.
2. Tucker TJ, Saggar S, Sisko JT, et al. *Bioorg Med Chem Lett.* 2008;18(9):2959-2966.
3. Côté B, Burch JD, Asante-Appiah E, et al. *Bioorg Med Chem Lett.* 2014;24(3):917-922.
4. Lai M, Feng M, Falgueyret, J, et al. *Antimicrob Agents Chemother.* 2014;58(3):1652-1663.
5. Feng M, Wang D, Grobler JA, et al. *Antimicrob Agents Chemother.* 2015;59(1):590-598.
6. Lai M, Sachs N, Xu M, et al. Abstract presented at: Conference on Retroviruses and Opportunistic Infections (CROI). February 22-25, 2016; Boston, MA. Abstract #506.
7. Feng M, Sachs NA, Xu M, et al. *Antimicrob Agents Chemother.* 2016;60(4):2241-2247.
8. Anderson MS, Gilmartin J, Cilissen C, et al. *Antivir Ther.* 2015;20(4):397-405.
9. Khalilieh S, Yee KL, Sanchez RI, et al. Abstract presented at: International AIDS Society (IAS) Conference on HIV Science; July 23-26, 2017; Paris, France. Poster MOPEB0334. Available at: http://programme.ias2017.org/Abstract/Abstract/3996. Accessed February 18, 2018.
10. Khalilieh S, Yee KL, Sanchez RI, et al. *Antimicrob Agents Chemother.* 2017;61(2).
11. Behm MO, Yee KL, Liu R, et al. *Clin Drug Investig.* 2017;37(6):571-579.
12. Sanchez RI, Fillgrove KL, Yee KL, et al. *Xenobiotica.* 2018:1-11.
13. Gallego L, Barreiro P, del Río R, et al. *Clin Infect Dis.* 2004;38(3):430-432.
14. Molina JM, Squires K, Sax P, et al. *Lancet HIV.* 2018;5(5):e211-e220.
15. Orkin C, Squires K, Molina J-M, et al. *Clin Infect Dis.* In press.

Disclosures

Carey Hwang, Ming-Tain Lai, and Daria Hazuda are employees of Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA.

The authors wish to thank Meredith Rogers, MS, CMPP, The Lockwood Group (Stamford, CT) for writing and editorial assistance.

https://bit.ly/2w4GUzG

Presented at IDWeek; San Francisco, CA; October 3–7, 2018.

Copyright © 2018 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.**, Kenilworth, N.J., U.S.A. All Rights Reserved.